A rational approach for the discovery of inhibitors of NSD2 for the treatment of cancer

Claudia Fromond, Xavier Espanel, Anne Soudé, Laurent Chene, Philippe Masson, Benaïssa Boubia, Christian Montalbetti and Pierre Broqua

Inventiva, Daix, France

Abstract # 105

BACKGROUND

The NSD2

- Belongs to the histone-lysine methyltransferase protein family
- Known to dimethylate histone H3 on lysine 36 (H3K36)
- H3K36 dimethylation is most associated with active transcription

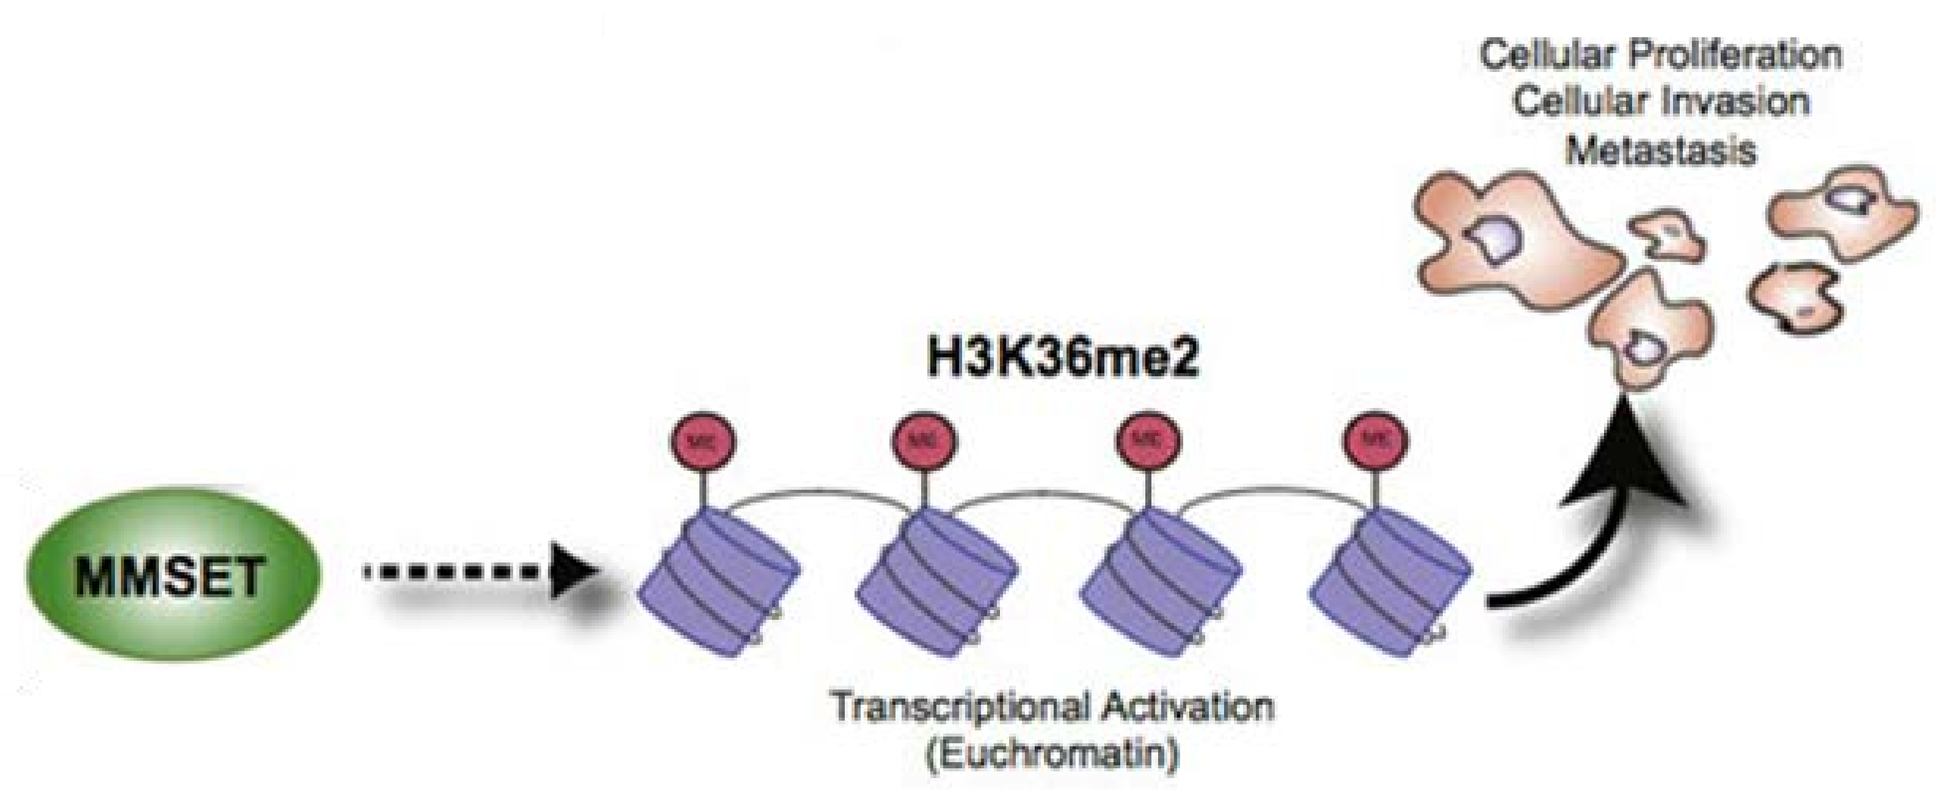
Cellular Proliferation
Cellular Invasion
Metastasis
H3K36me2
MMSET
Transcriptional Activation
(Euchromatin)

Rationale in cancer

- NSD2 triggers the expression of oncogenes, including TGFA, MET, PAK1 and RRAS2
- About 20% of Multiple Myeloma cases are caused by the t(4;14) chromosomal translocation, one result is increased expression of NSD2
- NSD2, harboring a functional SET domain, drives tumorigenesis by increasing H3K36me2, and knockdown of NSD2 leads to regression of multiple myeloma tumors carrying the t(4;14) translocation in mice
- The unmet clinical need and patient population size of t(4;14) multiple myeloma represents a key driver for drug discovery and development of NSD2 inhibitors

ASSAY PRINCIPLE AND VALIDATION

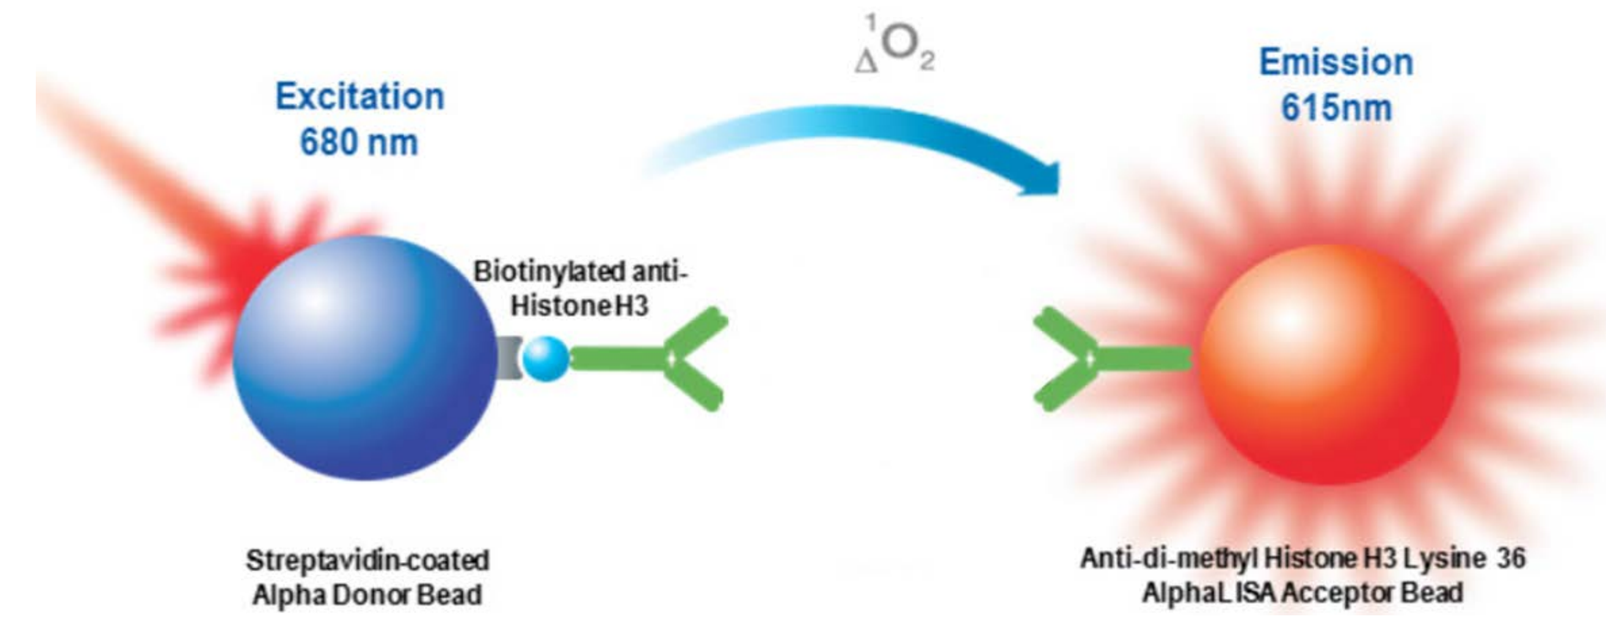
Excitation
680 nm
Emission
615nm
Biotinylated anti-HistoneH3
Streptavidin-coated Alpha Donor Bead
Anti-di-methyl Histone H3 Lysine 36 AlphaLISA Acceptor Bead

The assay is based on AlphaLISA technology and relies on the detection of H3K36me2 marks on nucleosome by a specific antibody

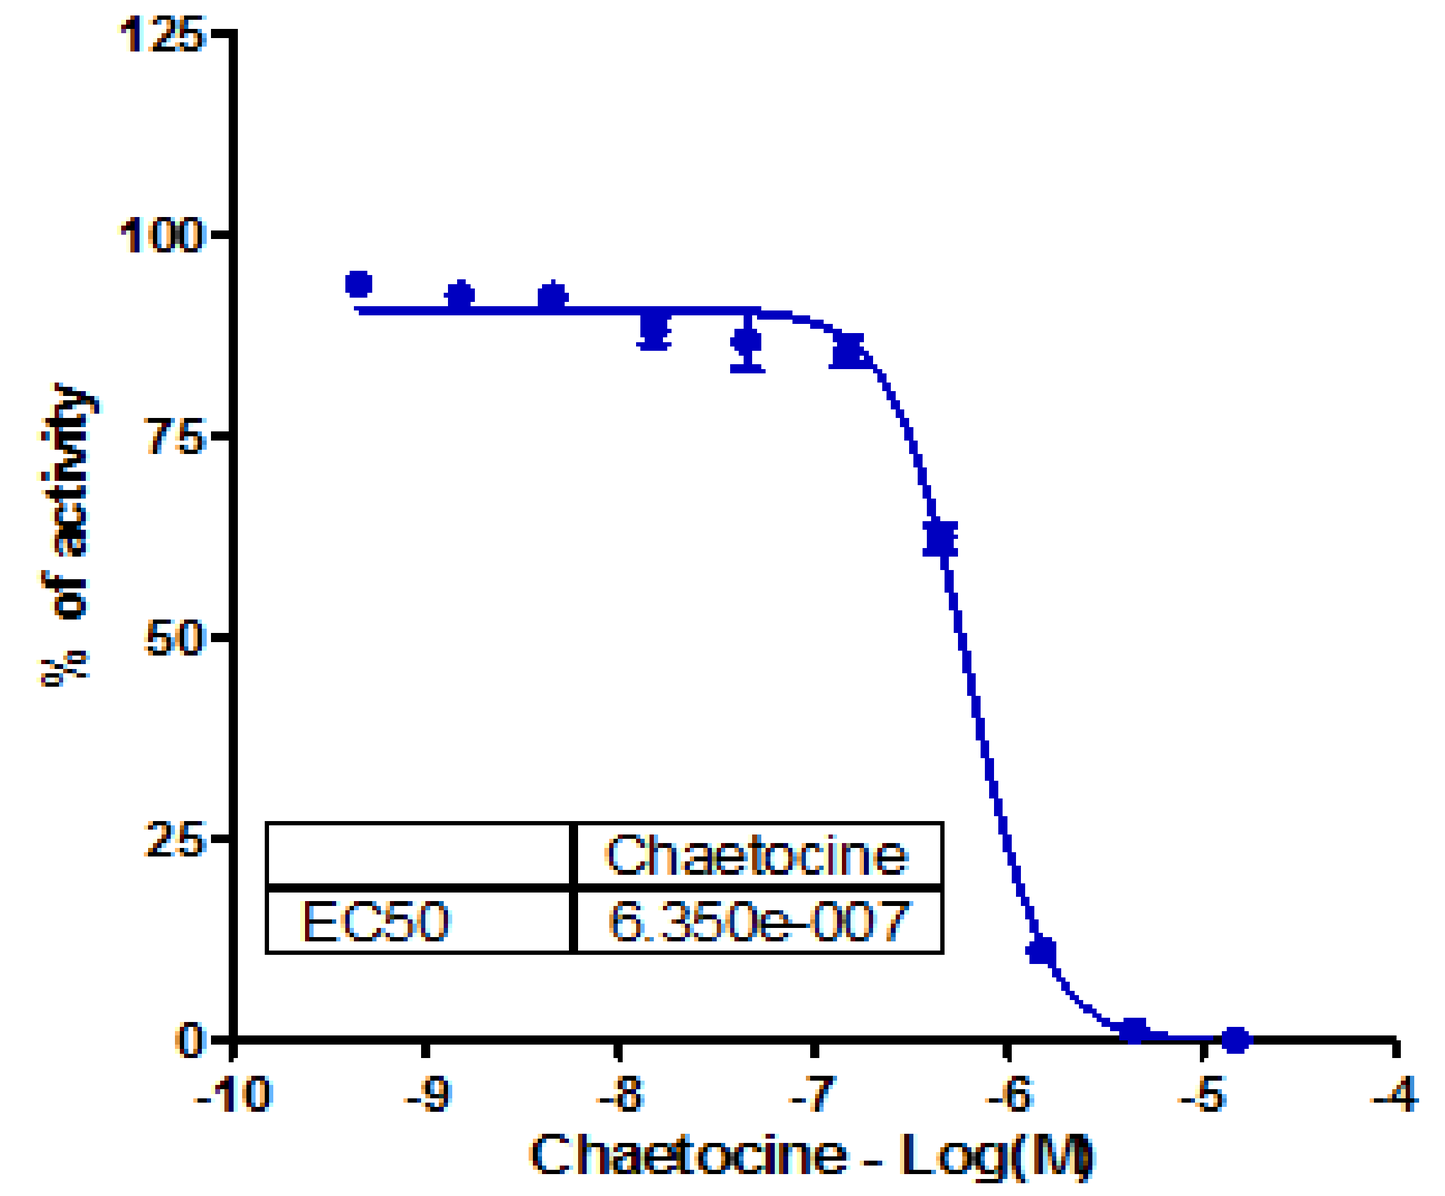
% of activity
Chaetocine - Log(M)
Chaetocine
EC50
6.350e-007

Chaetocin, a non specific inhibitor, is able to block NSD2 activity in a dose dependent manner, with an IC50 of 635 nM, similar to the ones described in the literature

IVALib

- 240,000 Compounds
- IVALib has been designed over years for drug discovery programs
- More than 70% of the compounds are original when compared to Zinc library
- Compounds are available as liquid solutions and 70% as powders, and all the library is stored in controlled environment
- Regular quality controls are performed and a collection enrichment to maintain diversity and originality is in place
- Good hit rate on internal screening programs achieved
- Library available for external drug discovery partnerships

FROM HTS TO VALIDATED HITS

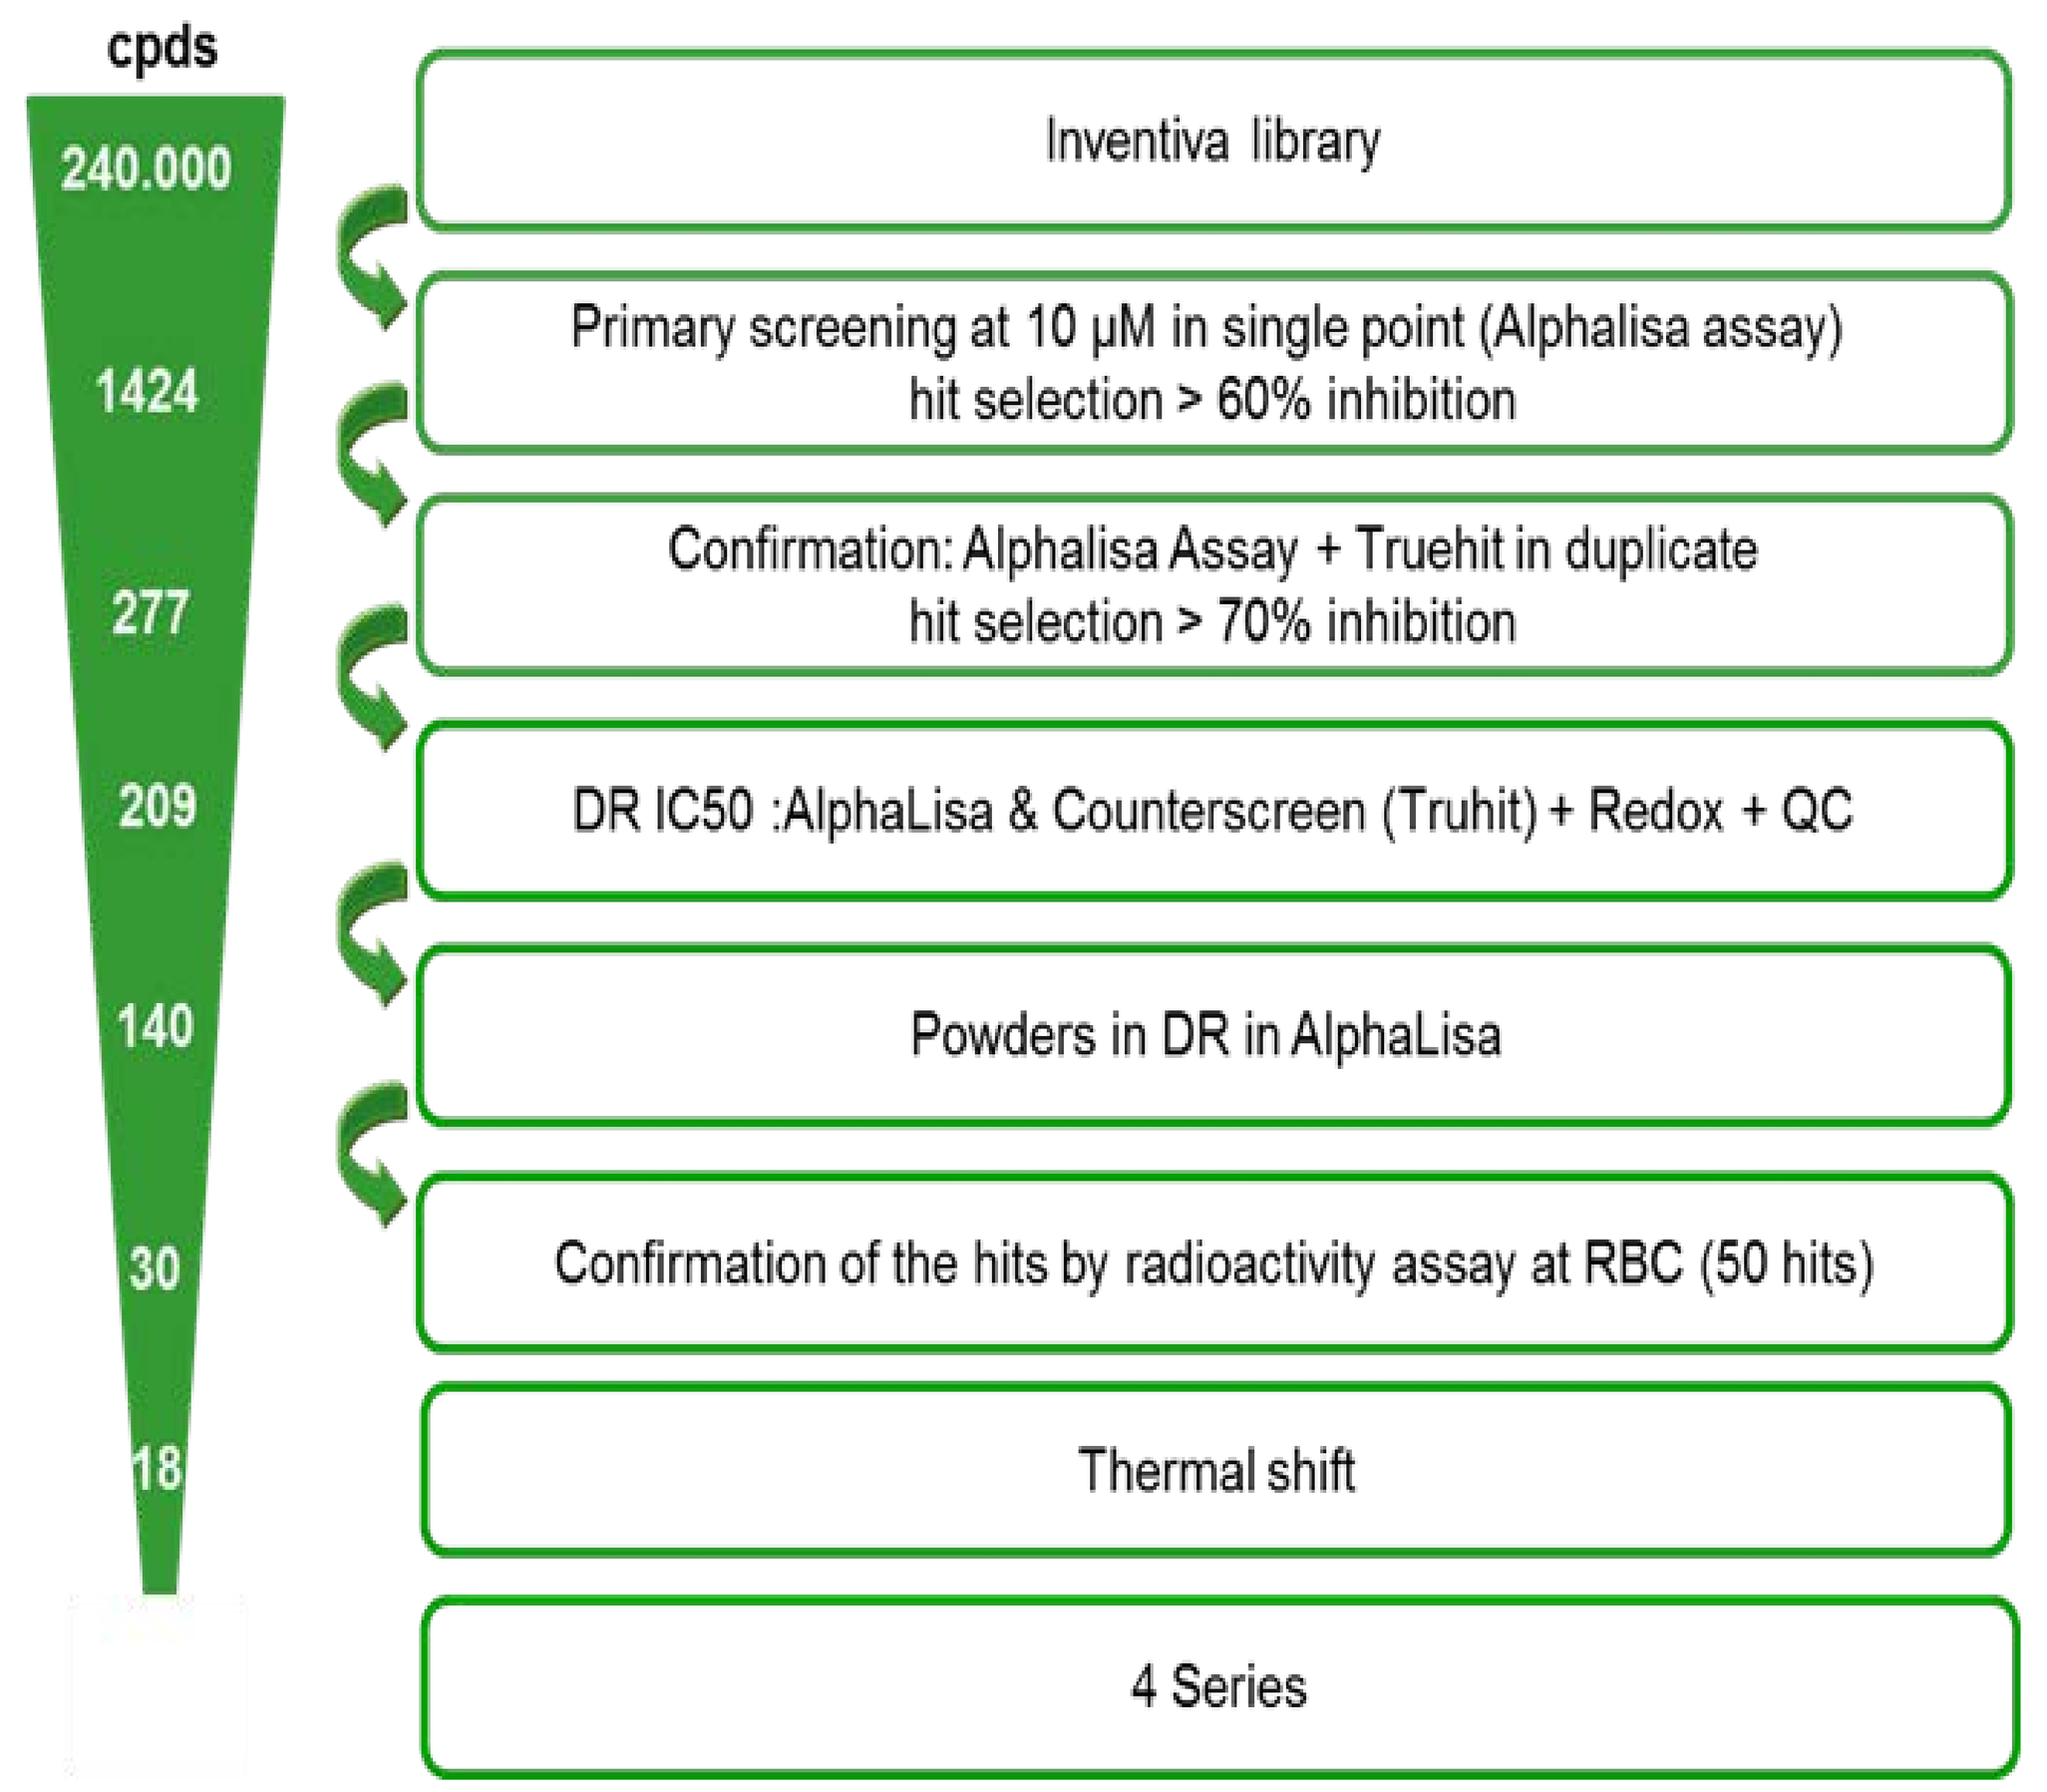
cpds
240.000
1424
277
209
140
30
18
Inventiva library
Primary screening at 10 µM in single point (Alphalisa assay)
hit selection > 60% inhibition
Confirmation: Alphalisa Assay + Truehit in duplicate
hit selection > 70% inhibition
DR IC50 :AlphaLisa & Counterscreen (Truhit) + Redox + QC
Powders in DR in AlphaLisa
Confirmation of the hits by radioactivity assay at RBC (50 hits)
Thermal shift
4 Series

Examples of hits confirmed by radioactivity assay

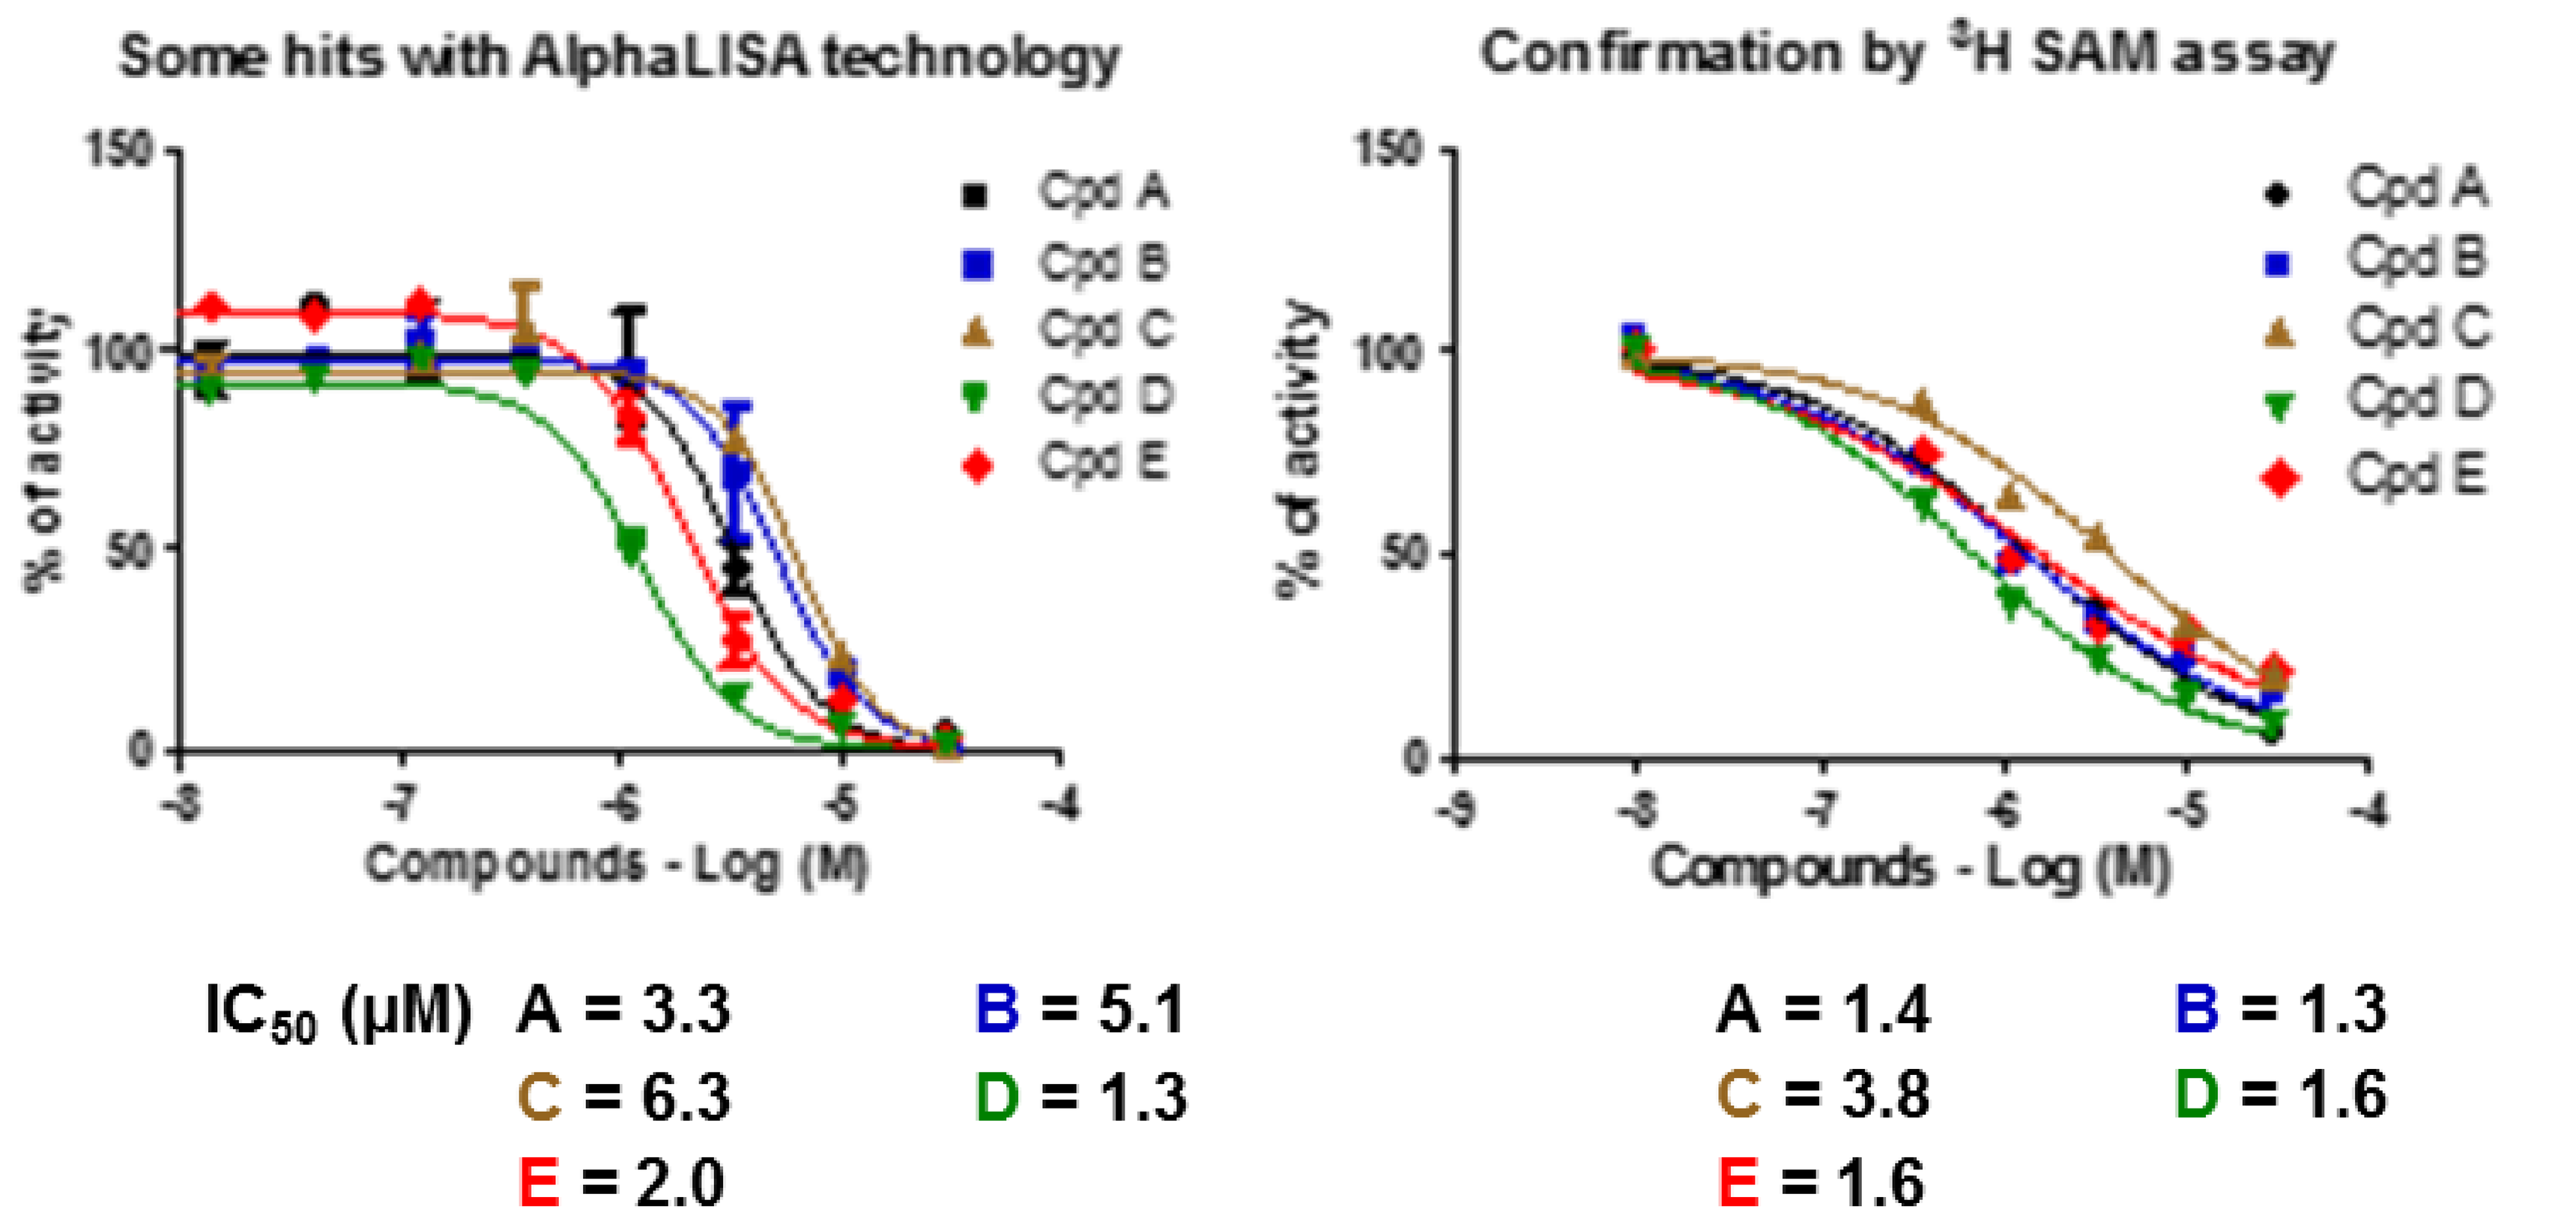
Some hits with AlphaLISA technology
Confirmation by 3H SAM assay
% of activity
Compounds - Log (M)
Cpd A
Cpd B
Cpd C
Cpd D
Cpd E
IC50 (µM) A = 3.3 B = 5.1
C = 6.3 D = 1.3
E = 2.0
A = 1.4 B = 1.3
C = 3.8 D = 1.6
E = 1.6

Thermal Shift Assay

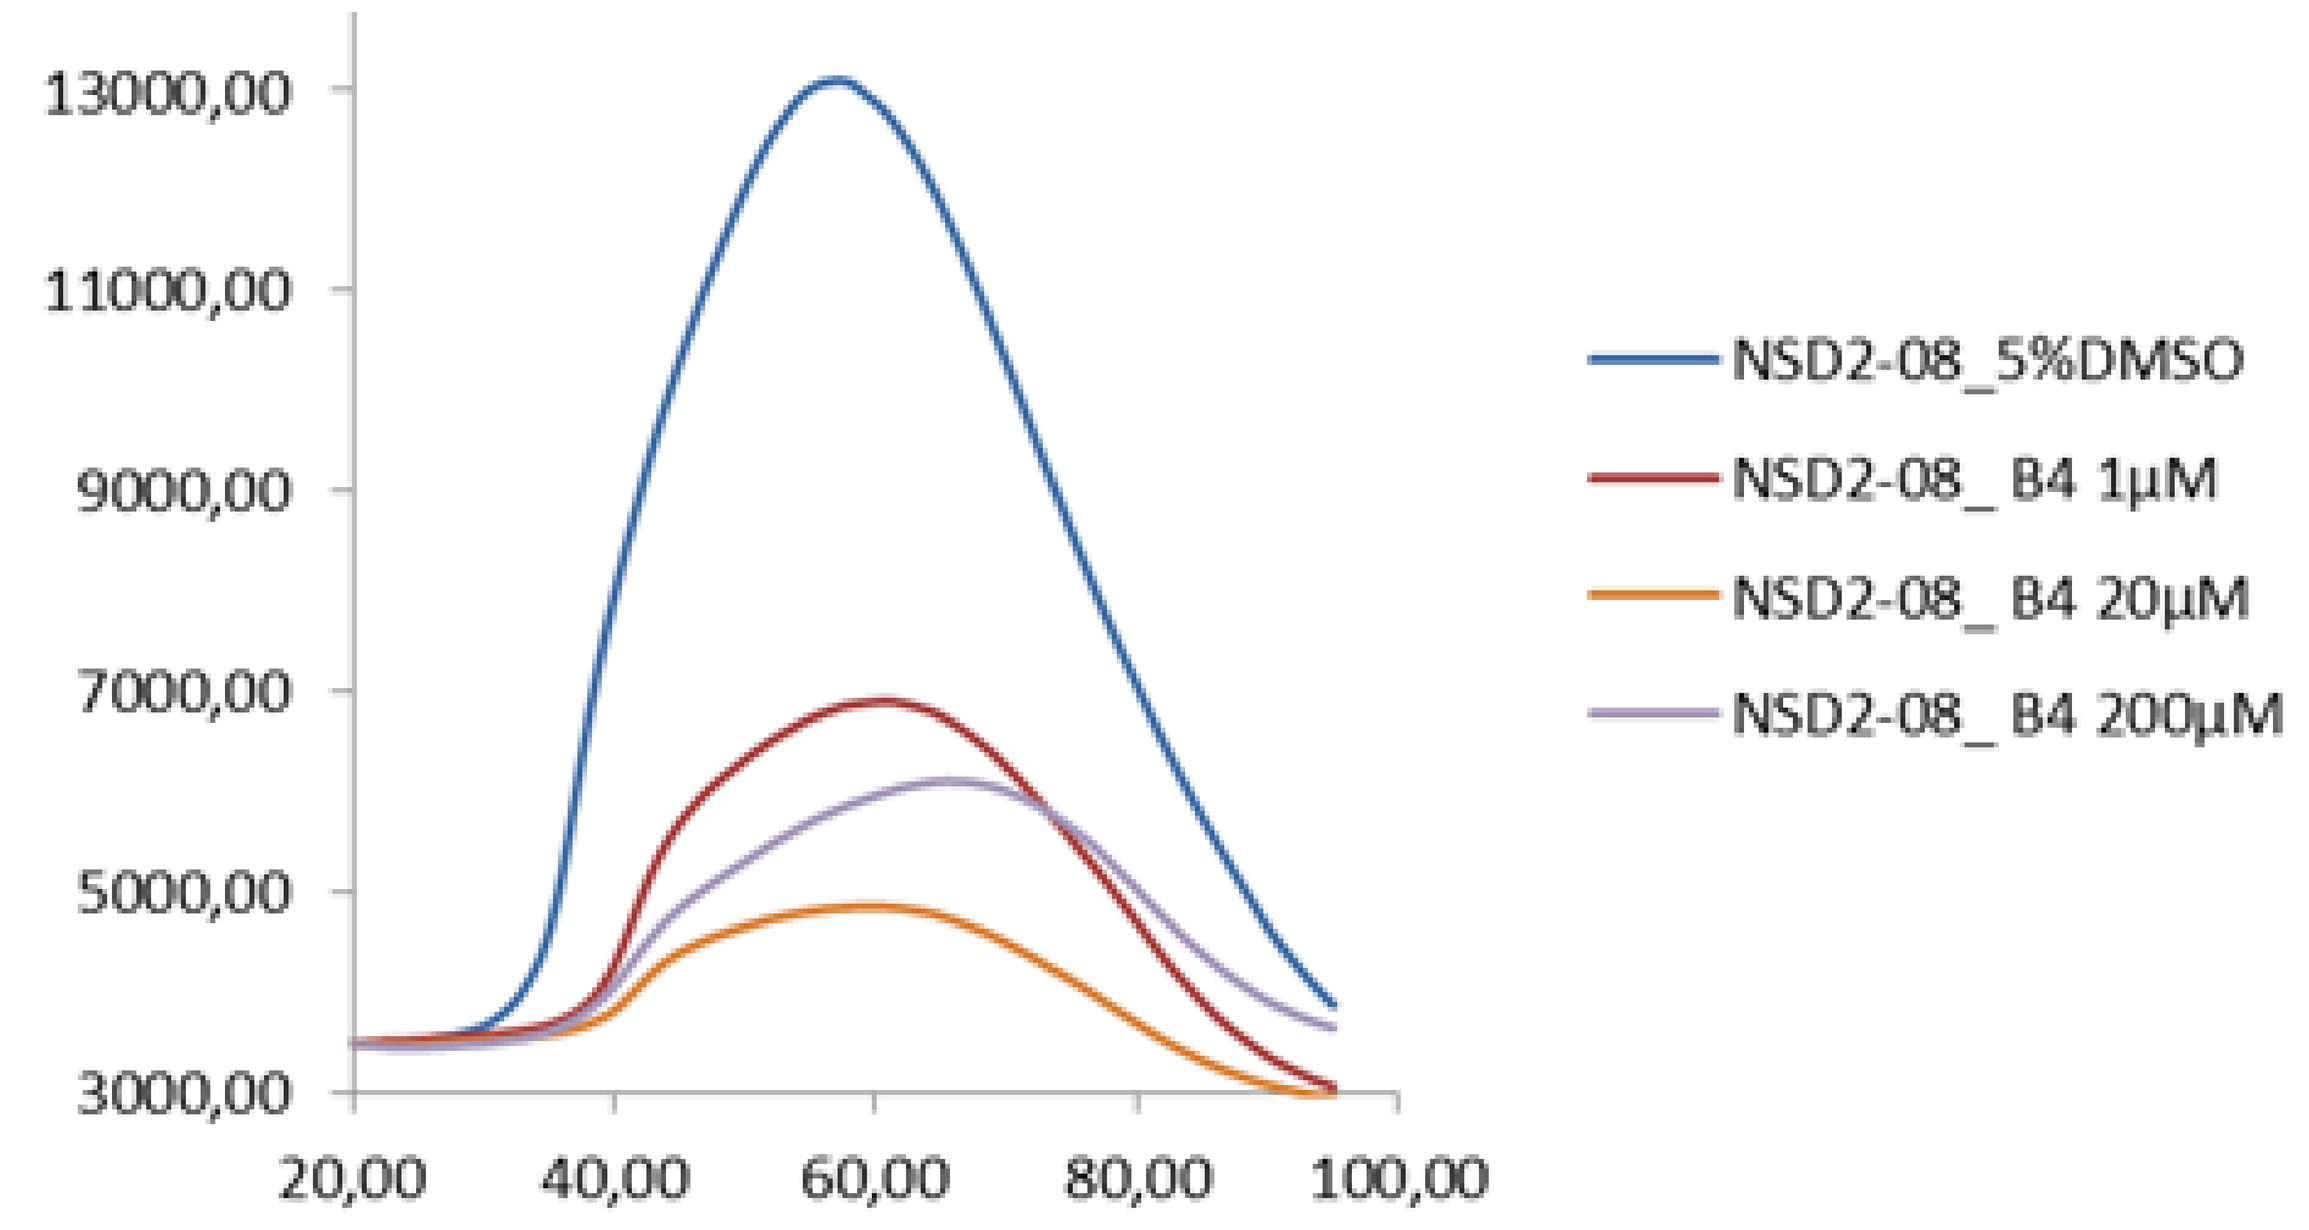
NSD2-08_5%DMSO
NSD2-08_ B4 1µM
NSD2-08_ B4 20µM
NSD2-08_ B4 200µM

Selected series exhibit ΔT_{NSD2} = 2 - 6.4°C

CONCLUSIONS

- Using the AlphaLisa™ technology followed by a subsequent orthogonal counter-screen based on 3H SAM incorporation, we have identified and selected chemical matters to enter into H2L phase
- The biomolecular interaction of our hits with NSD2 has been confirmed using a thermal shift assay. Furthermore, the SPR technology is currently being established in house to determine the compounds K_D for NSD2 and related isoforms (NSD1 and NSD3)
- In parallel, we are developing secondary cellular assays based on the H3K36me2 methylation and proliferation to further confirm the on-target hit activity
- NSD2 program is available for setting-up a drug discovery partnership
- Contacts: kristina.meyer@inventivapharma.com
 ursula.vogel@inventivapharma.com