Lanifibranor-associated adiponectin increase correlates with improvement of histological and serum markers of NASH severity both in terms of activity and fibrosis

Michael P. Cooreman[1], Manal F. Abdelmalek[2], Philippe Huot-Marchand[1], Lucile Dzen[1], Martine Baudin[1], Jean-Louis Junien[1], Pierre Broqua[1], Sven M. Francque[3]

1 INVENTIVA, Daix, France. **2** Division of Gastroenterology and Hepatology, Duke University, Durham, USA. **3** Department of Gastroenterology and Hepatology, Antwerp University Hospital, Belgium.

1-INTRODUCTION

Lanifibranor has shown efficacy on liver histology and markers of cardiometabolic health (CMH) in patients with pre-cirrhotic NASH. We previously showed that increases in adiponectin (ADP) with lanifibranor correlated with improvement of CMH markers. We further analysed the correlation between ADP increase and lanifibranor for 1) histological features of NASH severity, and 2) biomarkers of active NASH and fibrosis.

2-MATERIAL/METHODS

The phase 2 NATIVE study evaluated lanifibranor 800 and 1200 mg/d versus placebo in 247 patients for 24 weeks(1).
Paired samples for histological efficacy were available from 211 patients (72 on placebo, 66 and 73 on lanifibranor 800 and 1200 mg).
Serum levels of ADP, pro-C3, and MACK-3 score (a score for active and fibrotic NASH calculated from HOMA-IR, aspartate aminotransferase [AST] and cytokeratin 18 [CK-18]) were measured at baseline (BL) and end of treatment (EOT).
BL ADP levels were divided into low, medium and high (<5, 5-10 and >10 ug/mL); ADP increase at EOT was defined as unchanged, moderate and high (<1.5-fold, 1.5-4-fold and >4-fold change).
Histological NASH activity grading (steatosis, inflammation, ballooning) and fibrosis staging were per NASH-CRN and SAF scoring.
Histological endpoints were NASH resolution and no worsening of fibrosis (E1), improvement of fibrosis and no worsening of NASH (E2), and NASH resolution AND fibrosis improvement (E3).

3-RESULTS

BL ADP levels were low across groups.

Adiponectin (µg/mL) at BL	Placebo	Lanifibranor			Total
		800 mg	1200 mg	Pooled	
N	81	80	83	163	244
Mean (SD)	5.3 ± 4.2	5.0 ± 3.1	5.1 ± 3.2	5.1 ± 3.2	5.1 ± 3.5
Median	4.6	4.2	4.4	4.3	4.5
[Min ; Max]	1.2; 30.8	1.3; 22.3	0.8; 14.7	0.8; 22.3	0.8; 30.8

ADP increase at EOT

Fold Adiponectin at W24	Placebo	Lanifibranor		
		800 mg	1200 mg	Pooled
N	72	66	73	139
Mean (SD)	1.1 ± 0.3	3.8 ± 3.1	4.5 ± 3.5	4.2 ± 3.3
Median	1.0	2.8	3.5	3.0
[Min ; Max]	0.1; 2.5	0.8; 18.1	0.7; 17.4	0.7; 18.1
Unchanged	65 (90%)	9 (14%)	4 (5%)	13 (9%)
Fold [1.5 - 4]	7 (10%)	38 (58%)	38 (52%)	76 (55%)
Fold >4	0 (0.0%)	19 (29%)	31 (42%)	50 (36%)

With lanifibranor, ADP increase at EOT correlated with all histological endpoints: responders with lanifibranor (pooled) were for E1: 15, 44 and 57%, E2: 31, 35 and 53%, and E3: 15, 28 and 40% for ADP unchanged, moderate or high increase, respectively.

With placebo, a small (10)% of patients had a moderate ADP increase (all <4-fold), which also correlated with histological response, but to a lesser degree.

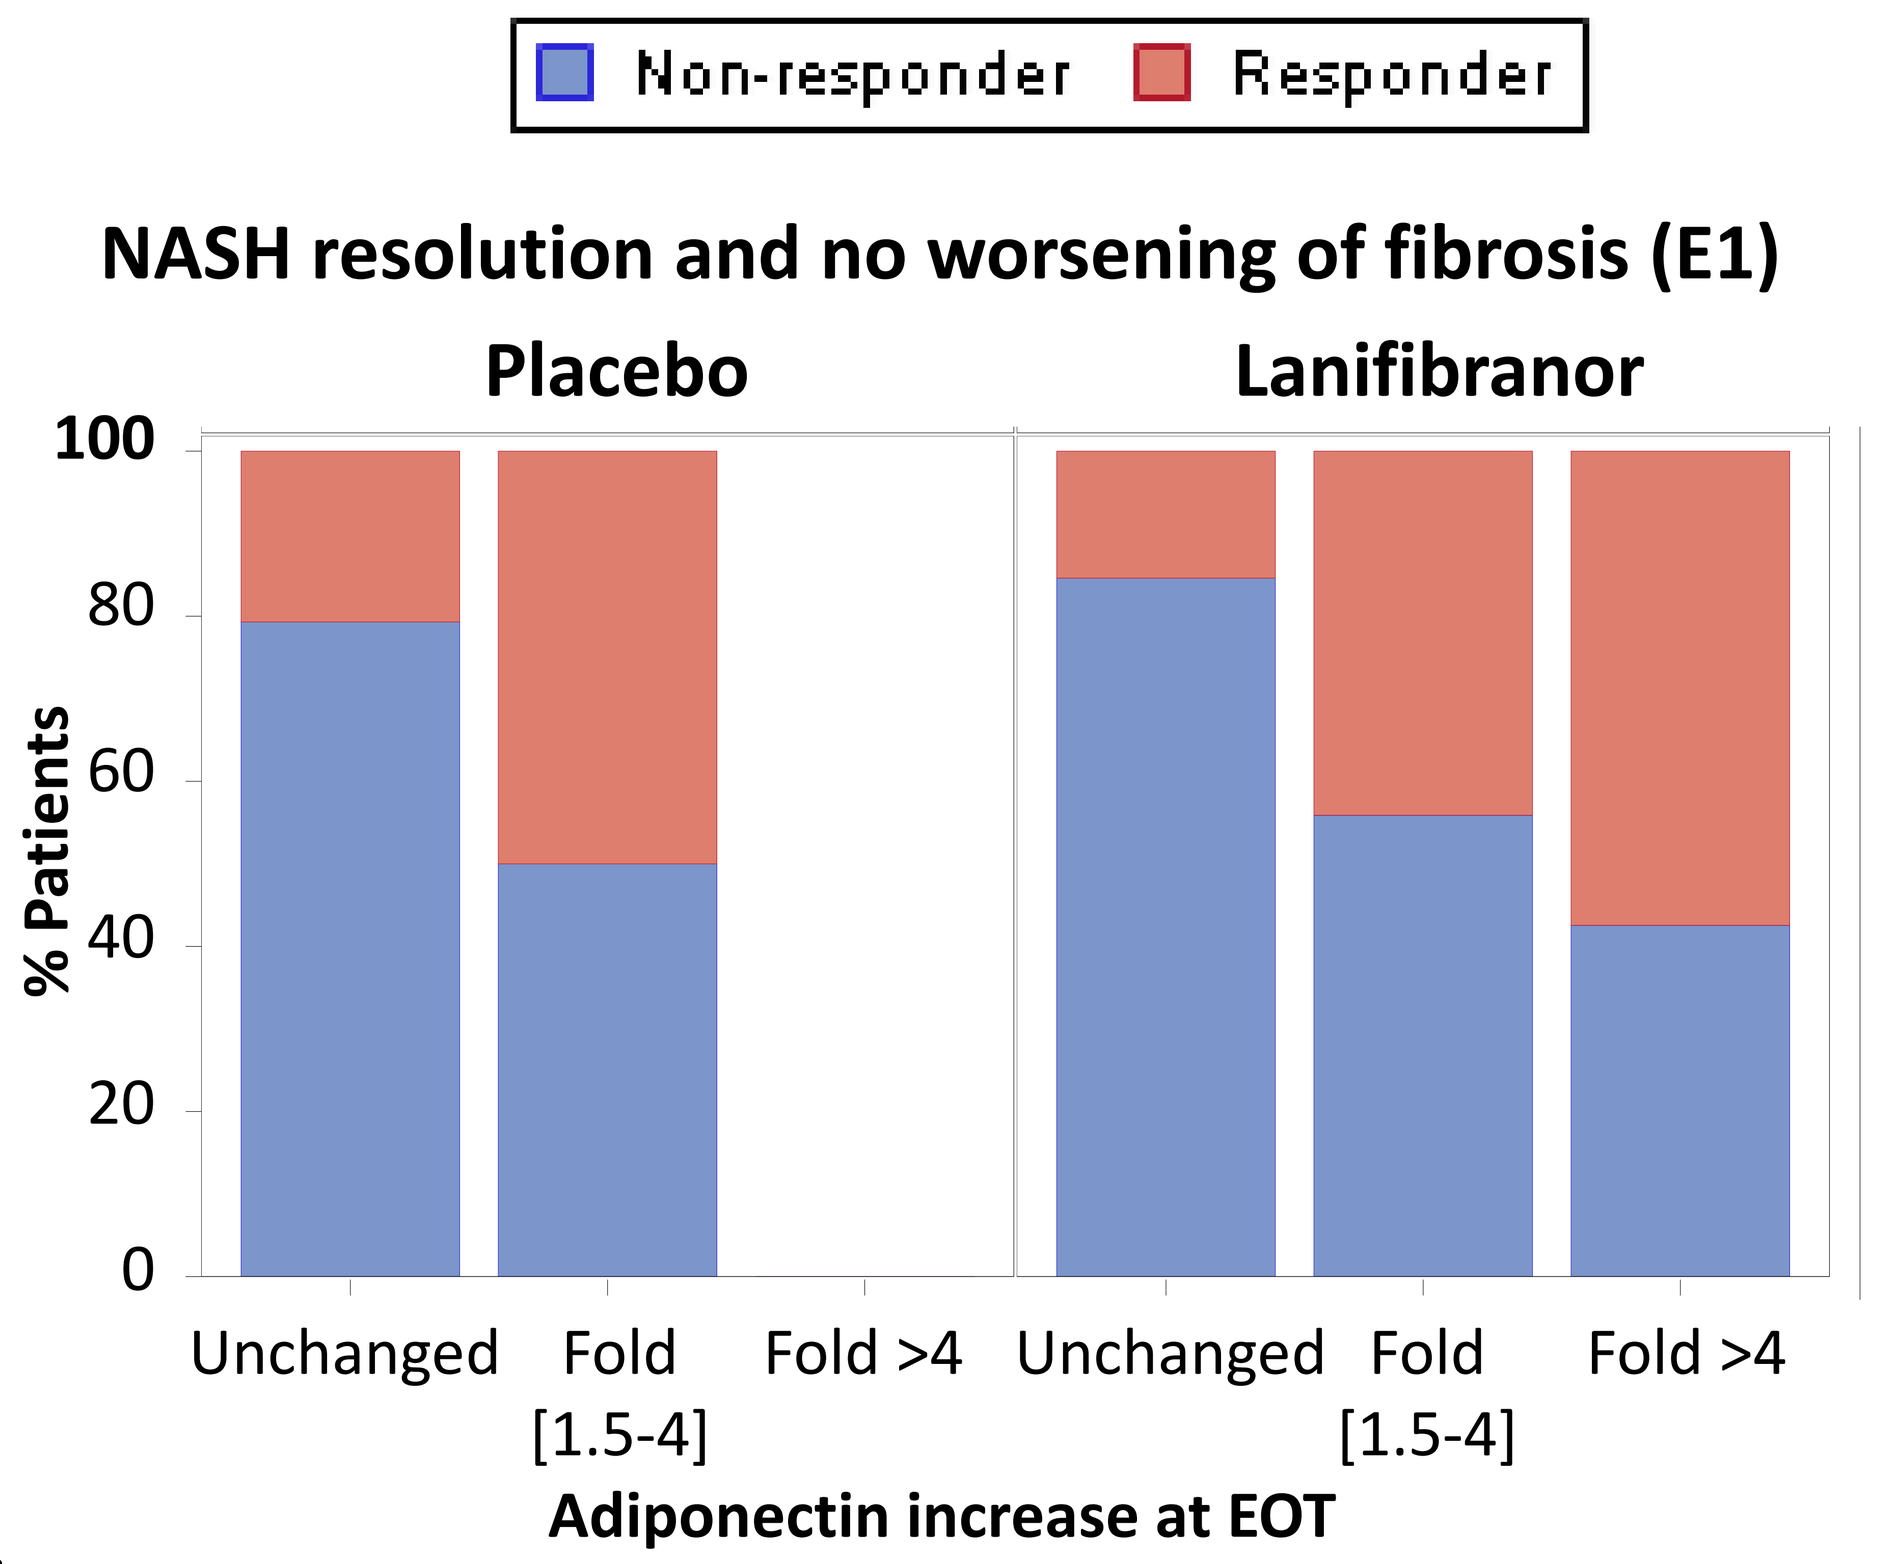
Non-responder
Responder
NASH resolution and no worsening of fibrosis (E1)
Placebo
Lanifibranor
% Patients
100
80
60
40
20
0
Unchanged
Fold [1.5-4]
Fold >4
Adiponectin increase at EOT

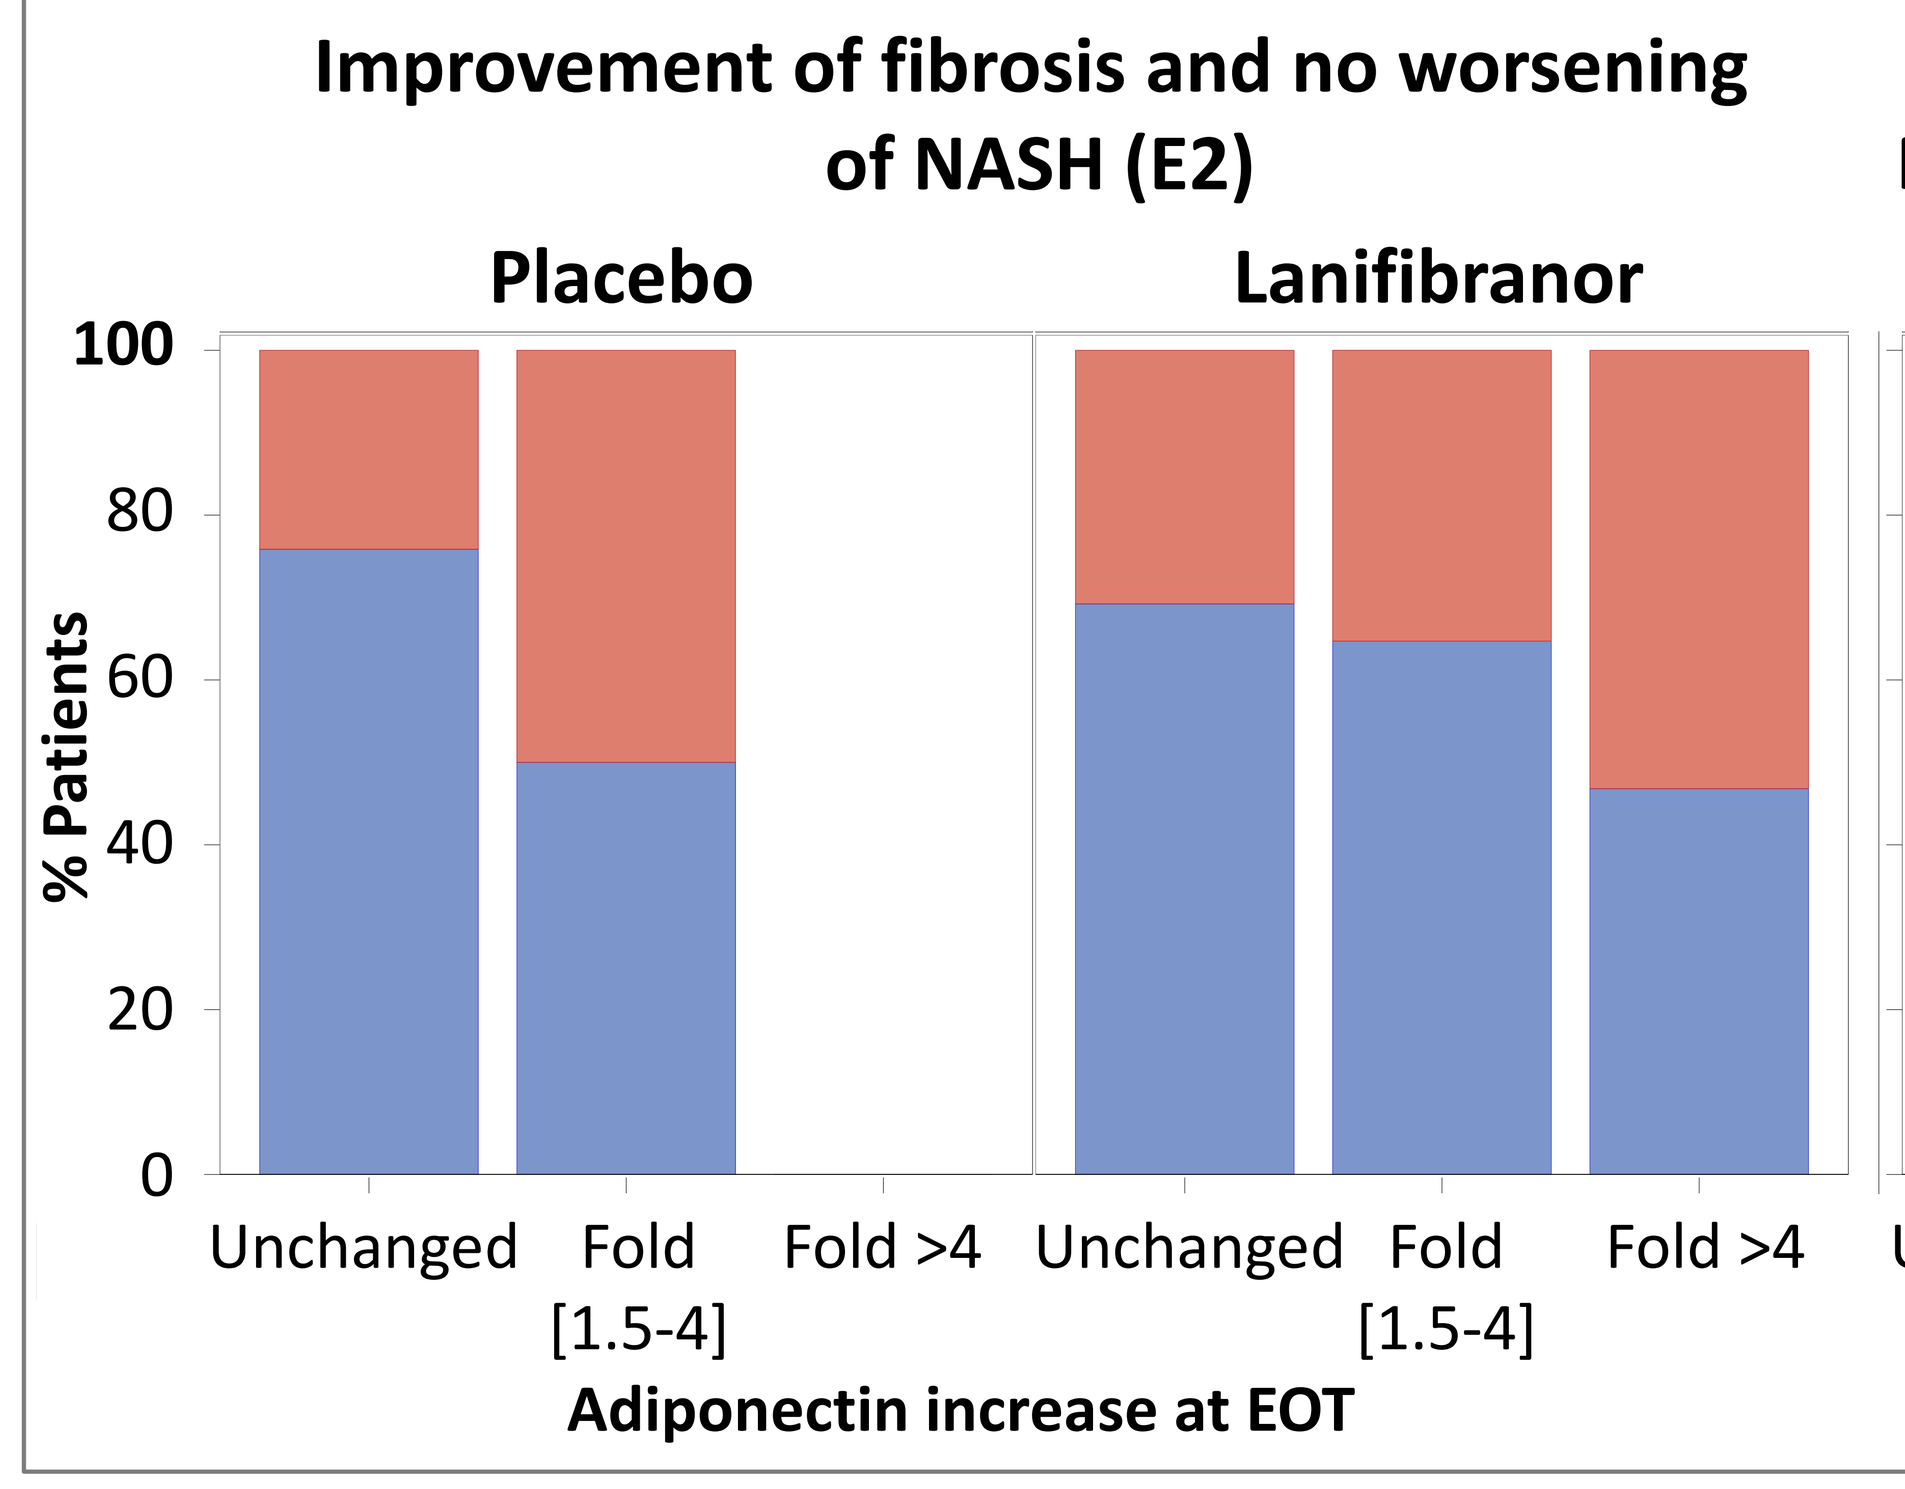
Improvement of fibrosis and no worsening of NASH (E2)
Placebo
Lanifibranor
% Patients
100
80
60
40
20
0
Unchanged
Fold [1.5-4]
Fold >4
Adiponectin increase at EOT

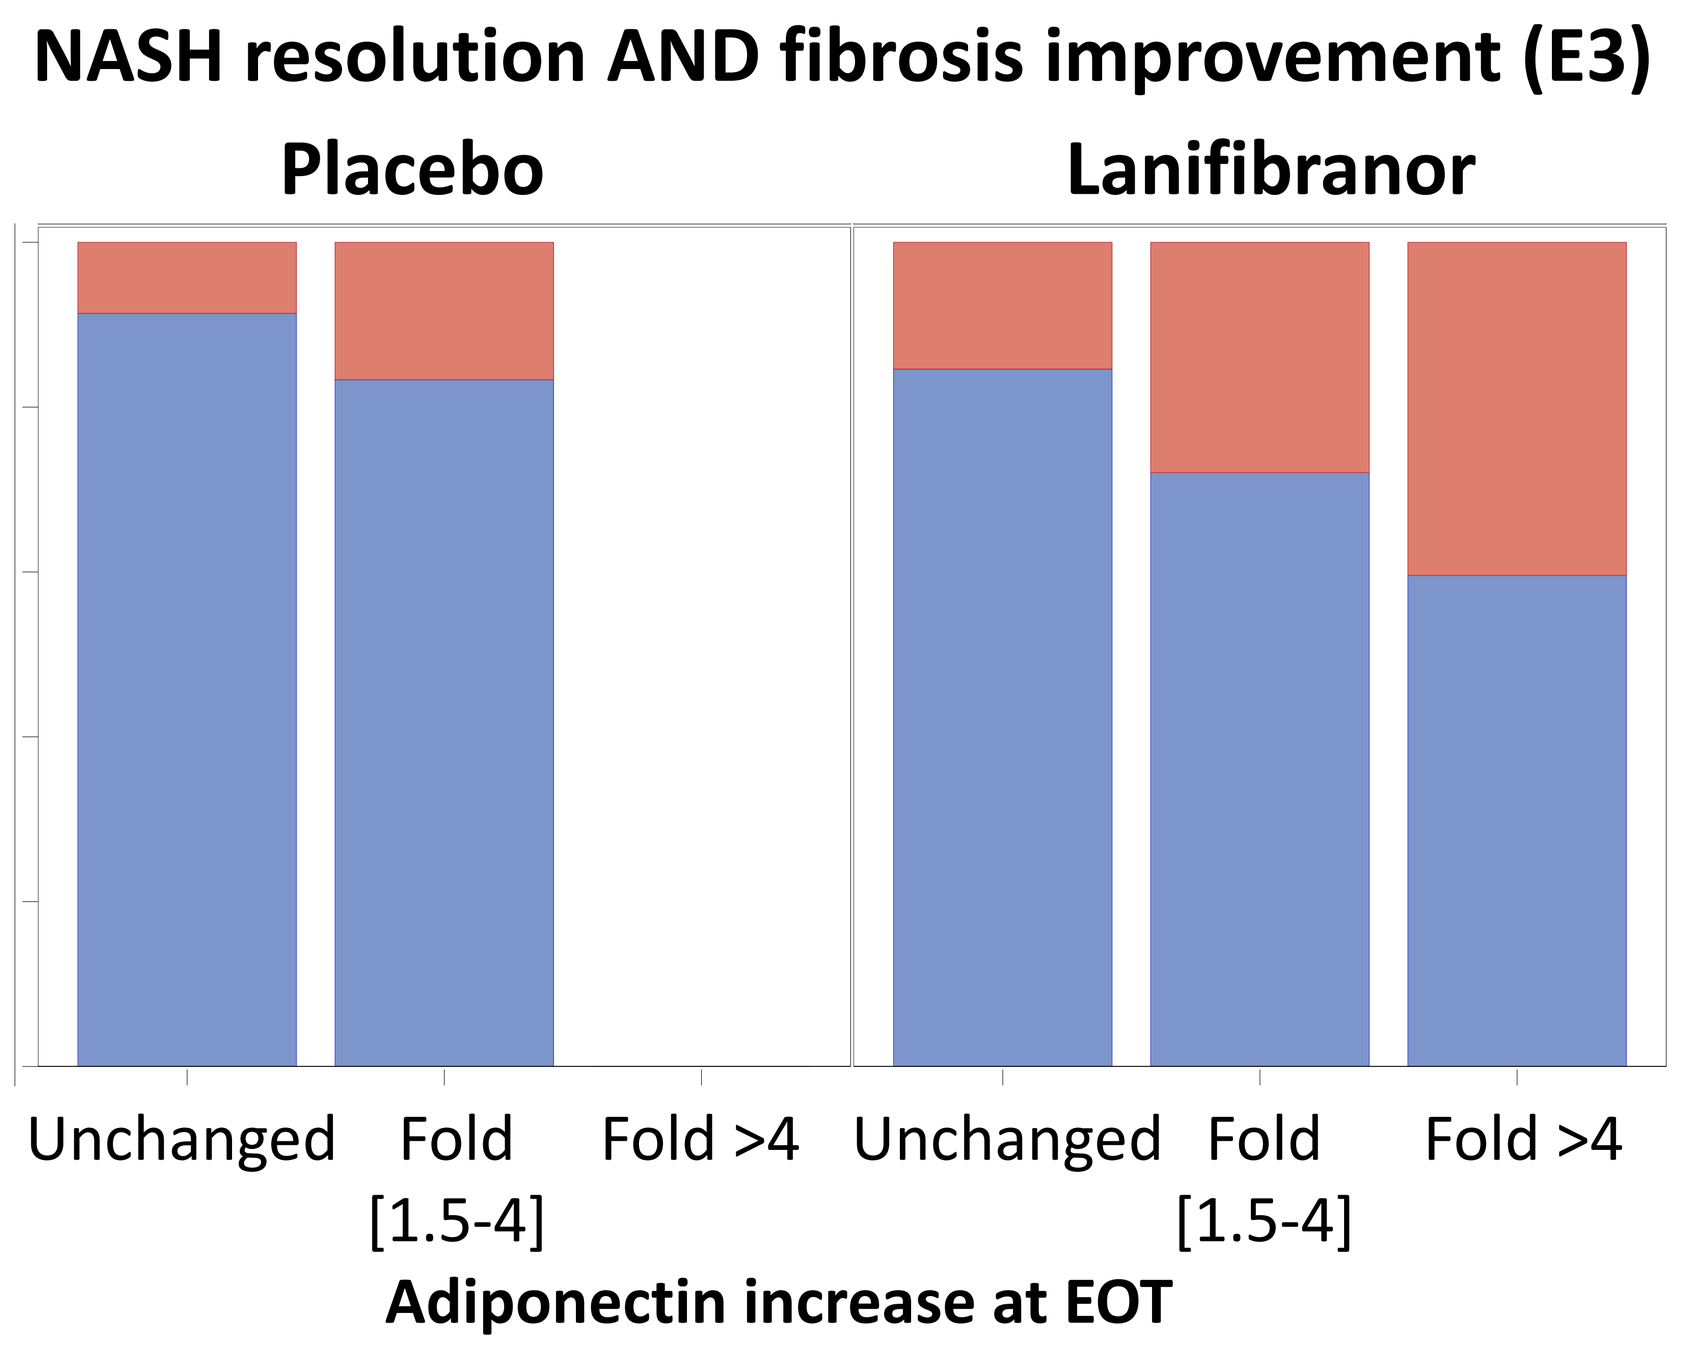
NASH resolution AND fibrosis improvement (E3)
Placebo
Lanifibranor
Unchanged
Fold [1.5-4]
Fold >4
Adiponectin increase at EOT

Degree of ADP increase, both absolute and categorical, correlated with improvement of CRN-NAS and SAF-Activity scores as well as with individual components (steatosis, inflammation, ballooning).

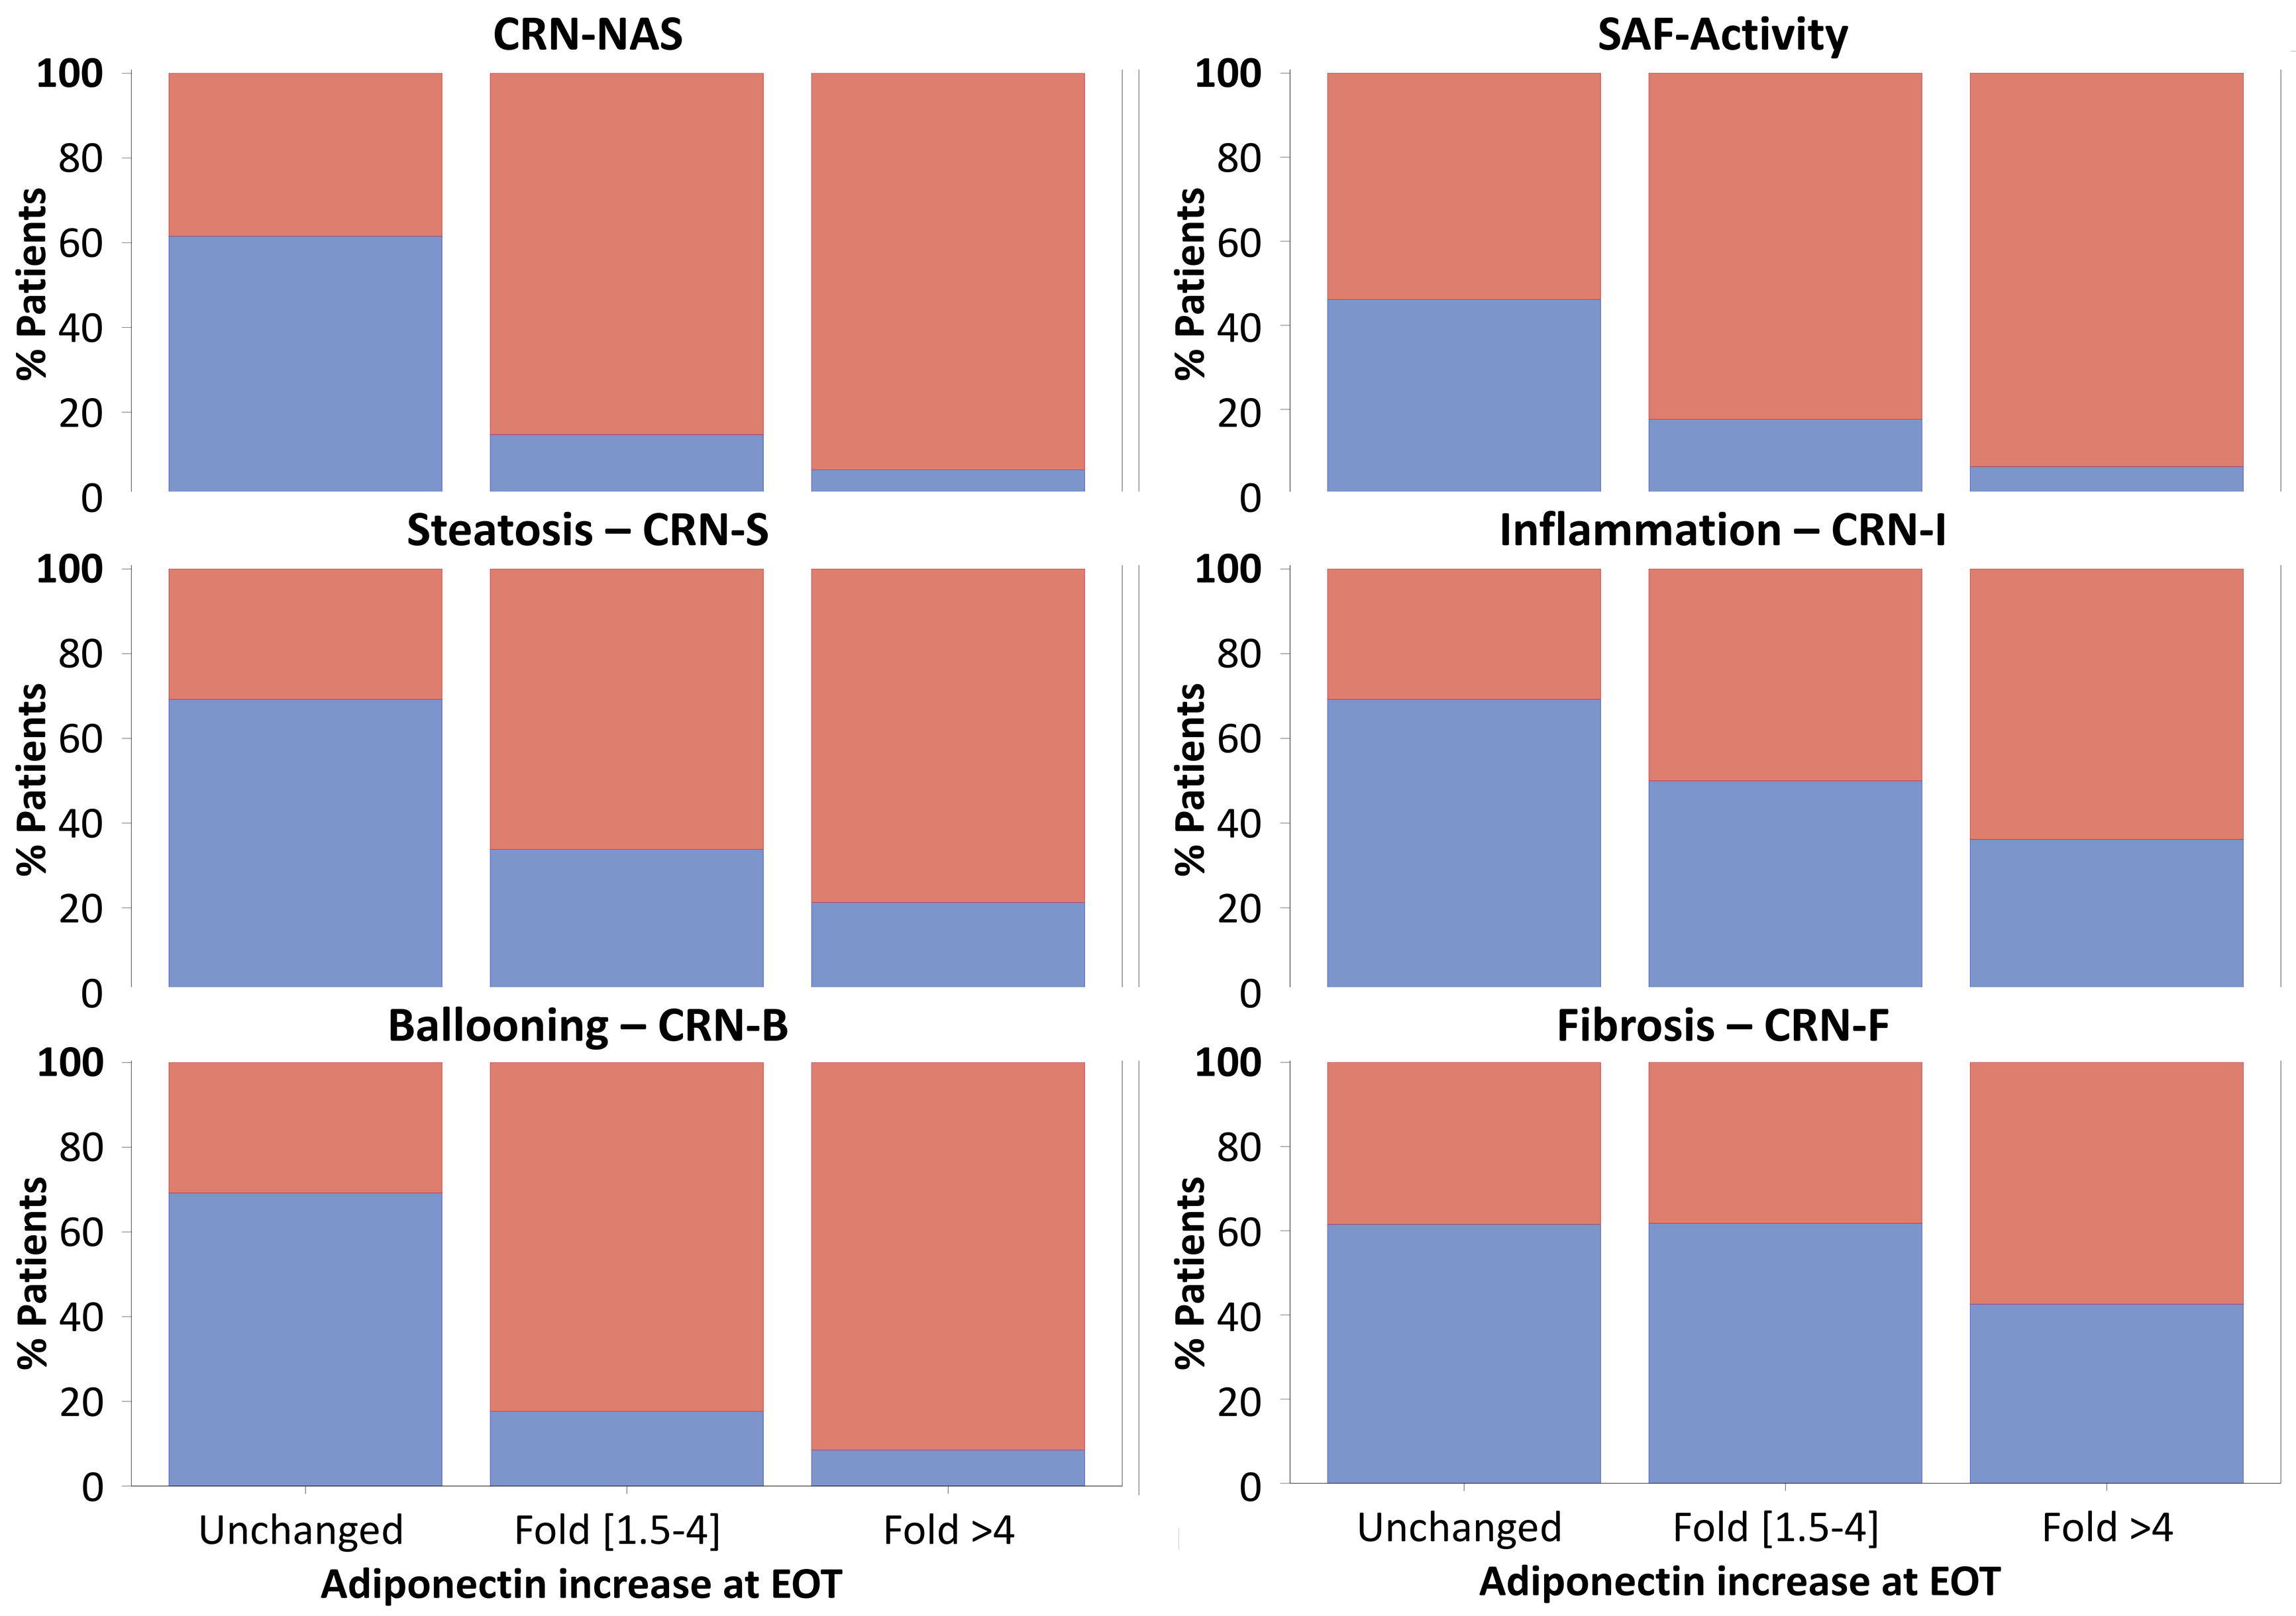
improvement
No
Yes
CRN-NAS
SAF-Activity
Steatosis – CRN-S
Inflammation – CRN-I
Ballooning – CRN-B
Fibrosis – CRN-F
% Patients
100
80
60
40
20
0
Unchanged
Fold [1.5-4]
Fold >4
Adiponectin increase at EOT

Lanifibranor-associated ADP increase correlated with improvement in fibrosis stage (38% vs 57% for ADP ≤4 vs >4-fold increase, respectively), pro-C3 levels and MACK-3 score.

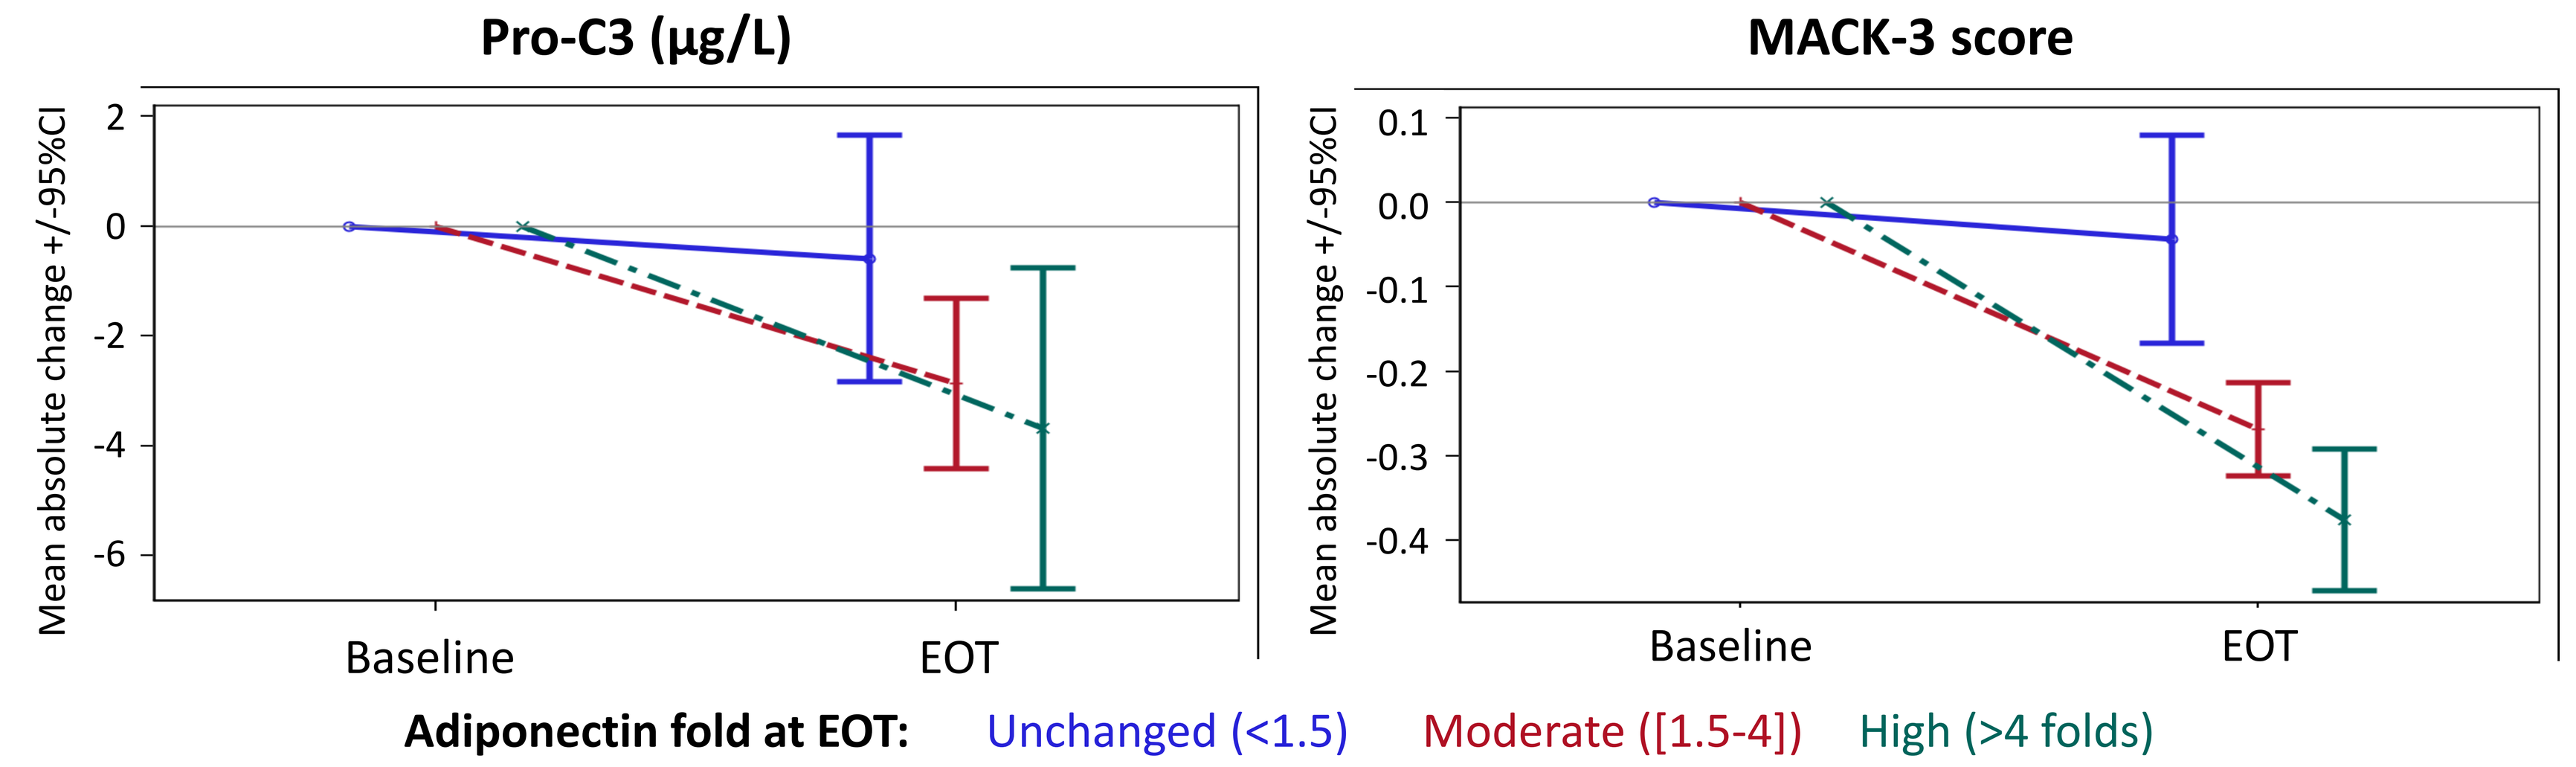
Pro-C3 (µg/L)
MACK-3 score
Mean absolute change +/-95%CI
2
0
-2
-4
-6
0.1
0.0
-0.1
-0.2
-0.3
-0.4
Baseline
EOT
Adiponectin fold at EOT:
Unchanged (<1.5)
Moderate ([1.5-4])
High (>4 folds)

4-CONCLUSION

ADP increase with lanifibranor in patients with NASH correlated with improvement of histological and serum markers of NASH activity and fibrosis, further supporting that the observed improvements correlate with lanifibranor target engagement. ADP, across the spectrum of NASH, is a biomarker of lanifibranor-associated treatment.

References:
[1] S.M. Francque and al. A Randomized, Controlled Trial of the Pan-PPAR Agonist Lanifibranor in NASH. *N Engl J Med.* 2021;385:1547-58.

Contact information:

Dr. Michael P. COOREMAN: Michael.COOREMAN@inventivapharma.com