Treatment of Mucopolysaccharidosis type VI patients with odiparcil alone or in addition to enzyme replacement therapy: a phase IIa study

inventiva

50 rue de Dijon, 21121 Daix, France
(www.inventivapharma.com)

Nathalie Guffon[a], Mireille Tallandier[b], Derralynn Hughes[c], Julia Hennermann[d], Elisa Leão Teles[e], Philippe Huot-Marchand[b], Anne Fiquet[b], Chris Hendriksz[f]

[a]Centre de Référence des Maladies Héréditaires du Métabolisme, HCL, Lyon,France; [b]Inventiva, Daix, France; [c]Royal Free Hospital, London, United Kingdom; [d]Villa Metabolica, Mainz, Germany; [e]Centro Hospitalar Universitário de São João, Porto, Portugal; [f]FYMCA, University of Pretoria, Manchester, United Kingdom and I would suggest: fFYMCA Medical , Manchester, United Kingdom h Steve Biko Academic Unit, University of Pretoria, South Africa.

1–INTRODUCTION

Background: Current treatments for mucopolysaccharidosis type VI (MPS VI) patients have limited effects on some organ systems, including ophthalmological, cardiovascular, respiratory and skeletal. In MPS VI animal models odiparcil reduces glycosaminoglycans (GAG) storage in various tissues including liver, but also hard-to-reach tissues such as cornea and joints of the animals.

Objective: Assess the safety of odiparcil and identify the early signals of efficacy and pharmacodynamic response in patients with MPS VI.

Endpoints included :

- Safety
- Efficacy assessments
- Pharmacokinetics profile
- Levels of GAG in urine and leukocytes

2–STUDY PLAN

Figure 1: Study plan

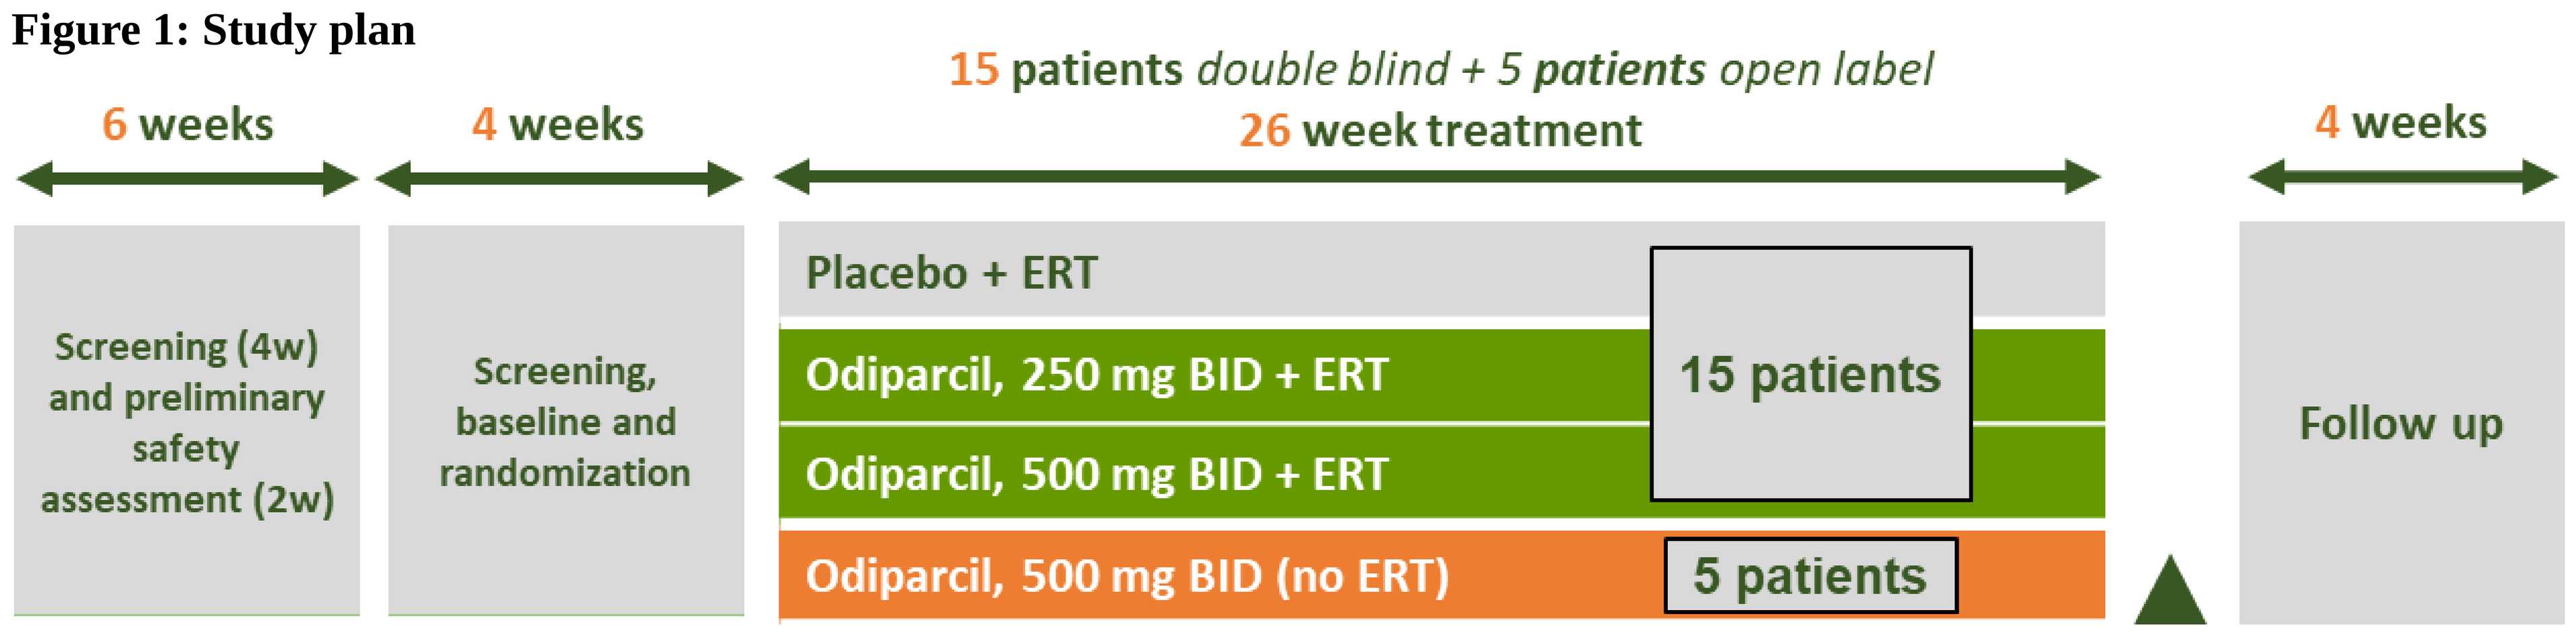

15 patients double blind + 5 patients open label
26 week treatment
6 weeks
4 weeks
4 weeks
Screening (4w) and preliminary safety assessment (2w)
Screening, baseline and randomization
Placebo + ERT
Odiparcil, 250 mg BID + ERT
Odiparcil, 500 mg BID + ERT
15 patients
Odiparcil, 500 mg BID (no ERT)
5 patients
Follow up
End of treatment

BID: twice a day; ERT: enzyme replacement therapy; w: week

3–STUDY CONDUCT

The 26-week multicenter clinical study (**iMProveS study**) was conducted in 4 sites in Europe (France, Germany, Portugal and United Kingdom) and head-line results were released mid-December-2019.

In the core phase of the study, 20 patients with MPS VI, 16 to 64 years of age were included in 2 cohorts (Figure 1):

- **Enzyme replacement therapy (ERT) cohort**: 15 patients treated with ERT were blindly randomized 1:1:1 to receive oral odiparcil at 250mg twice a day (BID), or 500mg BID or placebo, in addition to ERT
- **Non-ERT cohort**: 5 ERT-naïve patients received oral odiparcil at 500mg BID

Thirteen patients completed the 26-week study treatment, 10 in the ERT cohort (4 in the placebo group and 3 patients each in the odiparcil groups), and 3 in the non-ERT cohort. The remaining 7 patients terminated prematurely the study treatment.

4–DEMOGRAPHICS

- Gender was balanced between groups
- The median age varied from 18 to 28 years, however the patients in the placebo group were notably younger with a median age of 18 years versus 24 to 28 years in all odiparcil groups
- The median age at MPS diagnosis varied from 1 to 17 years old, notably MPS VI was diagnosed late in the non-ERT cohort (up to 34 years old). In the ERT cohort, all patients were diagnosed before the age of 10 except 1 patient diagnosed at 59 years old in the odiparcil 250mg BID
- The median duration of ERT ranged from 12 to 14 years

5–SAFETY RESULTS

- The clinical study **met its safety primary** endpoint further supporting the good overall safety profile of odiparcil already observed in previous Phase I and Phase II clinical studies (Table 1)
- Compared to previous Phase I and II clinical studies conducted with odiparcil for the prevention of thrombosis, **no new safety findings were observed**
- Most treatment-related adverse events (AEs) occurred in the odiparcil groups and were mild in intensity
- Three patients in the odiparcil groups of the ERT cohort reported a treatment-related serious AE (SAE):
 - *Proteinuria* further qualified as laboratory false positive in 1 patient in each odiparcil dose group
 - *Rash* (skin exanthema) that recovered, which was the severe treatment-related AE in the odiparcil 250mg BID group
- Seven patients terminated prematurely the study treatment:
 - One for patient's decision
 - One death for *bronchopneumopathy* (not related) occurred in the placebo group
 - Five withdrawals occurred in the odiparcil groups: 3 patients for a treatment-related SAE (see above), and 2 patients for a treatment-related non-serious AE (*dermatitis allergic and palpitations*)

Table 1: Number of patients with at least one treatment-related adverse event occurring from first to last dose of the study treatment in the core phase of the study

	Double-blind (ERT)			Open-label (non-ERT)
	Placebo N=5	Odiparcil 250mg BID N=5	Odiparcil 500mg BID N=5	Odiparcil 500mg BID N=5
At least 1 relAE	1	5	5	4
Serious relAE	0	2	1	0
Severe relAE	0	1	0	0
Moderate relAE	0	3	1	1
Mild relAE	1	5	5	4

BID: twice a day; ERT: enzyme replacement therapy; relAE: treatment-related adverse event

6–PHARMACOKINETICS PROFILE

- Odiparcil exposures correlated with dose per kg actually received by patients and were the expected ones
- There was no impact of ERT on odiparcil exposure in plasma

7–LEVELS OF GAG IN URINE AND LEUKOCYTES

A dose-dependent urinary GAG clearance, used as an activity biomarker, was clearly demonstrated in the entire patient population treated with odiparcil, consistent with odiparcil's mechanism of action of GAG clearance (see Figure 2)

GAG in leukocytes were not confirmed as biomarkers for decrease of GAG accumulation, in this study, similar to ERT.

Figure 2: Total urinary GAG levels

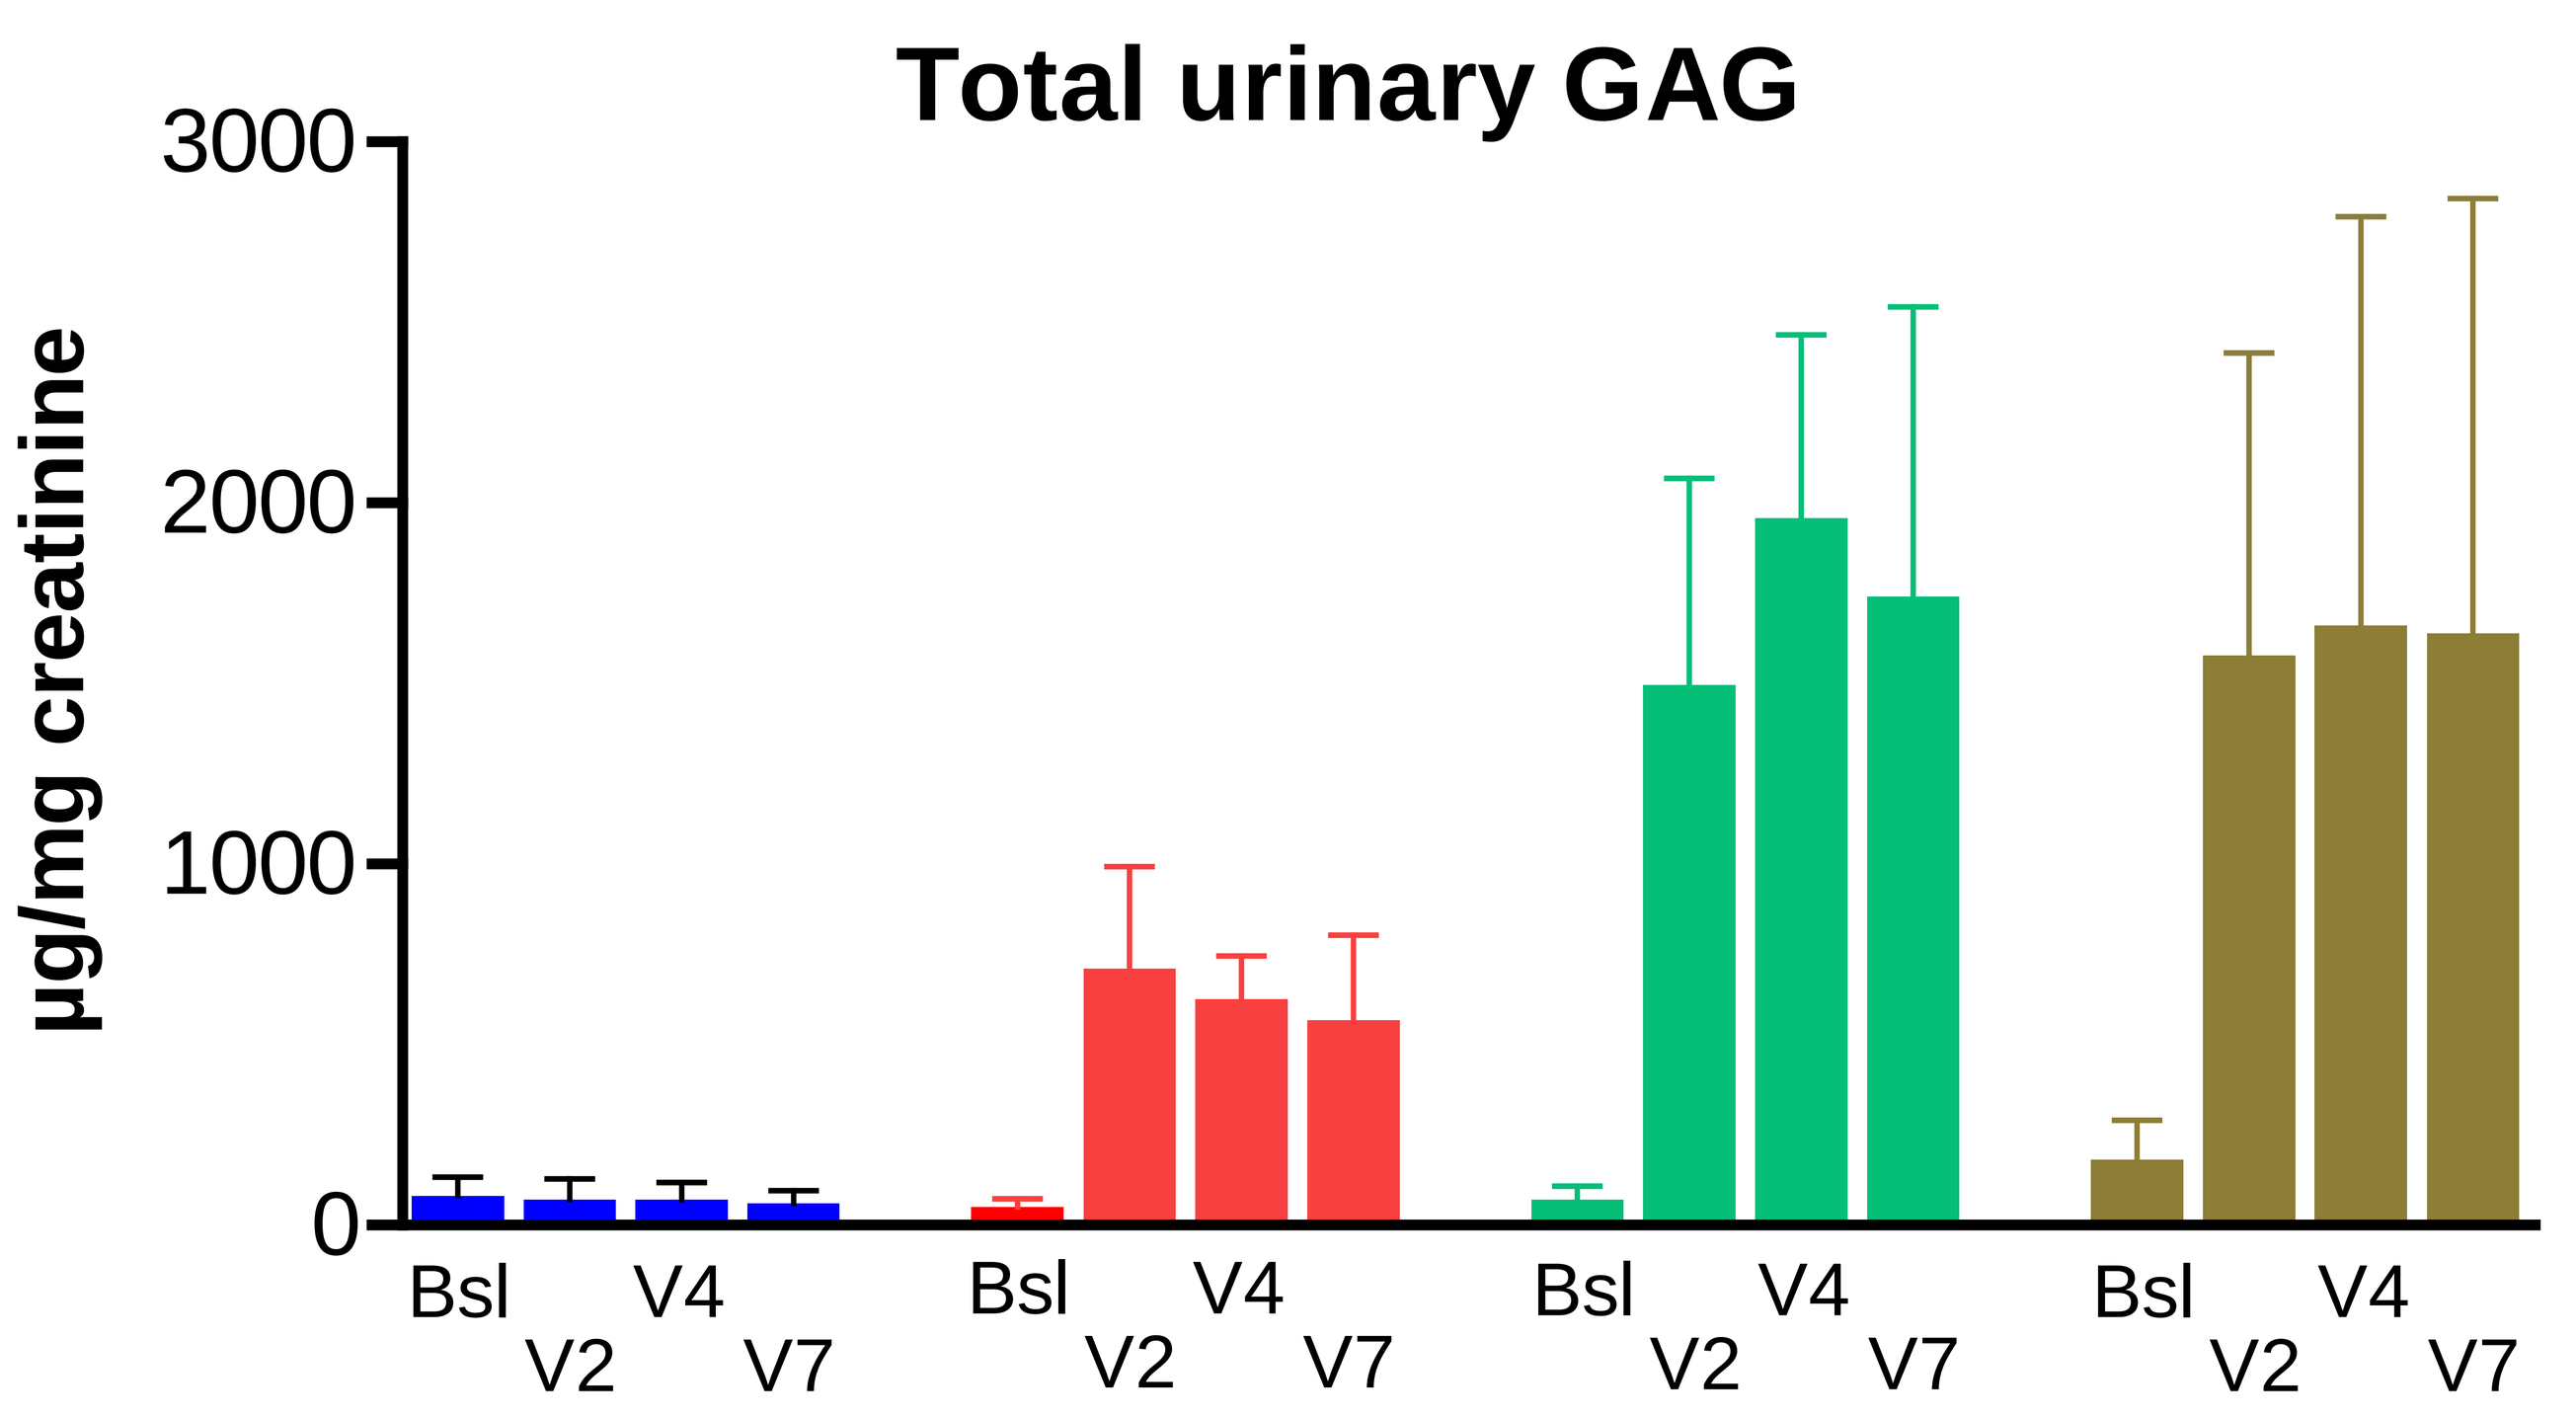

Total urinary GAG
µg/mg creatinine
3000
2000
1000
0
Bsl V2 V4 V7
Bsl V2 V4 V7
Bsl V2 V4 V7
Bsl V2 V4 V7

Bsl: baseline, V2, V4, V7: visits 2, 4, 7 (1-, 3-, 6-month treatment, respectively). ■ ERT-cohort placebo group, ■ ERT-cohort Odiparcil 250mg BID group, ■ ERT-cohort Odiparcil 500mg BID group, ■ non-ERT-cohort 500mg BID group.

8–EFFICACY RESULTS

Efficacy data measured in the locomotor, respiratory, cardiovascular, ophthalmology and audiology organ systems, and pain were analysed descriptively as well as reviewed blindly by Experts in cardiology, ophthalmology and audiology, and by the Trial Steering Committee (TSC).

Patient's responses were categorized by the Experts using a 5-point scale (improved, slightly improved, stable, slightly worsened or worsened) or non-assessable when no evaluation was possible due to lack of data, based on the blinded review of the patient's medical history, disease profile, and both absolute and relative changes of each measurement at 26 weeks compared to their screening/baseline value.

For the locomotor and respiratory organ systems, and pain, the TSC predefined thresholds to categorize patient's responses.

Improvements (improved/slightly improved) were observed in patients treated with odiparcil in addition to ERT with regards to corneal clouding as well as cardiovascular and respiratory functions (Table 2)

Table 2: Improvements in the Respiratory, Ophthalmology and Cardiovascular organ systems in the patients from the ERT cohort that completed the 26-week study treatment

Double-blind (ERT)	Organ system		
	Respiratory (FVC)	Ophthalmology (COM)	Cardiovascular (CIMT, echocardiogram)
Placebo (N=4)	0	1 (slightly improved)	1 (slightly improved)
Odiparcil 250mg or 500mg BID (N=6)	3 (slightly improved)	2 (improved)	4 (3 slightly improved + 1 improved)

BID: twice a day; ERT: enzyme replacement therapy; relAE: treatment-related adverse event

- Odiparcil led to improvement of respiratory function in 3 patients, but none in the placebo group
- The 3 patients who improved in 2 or 3 of the respiratory, ophthalmology and cardiovascular organ systems were all in the odiparcil groups of the ERT cohort (N=6):
 - 2 patients treated with odiparcil 250mg BID improved: i) FVC and left ventricular mass index; ii) COM of both eyes and disappearance of a mitral regurgitation
 - 1 patient treated with odiparcil 500mg BID improved for FVC, COM of the controlateral eye to a previously transplanted eye, and had decreased severity of mitral regurgitation.

In the non-ERT odiparcil 500mg BID cohort, 2 of the 3 patients improved for several functions: i) FVC, range of motion of both shoulders and pain; ii) range of motion of both shoulders and pain (data not shown).

Regarding locomotor function, no clear difference was observed among the different patient groups. However, for the 6-minute walk test, the absolute change at 26 weeks compared to screening/baseline value showed a trend in the ERT cohort in favour of odiparcil 500mg BID group compared to placebo (data not shown).

9–CONCLUSION

iMProveS clinical study showed:

- **A good safety profile of odiparcil** administered to 15 patients with MPS VI 16 years of age or older
- **A clear correlation between odiparcil dose received by patients and odiparcil exposure in plasma**
 - No impact of ERT on odiparcil exposure
- **A clear dose-dependent urinary GAGs clearance**, used as an activity biomarker
- **Positive results regarding the efficacy of odiparcil** considering the short study duration of 26 weeks, and the advanced status of the disease and the long ERT duration in patients included in the study

Based on the iMProveS clinical study results, Inventiva has decided to continue the clinical development of odiparcil for the treatment of MPS VI.