IVA336 a Potential Substrate Reduction Therapy for Mucopolysaccharidose type-VI, -I, and -II Diseases

inventiva
50 rue de Dijon, 21121 Daix, France
(www.inventivapharma.com)

Philippe Masson, Eugeni Entchev, Sebastien Jacquet, Ingrid Jantzen, Olivier Lacombe, Véronique Douet, Pierre Broqua, Jean-Louis Junien, Jean-Louis Abitbol, Mireille Tallandier

1–INTRODUCTION

Background: Medical need for the treatment of MPS is still very high as ERTs have not been able to act in certain regions of the CNS, ophthalmological system and joints due to the poor penetration of the recombinant enzymes into blood-brain and blood-ocular barriers as well as into tissues that are poorly vascularized, such as cartilages and bones. IVA336 is an orally-active compound that allows the synthesis of soluble glycosaminoglycans (GAGs), mainly chondroitin sulfate (CS) and dermatan sulfate (DS). The elimination of neosynthesized soluble GAGs is mainly via urine excretion. By diverting endogenous GAG synthesis to the synthesis soluble IVA336-linked GAGs,IVA336 should decrease the intracellular pool of GAGs and consequently decrease the lysosomal GAG charge in MPS. This mechanism of action should translate into a substrate reduction therapy.

Objective: To investigate the activity of IVA336 in *in vitro* and *in vivo* MPS relevant models.

Methods: *In vitro*, the effect of IVA336 was tested on intracellular and extracellular GAG contents in fibroblasts isolated from donors affected with MPS I, VI, or II or from non-affected donors (cells originate from the Coriell Institute repository). *In vivo*, effects of IVA336 on GAG storage was studied in mice disease relevant model. Tissue distribution of IVA336 was studied *in vivo* in rats after oral administration.

2–IVA336 MECANISM OF ACTION IN MPS

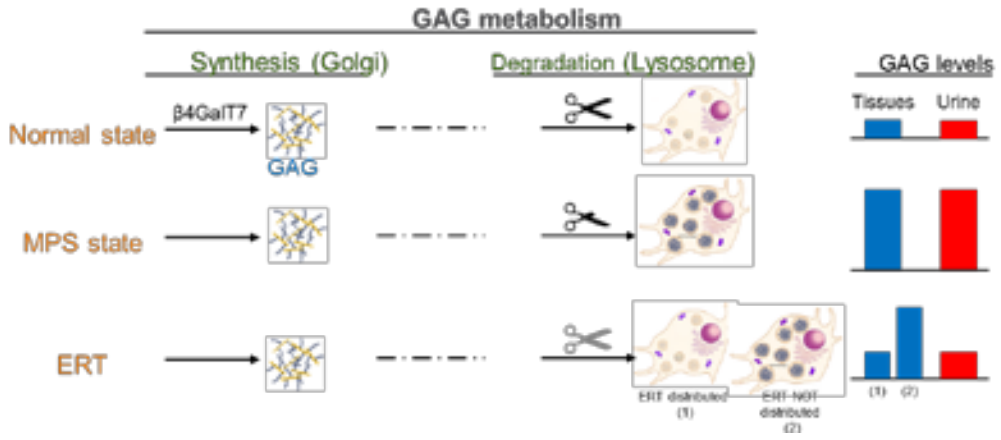
GAG metabolism
Synthesis (Golgi)
Degradation (Lysosome)
GAG levels
Tissues
Urine
Normal state
β4GalT7
GAG
MPS state
ERT
ERT distributed (1)
ERT NOT distributed (2)
(1) (2)

- HSCT and ERT bring quality-of-life improvements to patients
- Current therapeutic options present important limitations

New treatment strategies are needed to complete, improve or replace existing treatment and further improve patient quality-of-life

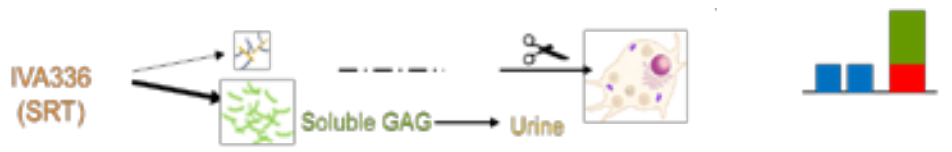
IVA336
(SRT)
Soluble GAG
Urine

By diverting endogenous PGs synthesis to soluble GAGs synthesis, IVA336 should decrease lysosomal GAGs charge in MPS

3–IVA336 DECREASES GAG STORAGE IN MPS VI FIBROBLASTS

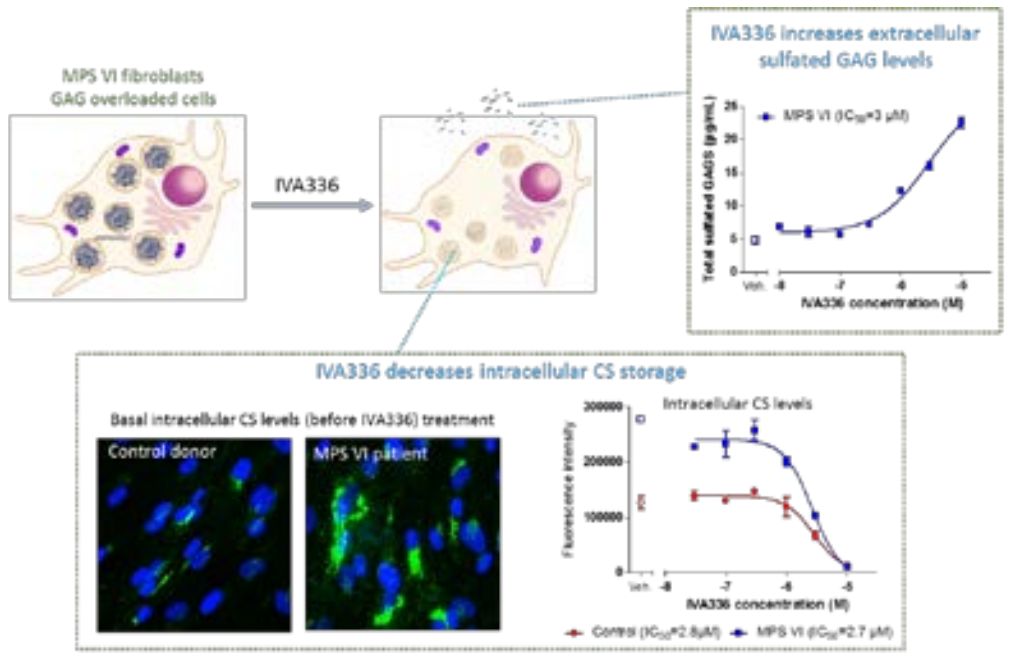
MPS VI fibroblasts
GAG overloaded cells
IVA336
IVA336 increases extracellular sulfated GAG levels
Total sulfated GAGS (µg/mL)
IVA336 concentration (M)
IVA336 decreases intracellular CS storage
Basal intracellular CS levels (before IVA336) treatment
Control donor
MPS VI patient
Intracellular CS levels
Fluorescence intensity
IVA336 concentration (M)

Activity of IVA336 *in vitro* on dermal fibroblasts isolated from MPS VI patients or from non-manifesting carriers.

- Before treatment the MPS VI fibroblasts are overloaded with CS, compared to non-affected fibroblasts.
- IVA336 decreases intracellular CS content in a dose dependent manner. Effect is comparable in MPS VI and non-affected fibroblasts.
- This intracellular decrease of GAG content is concomitant to increase of GAG content in the culture medium.

4–IVA336 ACTIVITY IN A MOUSE MPS MODEL

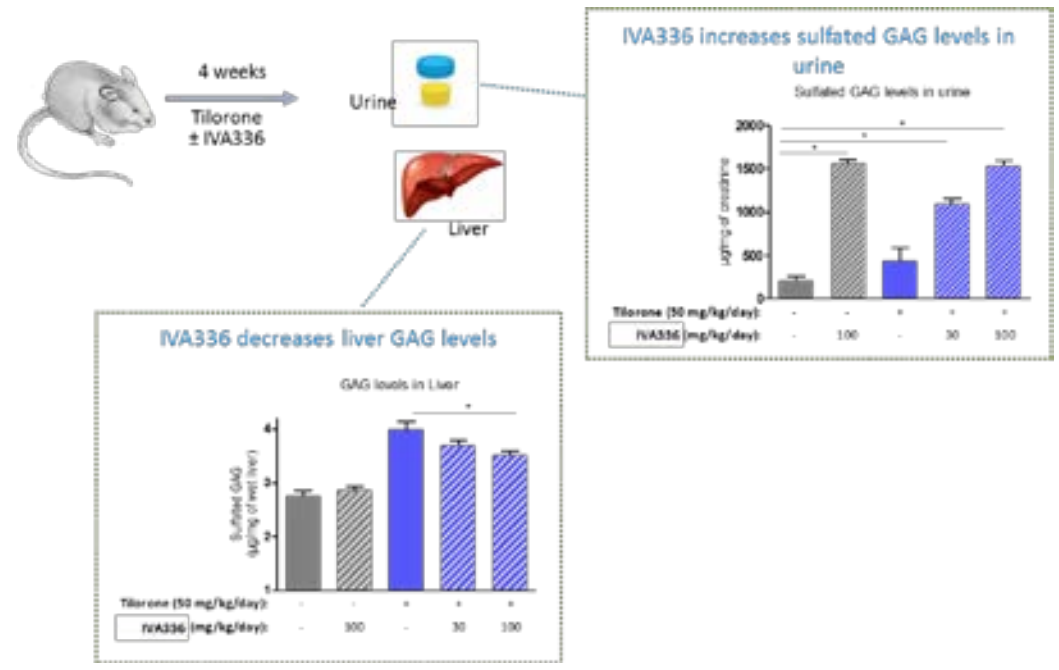
4 weeks
Tilorone
± IVA336
Urine
Liver
IVA336 increases sulfated GAG levels in urine
Sulfated GAG levels in urine
Tilorone (50 mg/kg/day):
IVA336 (mg/kg/day):
IVA336 decreases liver GAG levels
GAG levels in Liver

Activity of IVA336 *in vivo* a mice model were GAG accumulation is induced by tilorone, a non-specific chemical inducer.

- As expected, after a 4-week treatment, tilorone increases GAG levels in liver.
- In the liver, IVA336 decreases total GAG level induced by tilorone.
- This decrease of liver GAG is concomitant to increase of GAG levels in the urine.

5–IVA336 TISSUE DISTRIBUTION

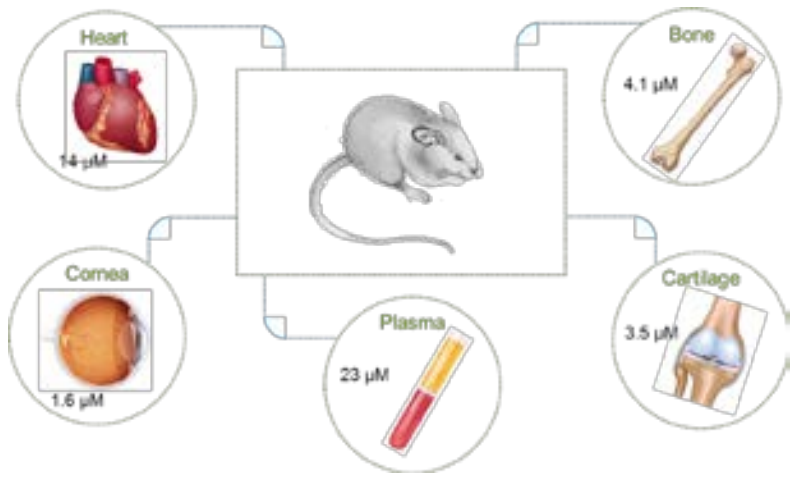
Heart
14 µM
Bone
4.1 µM
Cornea
1.6 µM
Plasma
23 µM
Cartilage
3.5 µM

In rat after repeat oral gavage, IVA336 is widely distributed in tissues and found at pharmacological active concentrations in tissues that are poorly targeted by ERTs

6–CLINICAL DATA IN POST OPERATIVE THROMBOSIS PREVENTION

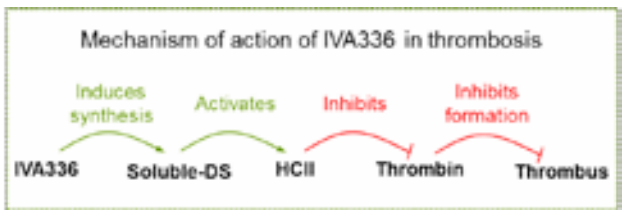
Mechanism of action of IVA336 in thrombosis
Induces synthesis
Activates
Inhibits
Inhibits formation
IVA336
Soluble-DS
HCII
Thrombin
Thrombus

- ~700 healthy volunteers in 29 phase 1, employing single-doses (50 to 1500 mg) and multiple-doses (50 to 1000 mg/day) administered for up to 14 days
- ~1100 subjects with thrombosis risk received IVA336 in 3 phase 2 clinical trials. IVA336 given orally bid (250 to 1000 mg/day) for up to 16 weeks
- IVA336 was safe and well tolerated
- IVA336 induced significant elevation in soluble-CS/DS levels

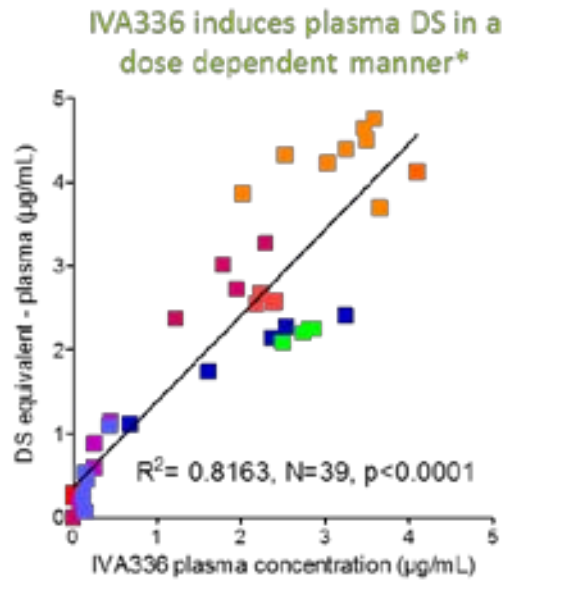
IVA336 induces plasma DS in a dose dependent manner*
DS equivalent - plasma (µg/mL)
R² = 0.8163, N=39, p<0.0001
IVA336 plasma concentration (µg/mL)

**Each data point is the mean values (Anti IIa activity) obtained in one clinical study for a group of subjects receiving the same dose; Each color represents a clinical study.*

7–CONCLUSION

IVA336 is a small molecule orally active that has demonstrated safety and tolerability in adult subjects in clinical trials. IVA336 is reducing GAG storage *in vitro* and *in vivo*. After oral administration, IVA336 is distributed throughout the body and is found at pharmacological active concentrations in bones, cartilages, retina, and heart. IVA336 is proposed as novel substrate reduction therapy for MPS in which CS and/or DS accumulate. Together, the data support the clinical investigation of IVA336 for the treatment of MPS I, II, and VI.