A rational approach for discovery of inhibitors of YAP-TEAD interaction

Laurent Chene, Anne Soude, Christelle Valaire, Severine Delaporte, Sebastien Jacquet, Yves Cambet,
Isabelle Braccini, Martine Barth, Christian Montalbetti, Pierre Broqua & Claudia Z-A Fromond

Inventiva, Daix, France

BACKGROUND

- The Hippo pathway plays a major role in organ growth regulation and tumorigenesis
- By sensing contact inhibition, the central kinase network of the hippo pathway controls YAP nuclear translocation
- YAP interacts with the TEAD family of transcription factors and drives expression of gene involved in cell survival and proliferation
- YAP, TAZ and TEADs over-expressed in cancer:
 - Colon
 - Lung
 - Gastric
 - Ovarian
- Knockdown of YAP inhibits proliferation and metastasis of gastric cancer cells
- A dominant-negative TEAD molecule suppresses YAP-induced liver tumorigenesis

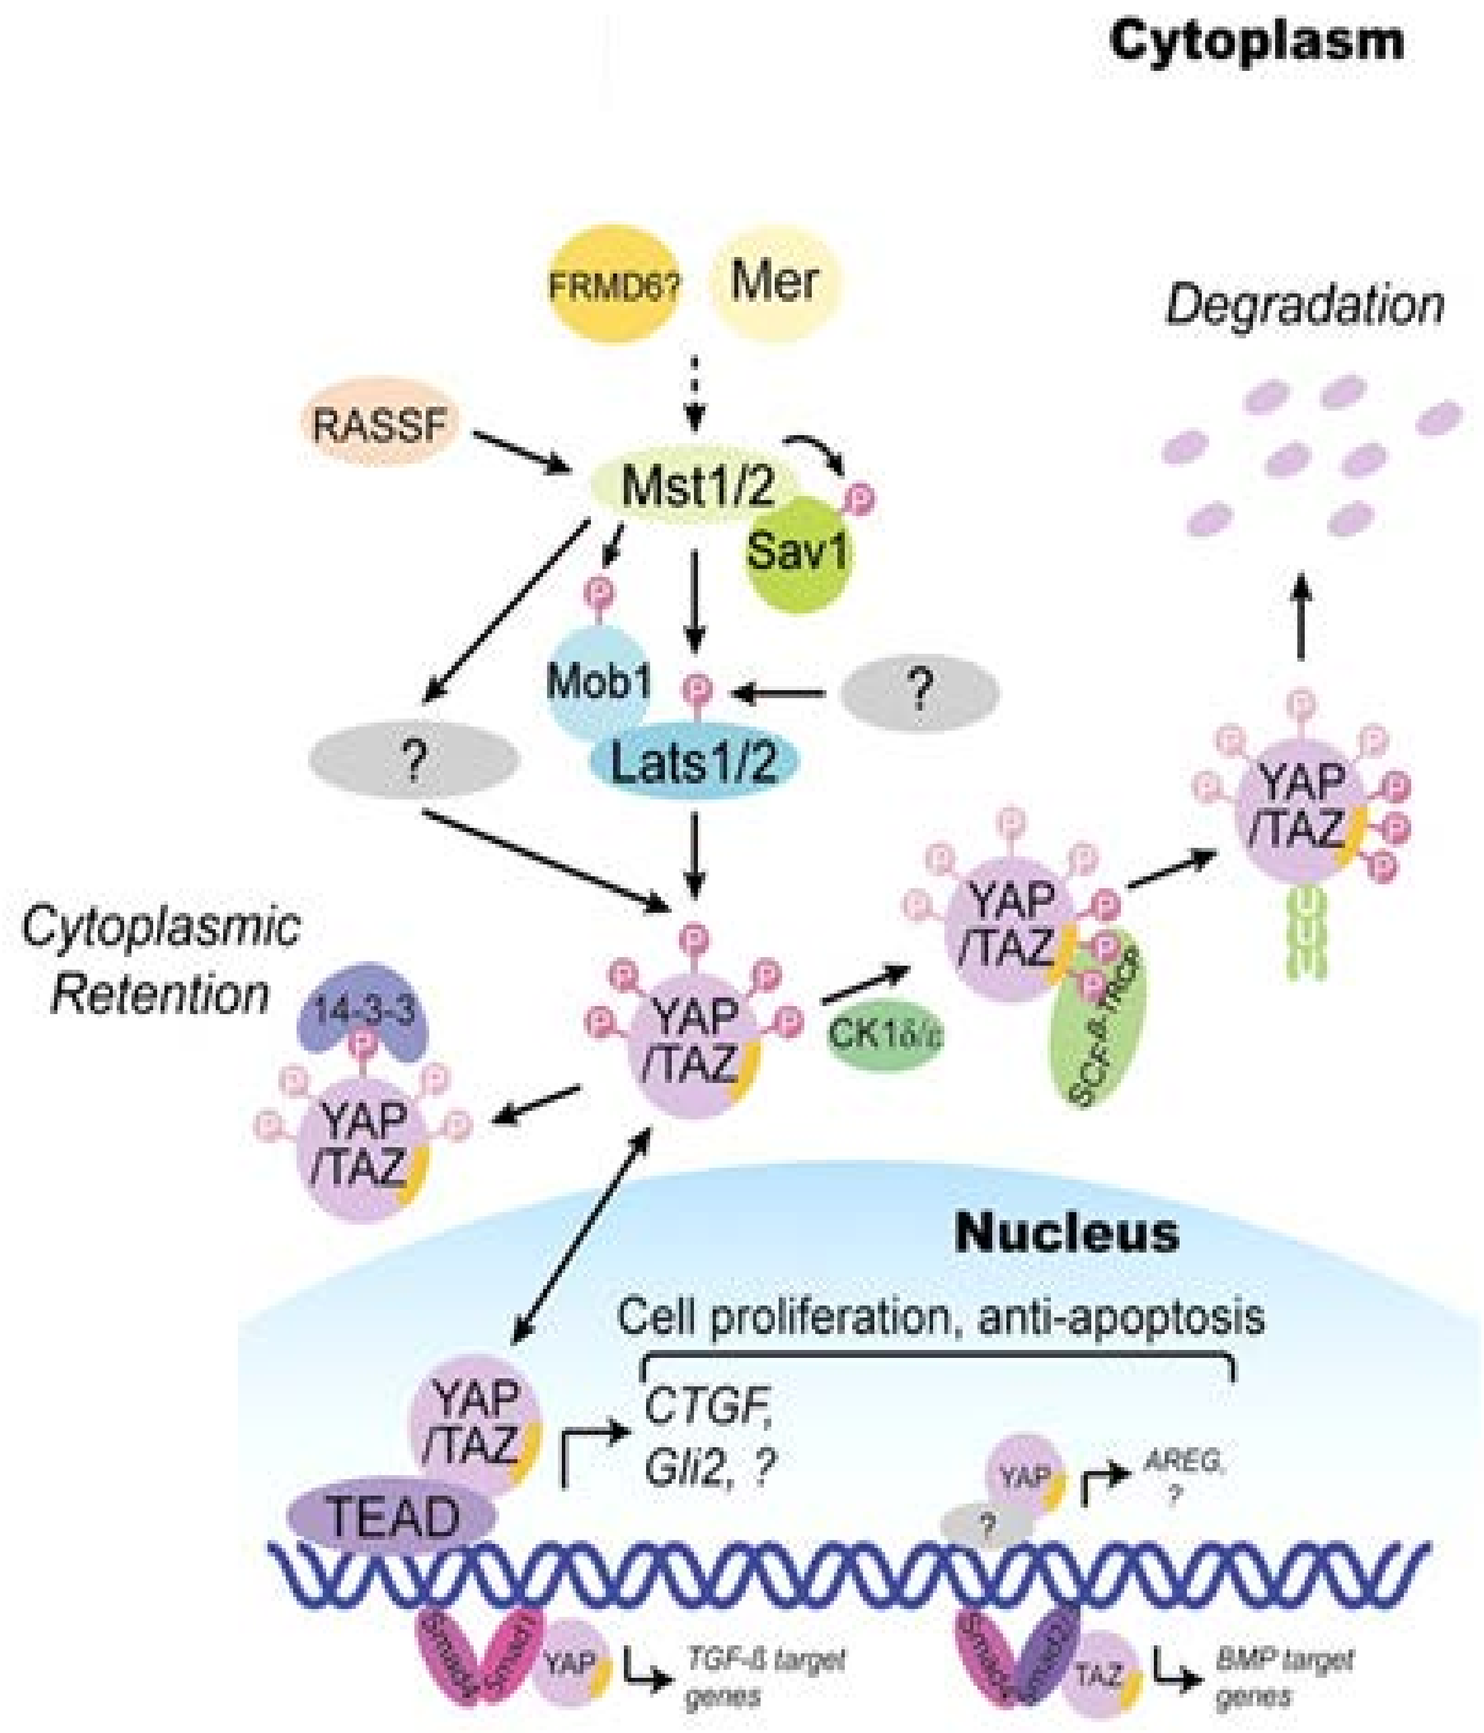
Cytoplasm
FRMD6?
Mer
Degradation
RASSF
Mst1/2
Sav1
Mob1
?
?
Lats1/2
Cytoplasmic Retention
14-3-3
YAP /TAZ
CK1δ/ε
SCF-β-TRCP
Nucleus
Cell proliferation, anti-apoptosis
CTGF, Gli2, ?
TEAD
AREG, ?
TGF-β target genes
BMP target genes

Our strategy: To block the YAP-TEAD interaction with a small molecule

YAP-TEAD A DRUGGABLE INTERACTION

- YAP-TEAD PPI has 3 interfaces
- YAP: IDP (Intrinsically Disordered Protein)
 - at least by sequence composition
 - YAP is stabilized by PPI with TEAD
- TEAD: globular protein
 - Hot Spot analysis by Ala-scan

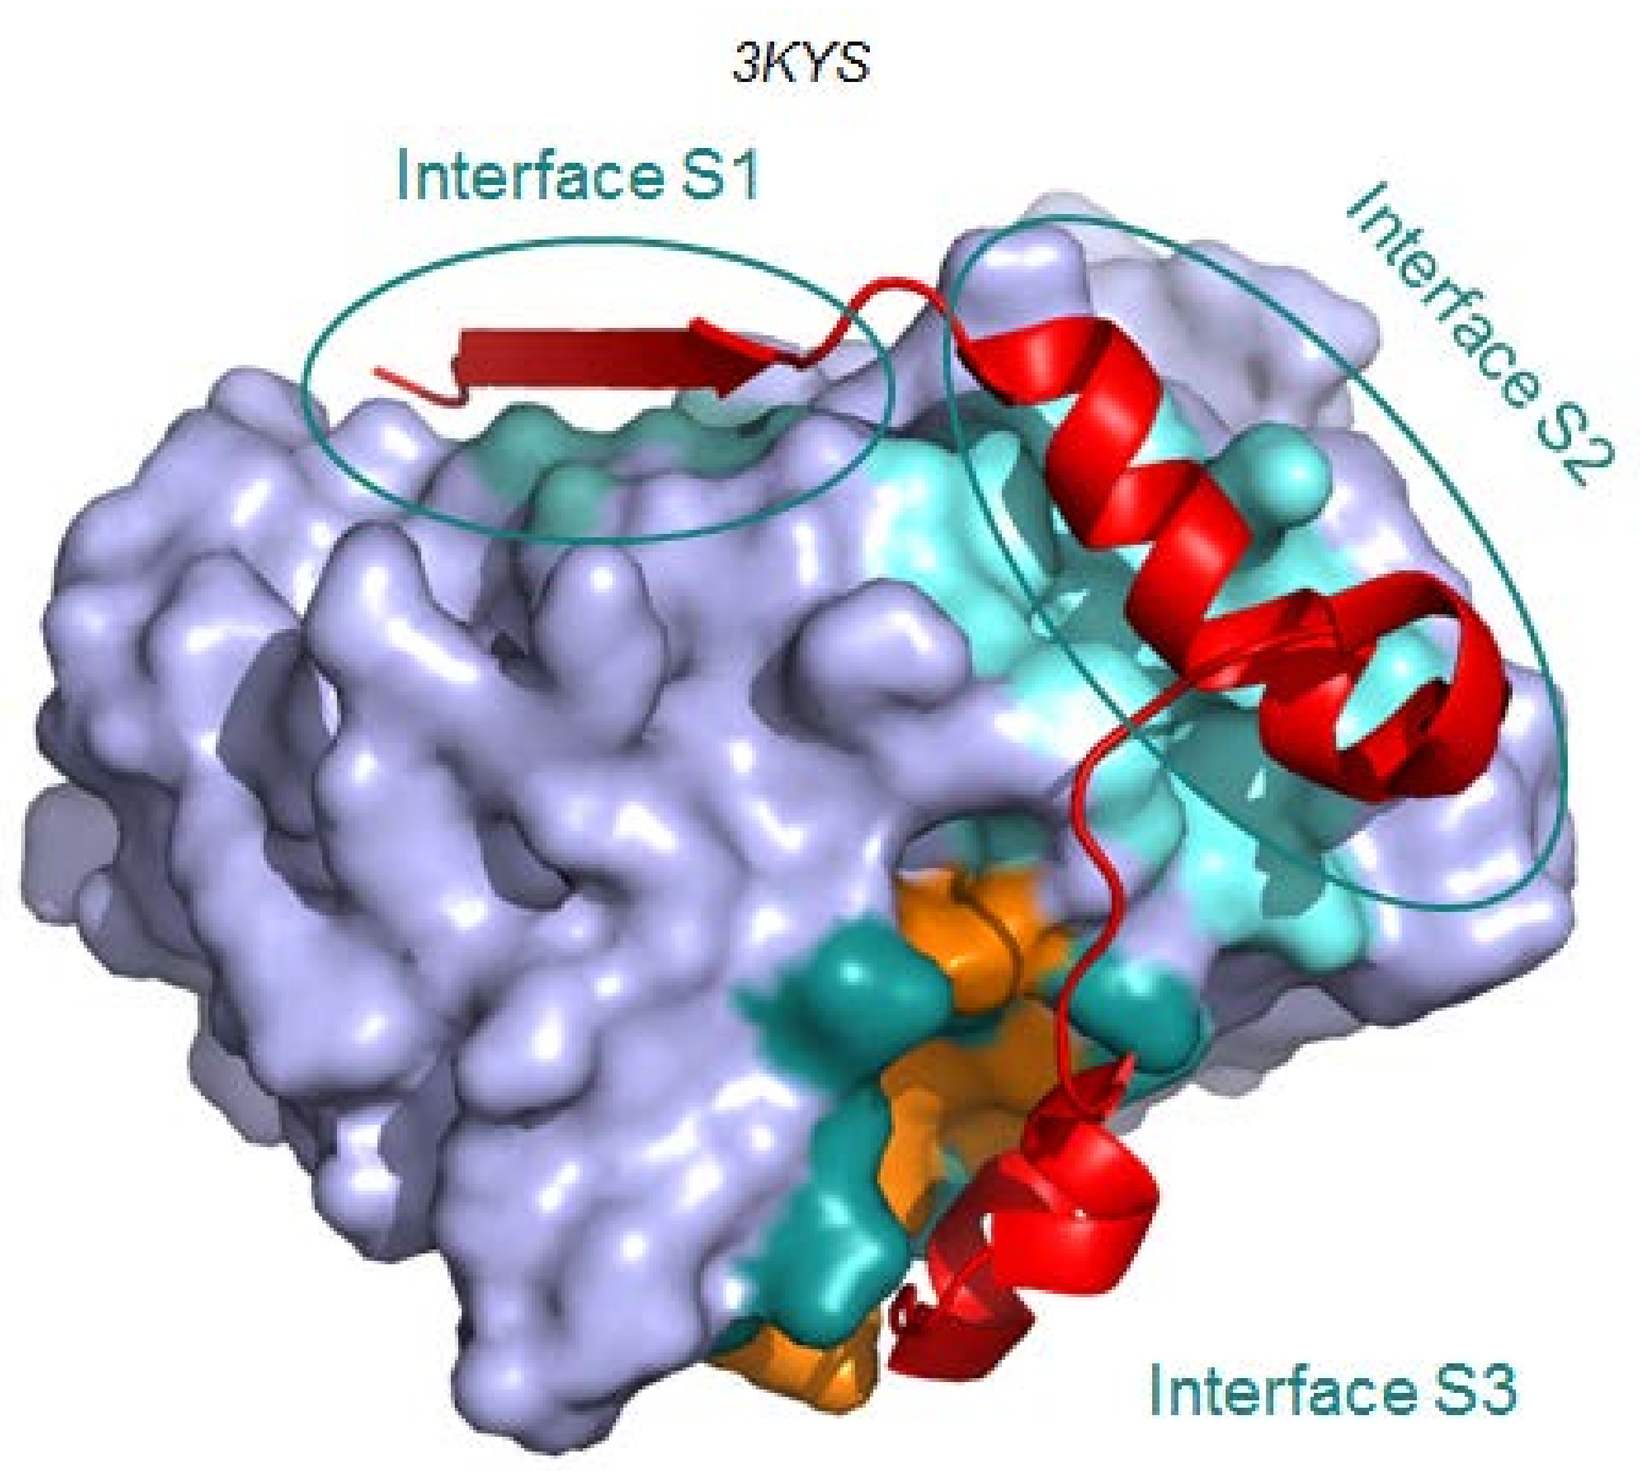
3KYS
Interface S1
Interface S2
Interface S3

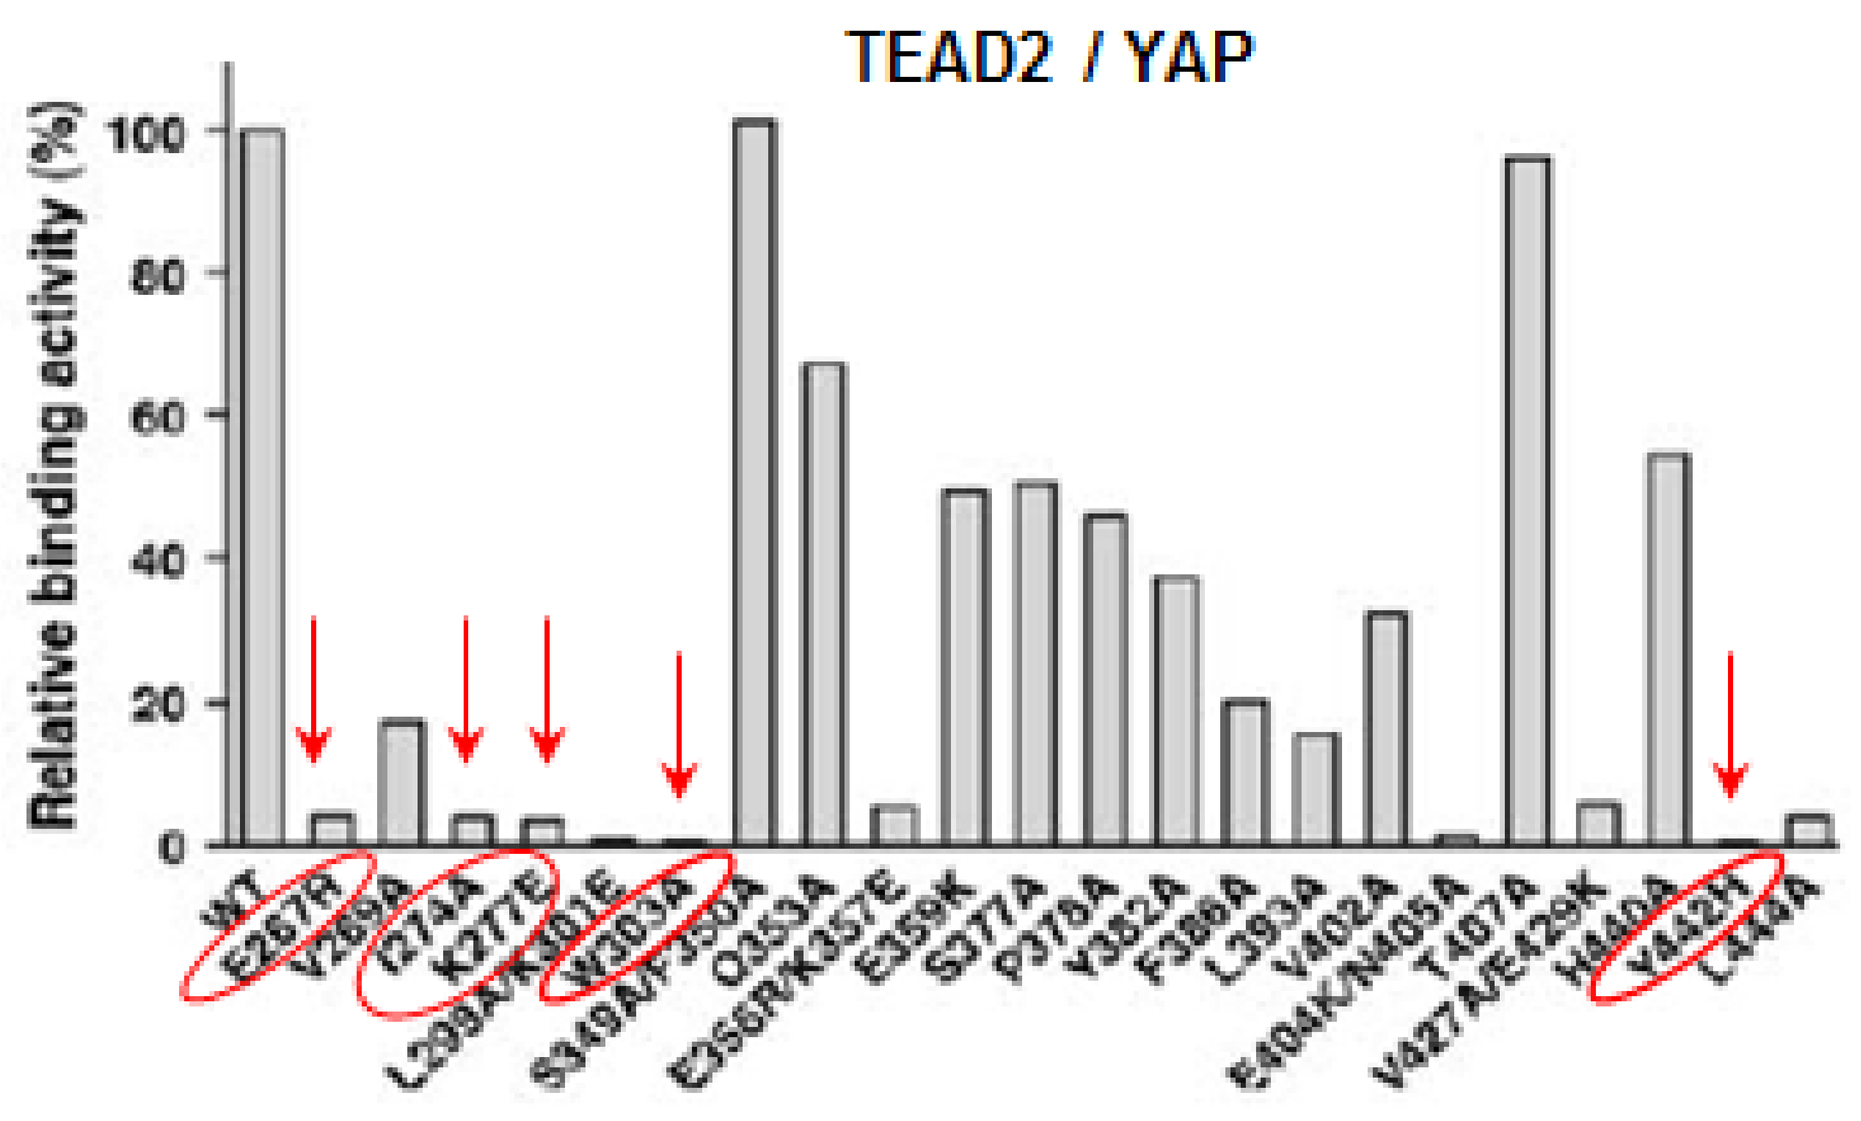
TEAD2 / YAP
Relative binding activity (%)
WT
E267R
V269A
I274A
K277E
L299A/K301E
W303A
S349A/F350A
Q353A
E256R/K357E
E359K
S377A
P378A
Y382A
F386A
L393A
V402A
E404K/N405A
T407A
V427A/E429K
H440A
Y442H
L444A

W. Tian et al, PNAS, April 2010, 107(16), 7293

All critical residues for YAP-TEAD interaction belong to interface 3

Discover interface S3-TEAD binders to inhibit YAP-TEAD interaction

RESULTS

HIT identification

A Dual FBDD and HTS Screening Approach

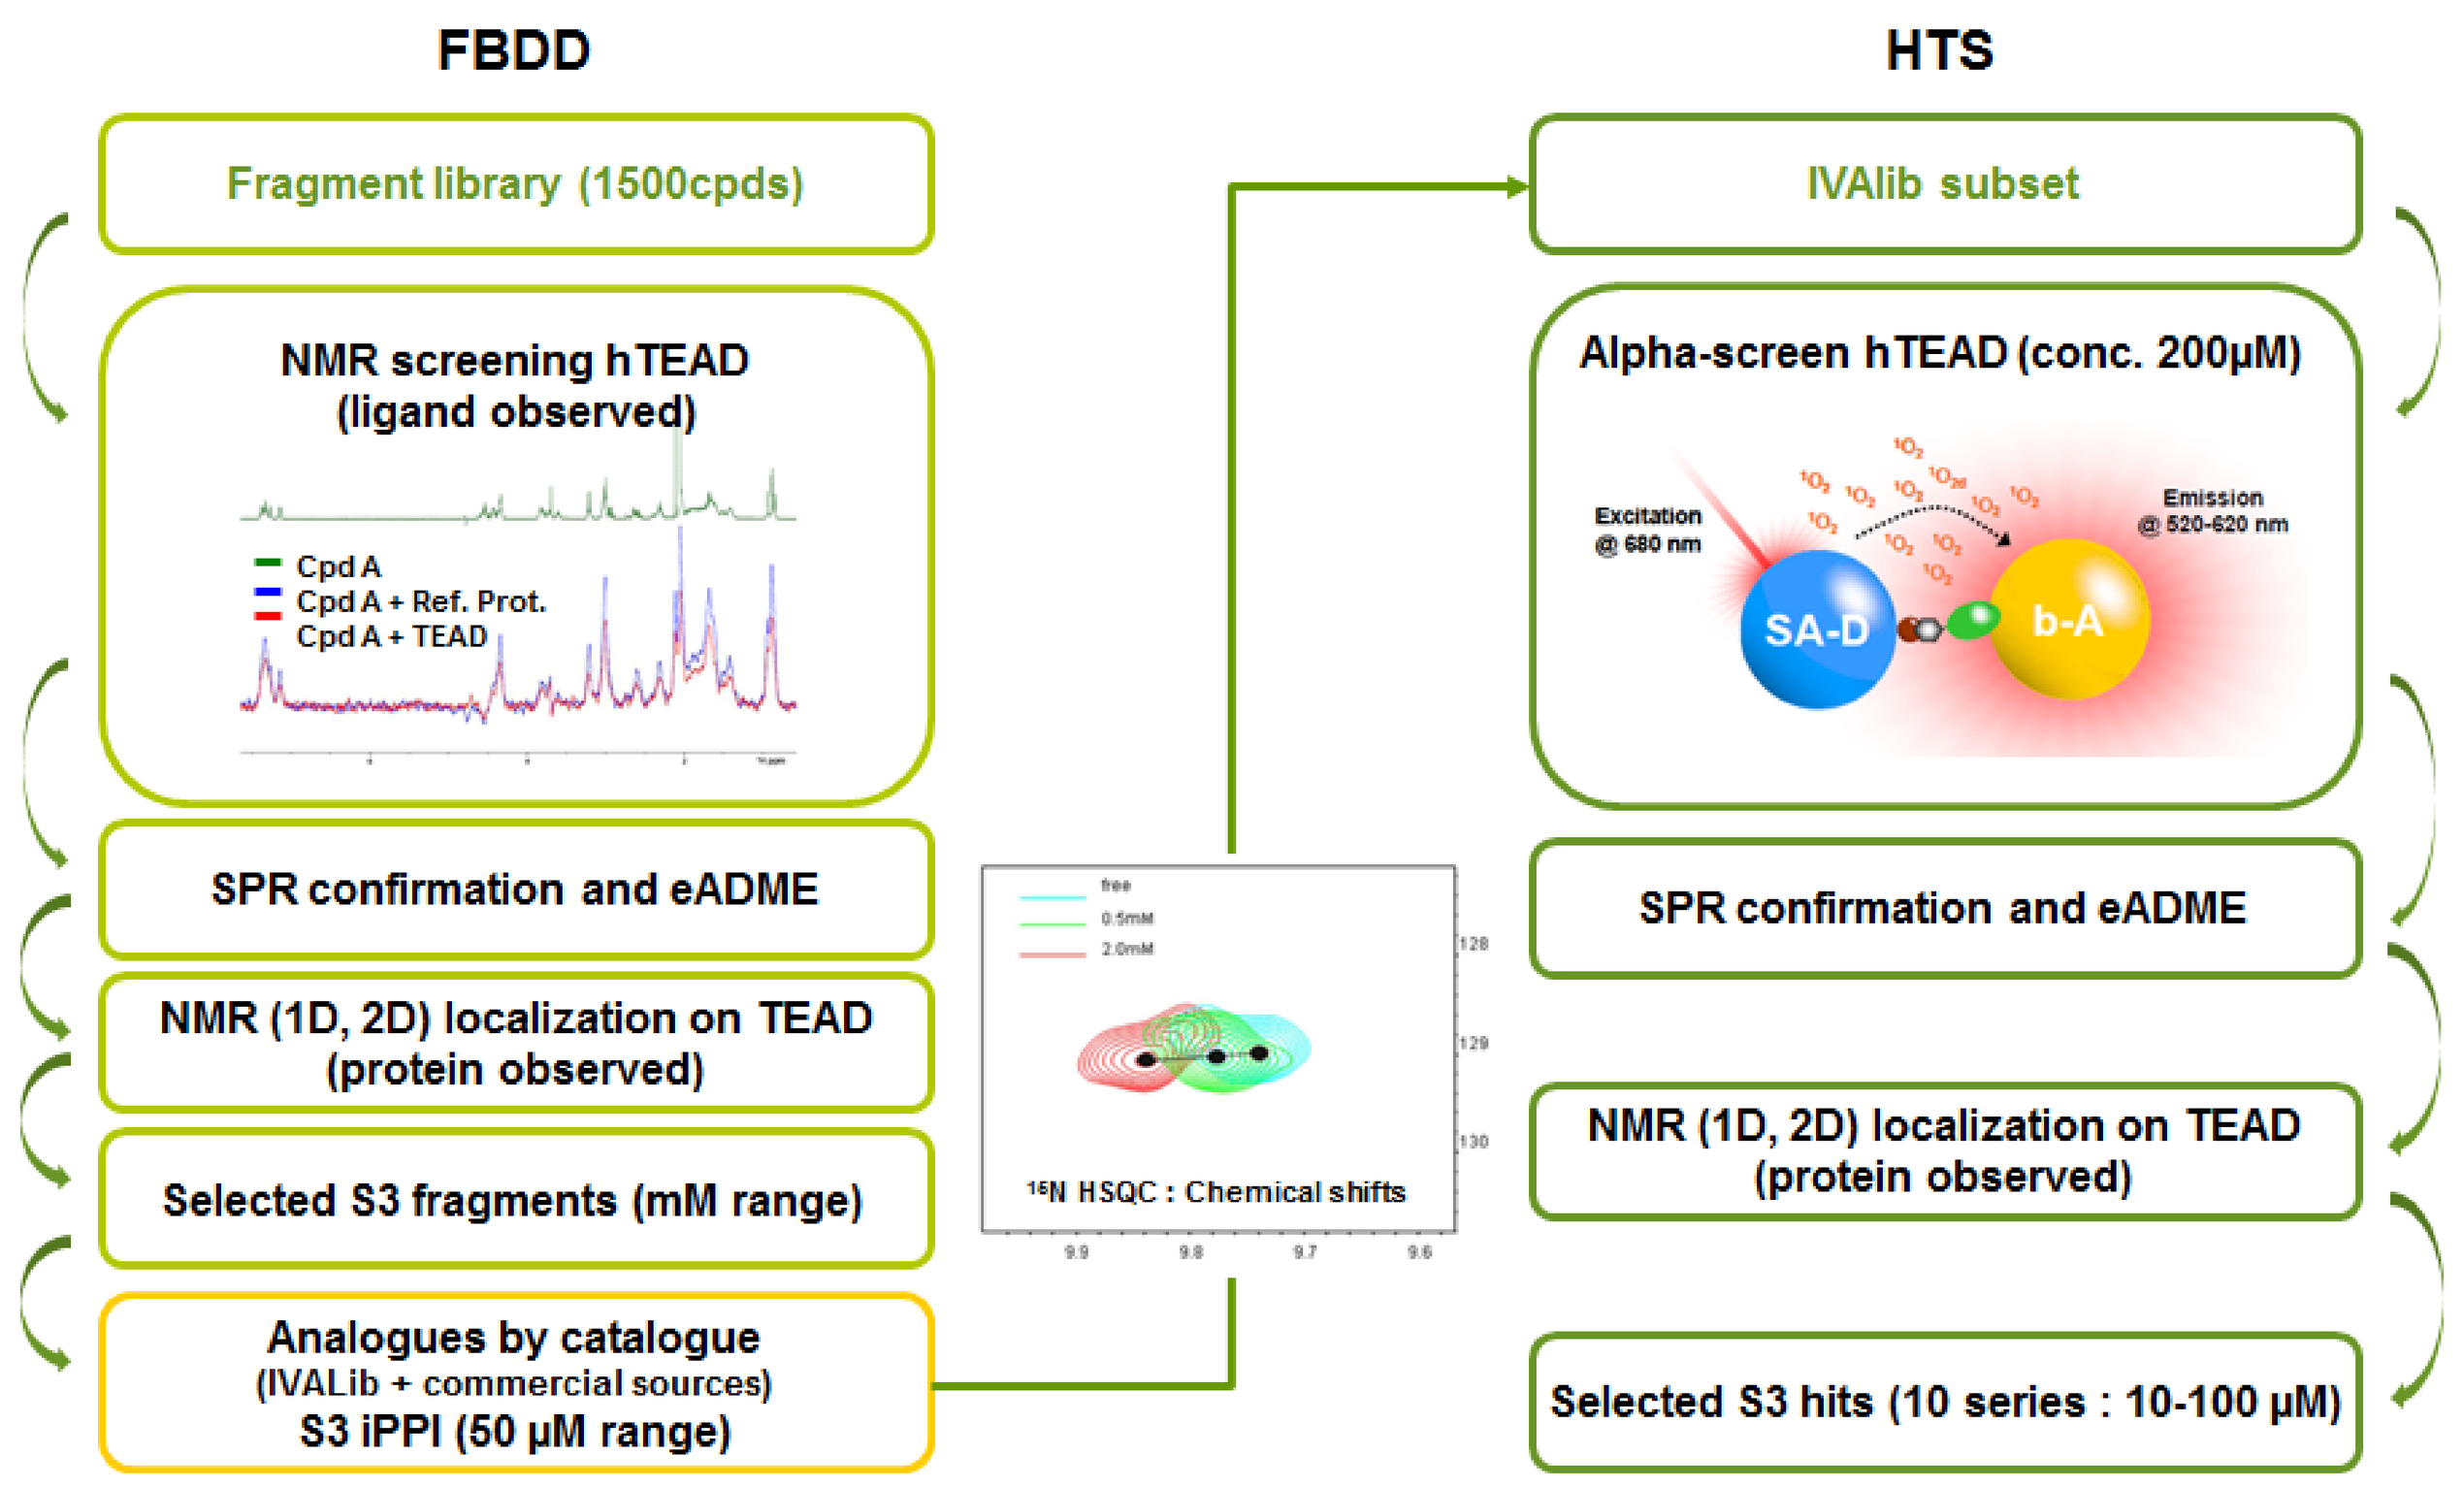
FBDD
HTS
Fragment library (1500cpds)
NMR screening hTEAD (ligand observed)
Cpd A
Cpd A + Ref. Prot.
Cpd A + TEAD
SPR confirmation and eADME
NMR (1D, 2D) localization on TEAD (protein observed)
Selected S3 fragments (mM range)
Analogues by catalogue (IVALib + commercial sources) S3 iPPI (50 µM range)
¹⁵N HSQC : Chemical shifts
IVAlib subset
Alpha-screen hTEAD (conc. 200µM)
Excitation @ 680 nm
Emission @ 520-620 nm
SA-D
b-A
SPR confirmation and eADME
NMR (1D, 2D) localization on TEAD (protein observed)
Selected S3 hits (10 series : 10-100 µM)

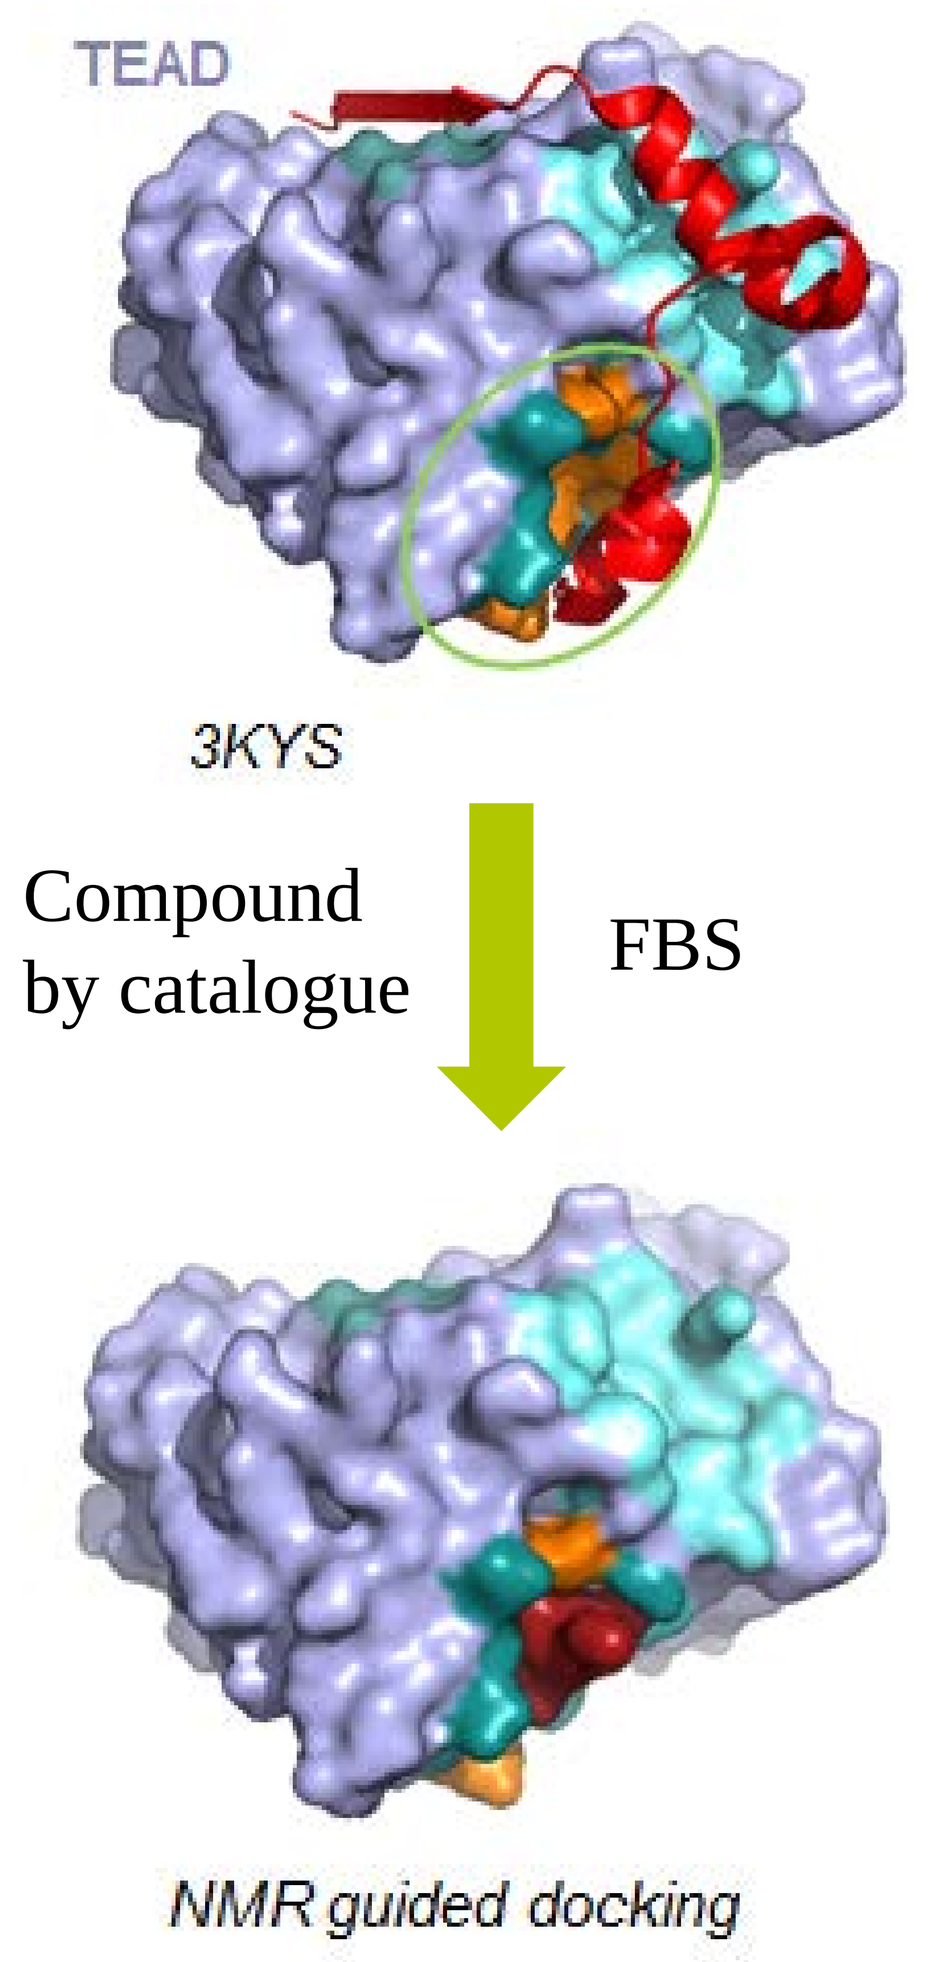
TEAD
3KYS
Compound by catalogue
FBS
NMR guided docking

- Drugability assessment:
 - FBS by NMR: Identification of S3 binding fragments (mM range)
 - Compound by Catalogue (IVALib and commercial sources): Identification of S3 binders that were confirmed as PPI inhibitors (µM range)
- HTS by Apha-Screen: Identified multiple YAP-TEAD iPPI series confirmed to bind at S3 (RMN and SPR)

Multiple series undergoing H2L program

Rapid First Round of Optimization µM to sub µM IC_{50}

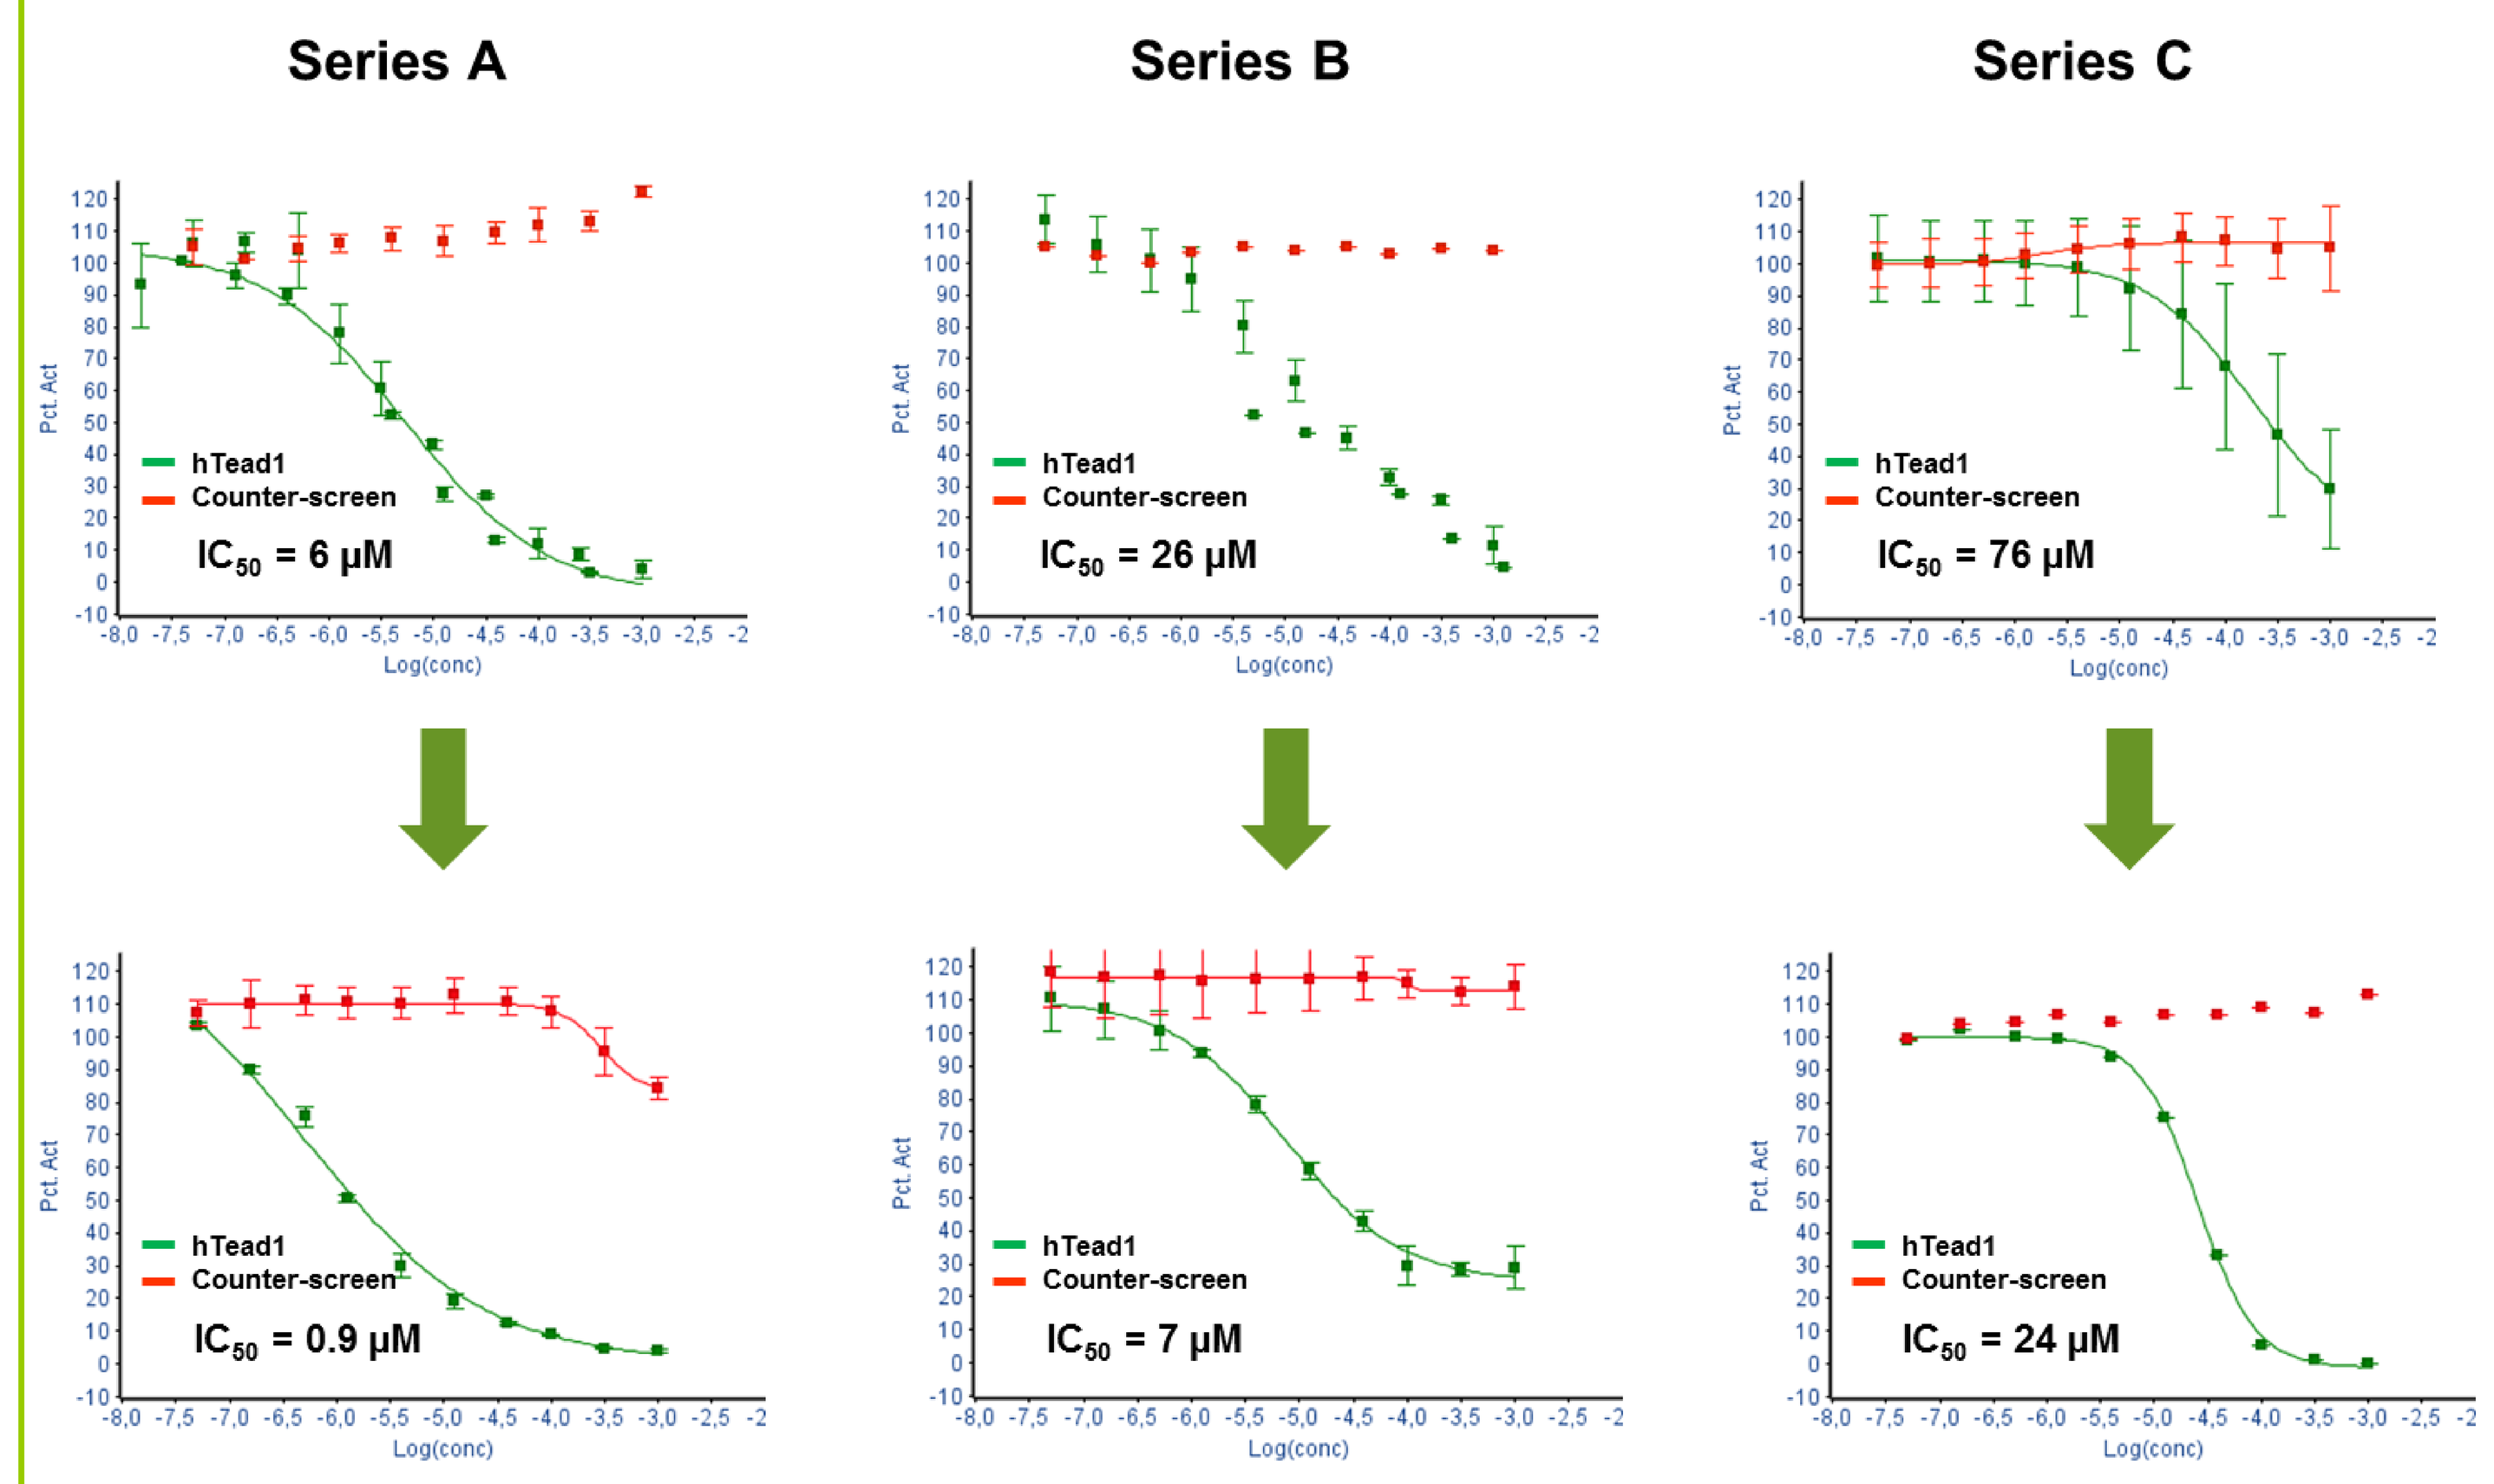
Series A
Series B
Series C
hTead1
Counter-screen
IC50 = 6 µM
IC50 = 26 µM
IC50 = 76 µM
IC50 = 0.9 µM
IC50 = 7 µM
IC50 = 24 µM
Pct Act
Log(conc)

TEAD ligands	✓	• SPR, KD : 60µM - 3mM • LE : 0.17 – 0.27
YAP – TEAD inhibitors	✓	• Alpha-screen : IC50 : 0.9µM - 10µM with max > 80%
TEAD S3 binders	✓	• NMR HSQC localization studies
Lipinski rules	✓	• Mw : 213 – 500 • HA : 0 – 7 • HD : 0 – 2 • LogP : 0.6 – 4 • PSA : 15 -150
Early ADME	✓	• Solubility > 100µM • Metabolic stability < 21µl/min/mg protein • Permeability moderate to high on Caco2
SAR	✓	• Emerging trends

CONCLUSIONS

- We have been able to demonstrate TEAD S3 drugabbility
- We have identified multiple YAP-TEAD iPPI series confirmed to bind at S3 (RMN and SPR)
- Three series have been selected for hit to lead phase, and optimization µM to sub µM IC_{50}