Comparative Modulation of Adiponectin Across Select Therapeutics in Clinical Development for MASH with Fibrosis Identifies Lanifibranor as a Differentiated Metabolic Modulator.

Mary E. Rinella[1], Guillaume Wettstein[2], Philippe Huot-Marchand[2], Jason Campagna[2], Sven Francque[3], Manal F. Abdelmalek[4]

[1] Division of Gastroenterology, Hepatology and Nutrition, University of Chicago, Pritzker School of Medicine, Chicago, USA; [2] Inventiva pharma, Daix, France; [3] Department of Gastroenterology and Hepatology, Antwerp University Hospital, Belgium. [4] Division of Gastroenterology and Hepatology, Mayo Clinic, Rochester, MN, USA.

1-INTRODUCTION

Adiponectin (ADP) regulates lipid oxidation, insulin sensitivity, inflammation and fibrogenesis. Low circulating levels of adiponectin are linked to increased insulin resistance, cardiovascular risk and correlate with MASH histologic severity. Activation of PPAR-γ pathways is known to restore adipose-tissue function and increase circulating adiponectin. In the phase 2b NATIVE study, lanifibranor produced a dose-dependent increase in circulating adiponectin together with improvements in histologic MASH resolution and fibrosis regression.

2-Material/Methods

NATIVE evaluated lanifibranor 800 and 1200 mg/day versus placebo in 247 patients with non-cirrhotic MASH for 24 weeks of treatment. ADP serum levels, markers of lipid and glucose metabolism, Insulin Resistance (IR), inflammation, liver chemistries were also measured at baseline (BL) and end-of-treatment (EOT). ADP increase at EOT was defined as unchanged, moderate and high (<1.5-fold (N=78), 1.5-4-fold (N=83) and >4-fold (N=50) change). Data from late-stage therapeutics in MASH were collected from publications.

3-RESULTS

	Lanifibranor (NATIVE) 24 weeks[1]		Resmetirom (MAESTRO-NASH) 52 weeks[2]		Efruxifermin (HARMONY) 24 weeks[3]		Pegozafermin (ENLIVEN) 24 weeks[4]		
Dose	800mg	1200mg	80mg	100mg	28mg	50mg	15mg	30mg	44mg
Baseline µg/ml	4.98 (3.12)	5.13 (3.24)	4.1 (2.6)	4.2 (2.5)	3.5 (1.6)	3.5 (1.5)	4.94		
Absolute change from BL at EOT µg/ml	+ 11.95 (8.97 to14.94)[$]	+ 17.12 (14.29 to 19.96)[$]	+ 0.86 (0.19)*	+ 1.1 (0.19)*	+ 1.4 (0.74 to 2.08)[$]	+ 3.0 (2.34 to 3.71)[$]	+ 1.1 (0.7)[£]	+ 1.1 (0.3)[£]	+ 1.2 (0.4)[£]
Placebo	-0.35 (-3.20 to 2.50)		- 0.1 (0.18)		+ 0.28 (0.32)		- 0.6 (0.4)		

[$] Least Square Mean, 95% confidence intervals; *Mean (Standard Deviation); [£] Least Square Mean (Standard error)

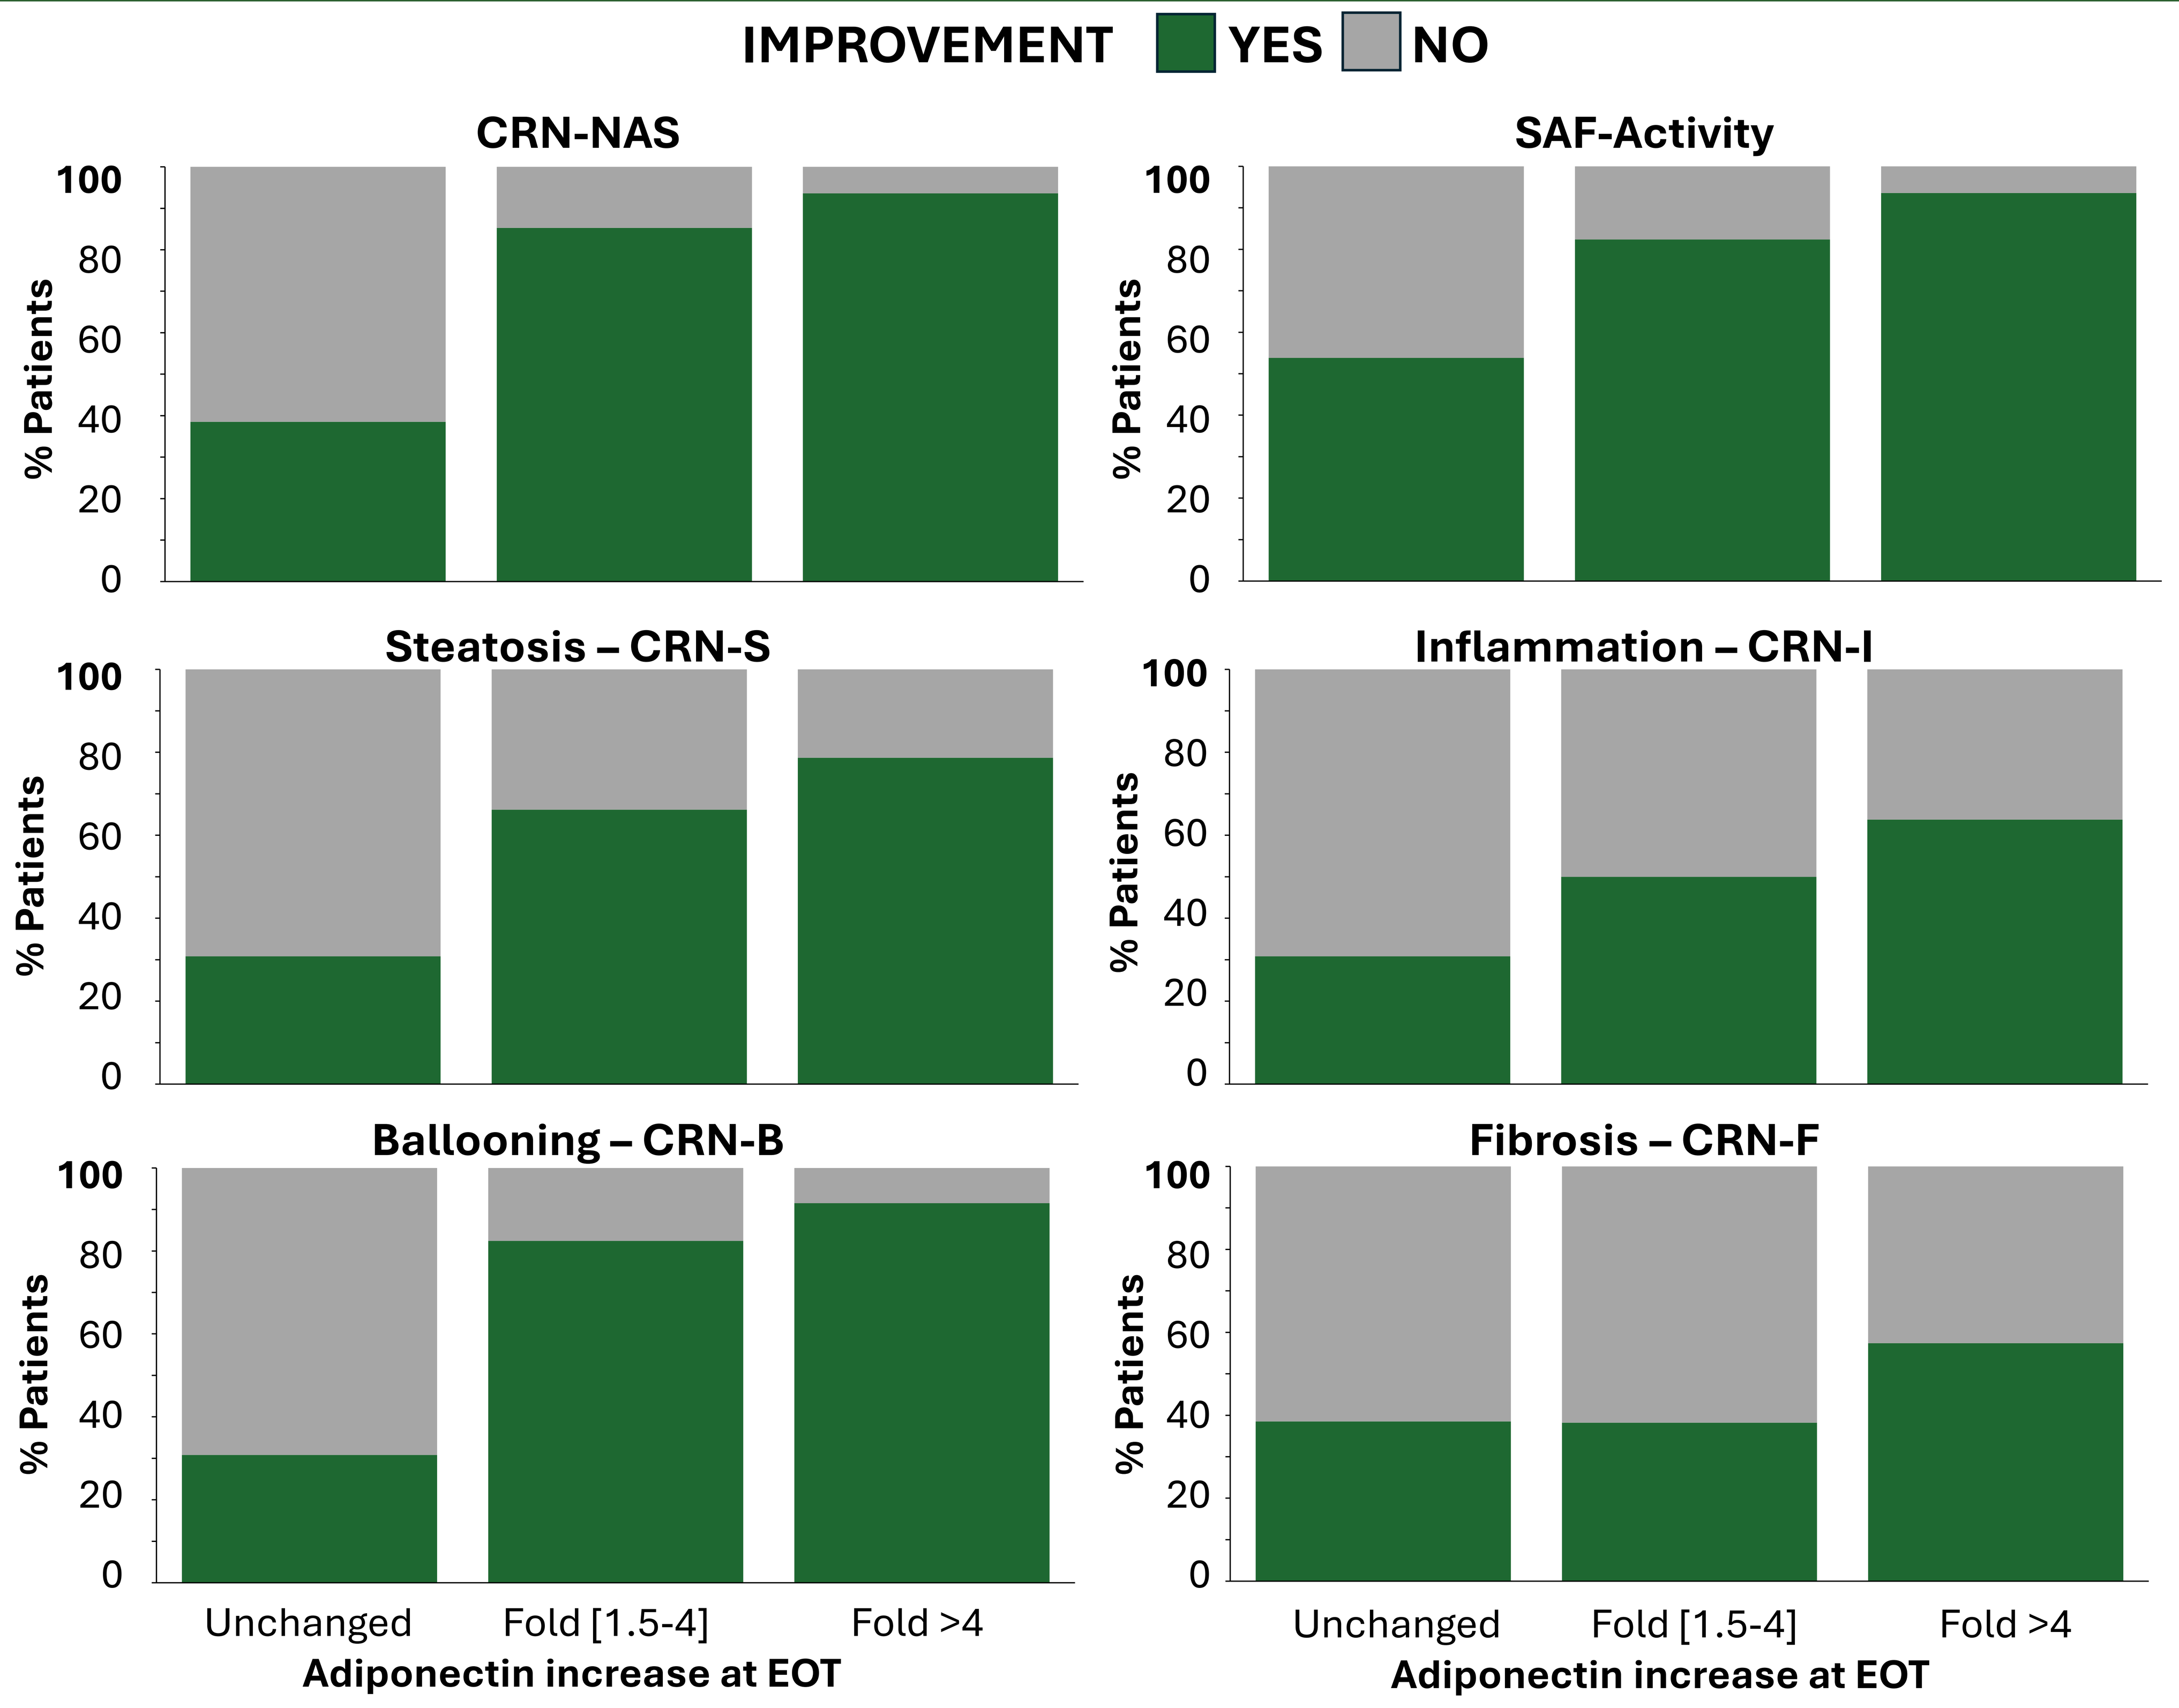
IMPROVEMENT YES NO
CRN-NAS
SAF-Activity
Steatosis – CRN-S
Inflammation – CRN-I
Ballooning – CRN-B
Fibrosis – CRN-F
% Patients
Unchanged
Fold [1.5-4]
Fold >4
Adiponectin increase at EOT

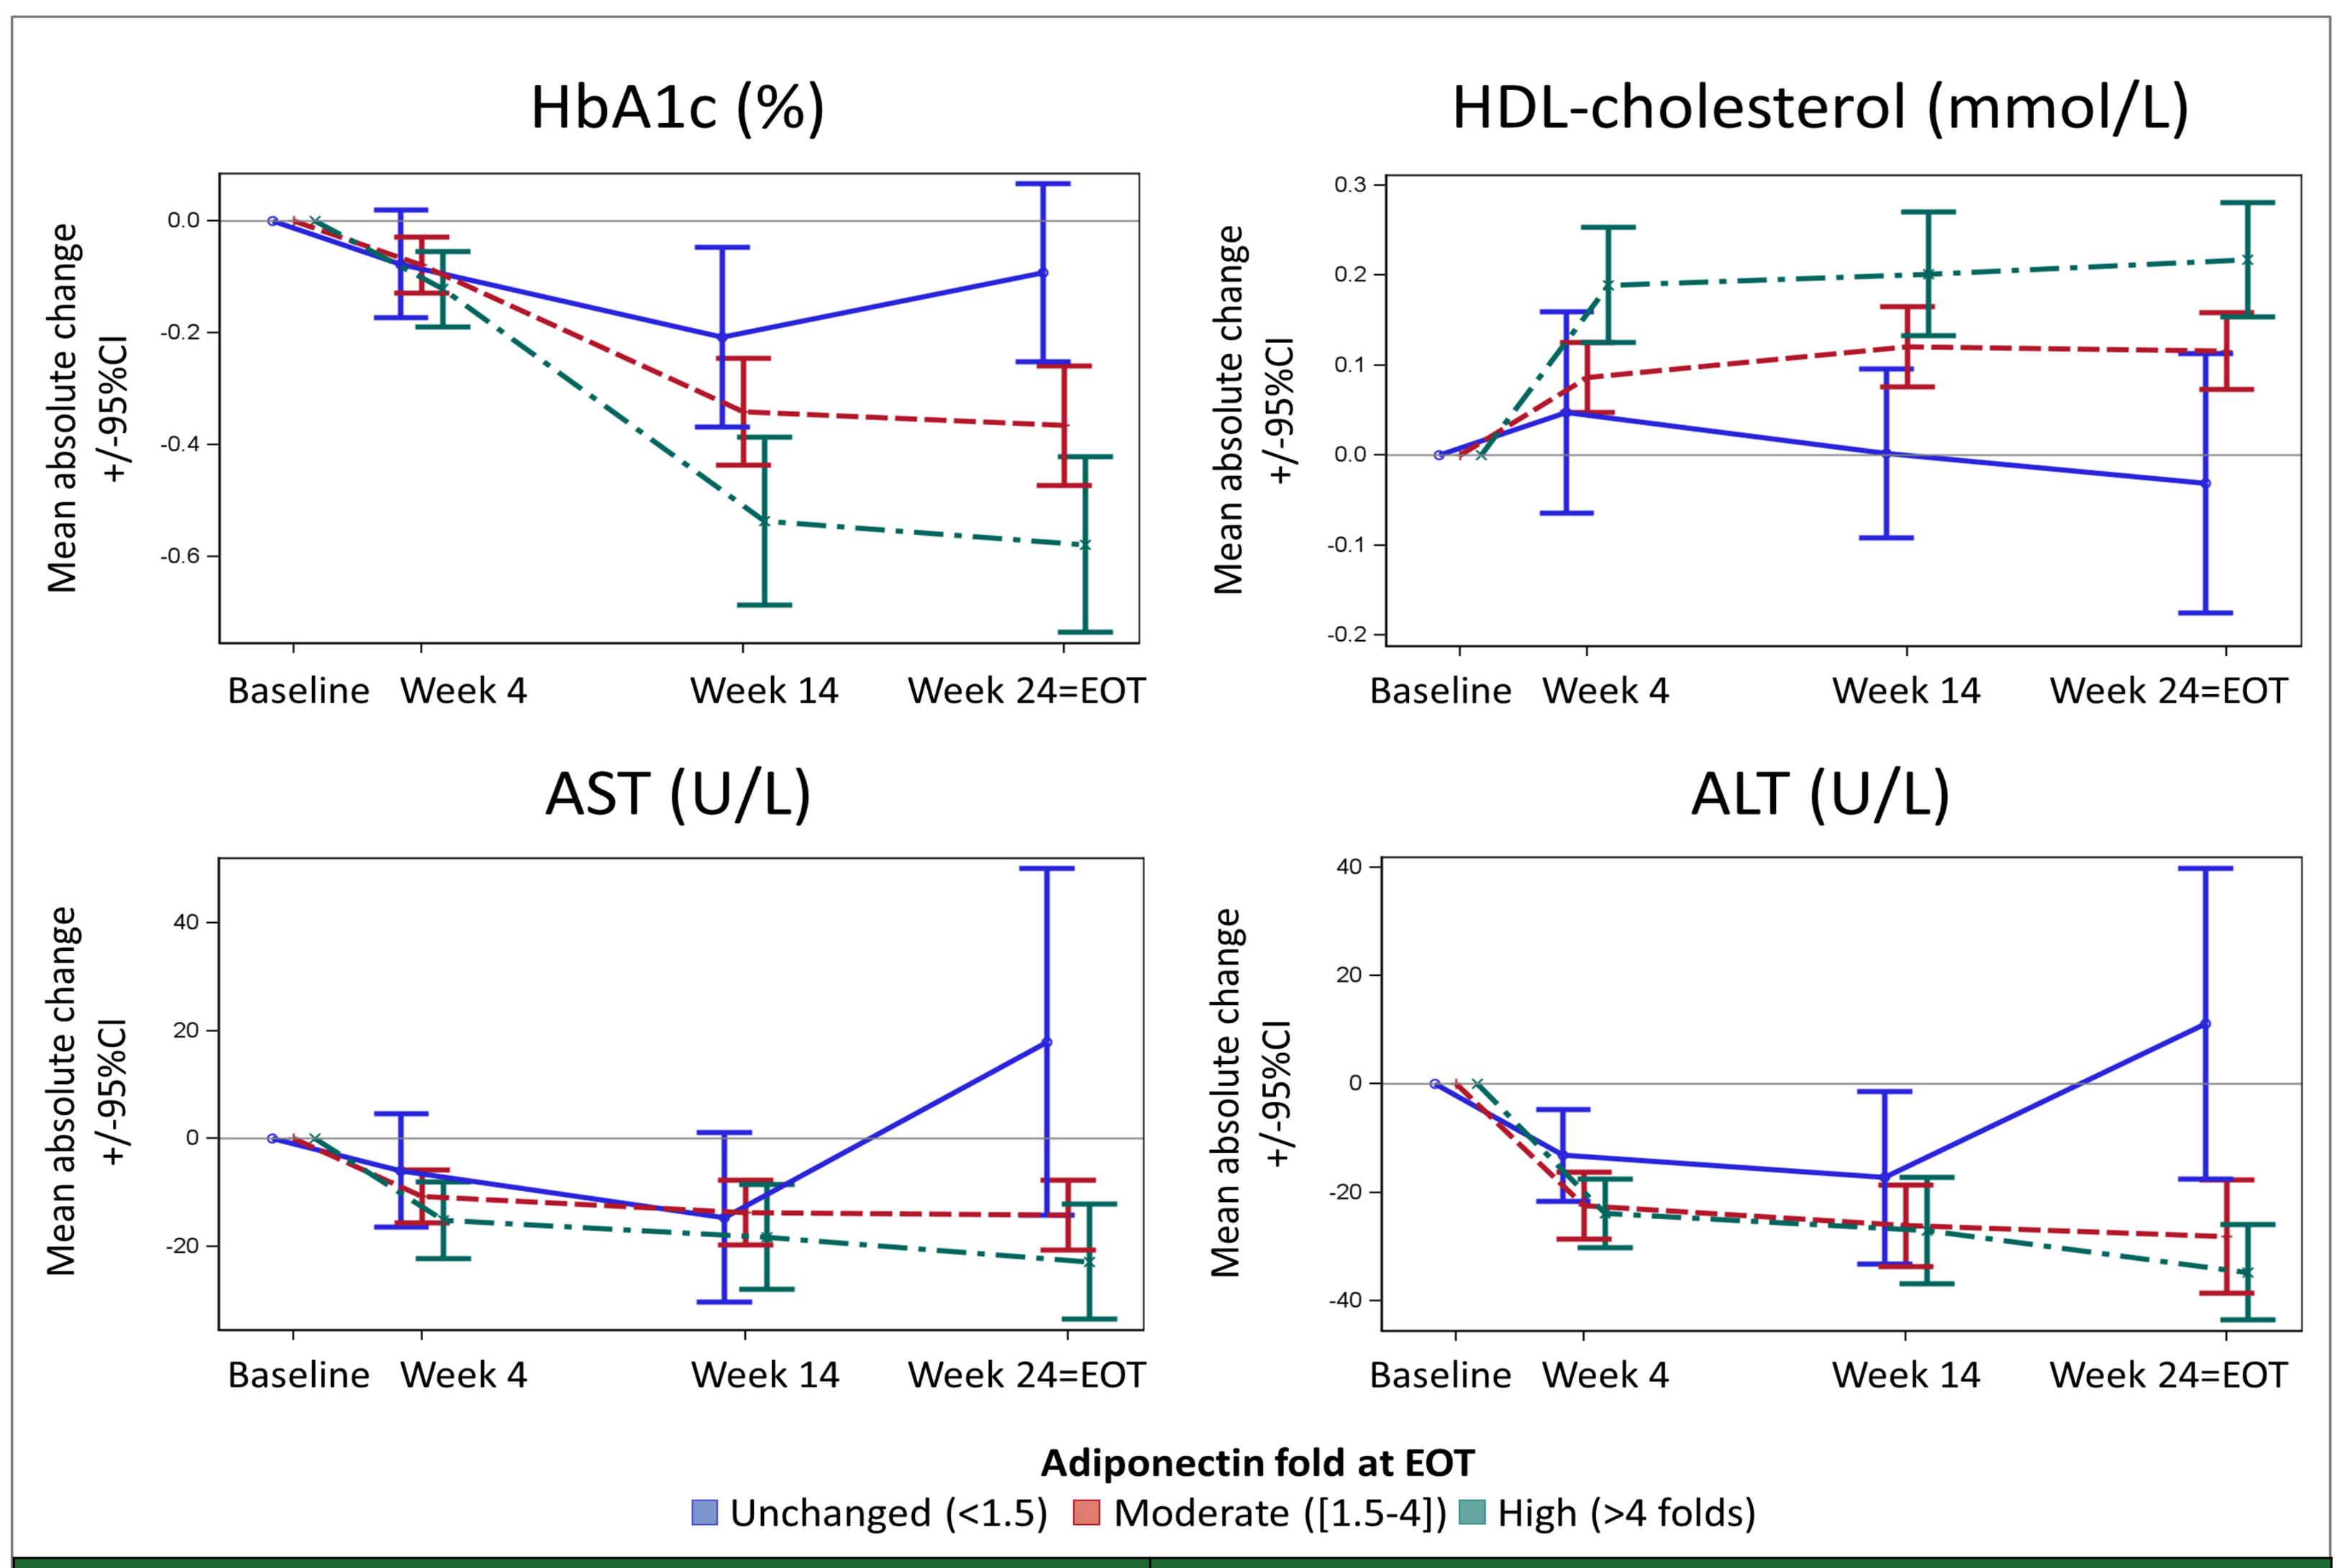
HbA1c (%)
HDL-cholesterol (mmol/L)
AST (U/L)
ALT (U/L)
Mean absolute change +/-95%CI
Baseline
Week 4
Week 14
Week 24=EOT
Adiponectin fold at EOT
Unchanged (<1.5)
Moderate ([1.5-4])
High (>4 folds)

Absolute change at EOT (mean ± SD)	Adiponectin fold at EOT		
	Unchanged <1.5 folds	Moderate [1.5 - 4] folds	High >4 folds
Glucose (mmol/L)	0.20 ± 0.79	-0.83 ± 1.33	-1.01 ± 1.63
Insulin (pmol/L)	-17.3 ± 64.3	-115.4 ± 207.4	-177.8 ± 342.1
HOMA-IR	-0.50 ± 3.04	-5.70 ± 10.53	-9.17 ± 20.13
Triglycerides (mmol/L)	-0.23 ± 0.62	-0.36 ± 0.96	-0.59 ± 0.51
Apolipoprotein B (mg/dL)	-3 ± 24	-9 ± 17	-19 ± 22
High-sensitivity C Reactive Protein (mg/L)	1.02 ± 6.63	-1.73 ± 5.60	-2.90 ± 4.82

In the NATIVE study, the degree of adiponectin increase correlated with improvement of CRN-NAS and SAF-Activity scores as well as with individual components (steatosis, inflammation, ballooning) other liver-related parameters as well as established cardiometabolic parameters.

4-CONCLUSION

Among MASH therapeutics in clinical development, lanifibranor induces the largest reported increase in ADP, with substantial changes that correlate with histologic improvement in both MASH activity and fibrosis[6]. This relationship aligns mechanistically with pan-PPAR-mediated restoration of adipose tissue function and enhanced hepatic and muscle insulin sensitivity. Importantly, these increases also correlate with improvements in cardiometabolic parameters (e.g., HbA1c, HDL, and hepatic steatosis)[7], supporting ADP as a systemic biomarker of metabolic normalization , driving liver improvement. Together, these findings support the potential of lanifibranor as a differentiated therapy capable of addressing both intrahepatic and extrahepatic complications of MASH through multi-pathway PPAR engagement.

REFERENCES

[1] Francque S., et al. A Randomized, Controlled Trial of the Pan-PPAR Agonist Lanifibranor in NASH. N Engl J Med. 2021; 385(17):1547-1558
[2] Harrison S., *et al.* A Phase 3, Randomized, Controlled Trial of Resmetirom in NASH with Liver Fibrosis. N Engl J Med, 2024; 390(6):497-509
[3] Harrison S., *et al.* Safety and efficacy of once-weekly efruxifermin versus placebo in non-alcoholic steatohepatitis (HARMONY): a multicentre, randomised, double-blind, placebo-controlled, phase 2b trial. Lancet Gastroenterol Hepatol, 2023; 8(12):1080-109
[4] Loomba R., *et al*. Randomized, Controlled Trial of the FGF21 Analogue Pegozafermin in NASH. N Engl J Med, 2023; 389(11):998-1008
[5] P.J. Eddowes and al. Accuracy of FibroScan Controlled Attenuation Parameter and Liver Stiffness Measurement in Assessing Steatosis and Fibrosis in Patients With Nonalcoholic Fatty Liver Disease. Gastroenterology. 2019 May;156(6):1717-1730. doi: 10.1053/j.gastro.2019.01.042. Epub 2019 Jan 25. PMID: 30689971
[6] No head-to-head clinical trials amongst late stage agents have been conducted, results obtained from different published trials with different patient populations. Results may not be comparable.
[7] Cooreman M. et al., The pan-PPAR agonist lanifibranor improves cardiometabolic health in patients with metabolic dysfunction-associated steatohepatitis. Nat Commun. 2024; 15(1):3962